2716

The O-glycan Epitope Targeting Anti-Human Carcinoma Monoclonal Antibody (mAb) NEO-201 can also target human regulatory T cells (Tregs)

Atsushi Tanaka[1], Massimo Fantini[2], Philip M. Arlen[2], Christina M. Annunziata[3], Kwong Y. Tsang[2], Shimon Sakaguchi[1]

[1]Laboratory of Experimental Immunology, WPI Immunology, Frontier Research Center, Osaka University, Osaka, Japan,[2]Precision Biologics, Inc., Bethesda, MD,[3]Women's Malignancies Branch, Center for Cancer Research, National Cancer Institute, National Institutes of Health, Bethesda, MD

Introduction: NEO-201 is a humanized IgG1 mAb that targets Core 1 and/or extended Core 1 O-glycans expressed by several human solid and blood tumors, as well as neutrophils, but it does not bind to most normal tissues and human immune cell subsets (B cells, CD4+ T cells, CD8+ T cells, NK cells, monocytes). Functional analysis revealed that NEO-201 mediates the killing of its target cells through ADCC and CDC (Tsang et al 2022; Fantini et al 2018). Previous study has shown that approximately 4.6% of CD4+ T cells express NEO-201 target antigen and that those CD4+ T cells have Tregs phenotype. In addition, in a recent published phase 1 study we observed that NEO-201 binds to CD4+/ CD25+/, CD127-/, Foxp3+/, CD15s+ cells from peripheral blood mononuclear cells (PBMCs) from cancer patients with solid tumors (Cole et al 2023). The purpose of this study was to further characterize the phenotype of the specific subset of CD4+ T cells expressing the NEO-201 target antigen.

Experimental Design: Human PBMCs were obtained from 7 healthy donors (HD) collected at Osaka University and 6 cancer patients from our ongoing phase II clinical trial (Clinical Trial NCT03476681) evaluating the efficiency of the combination of NEO-201 with pembrolizumab in adults with solid tumors resistant to checkpoint inhibitors. Phenotypic analysis was performed by flow cytometry using NEO-201 and antibodies CD3, CD4, CD25, CD45RA, FoxP3, and CD15s. The same gating strategy was applied for both normal donors and patients to evaluate which fraction of CD4+ T cells is recognized by NEO-201. Fraction (Fr) 1 is naïve Treg, Fr II is effector Treg, Fr III is non-Treg, Fr IV is effector CD4 T cells and Fr V is naïve CD4 T cells (Tanaka et al 2017).

Results

1. NEO-201 Binds to O-Glycans

O-Glycan array containing 94 different O-glycans to test the binding to NEO-201

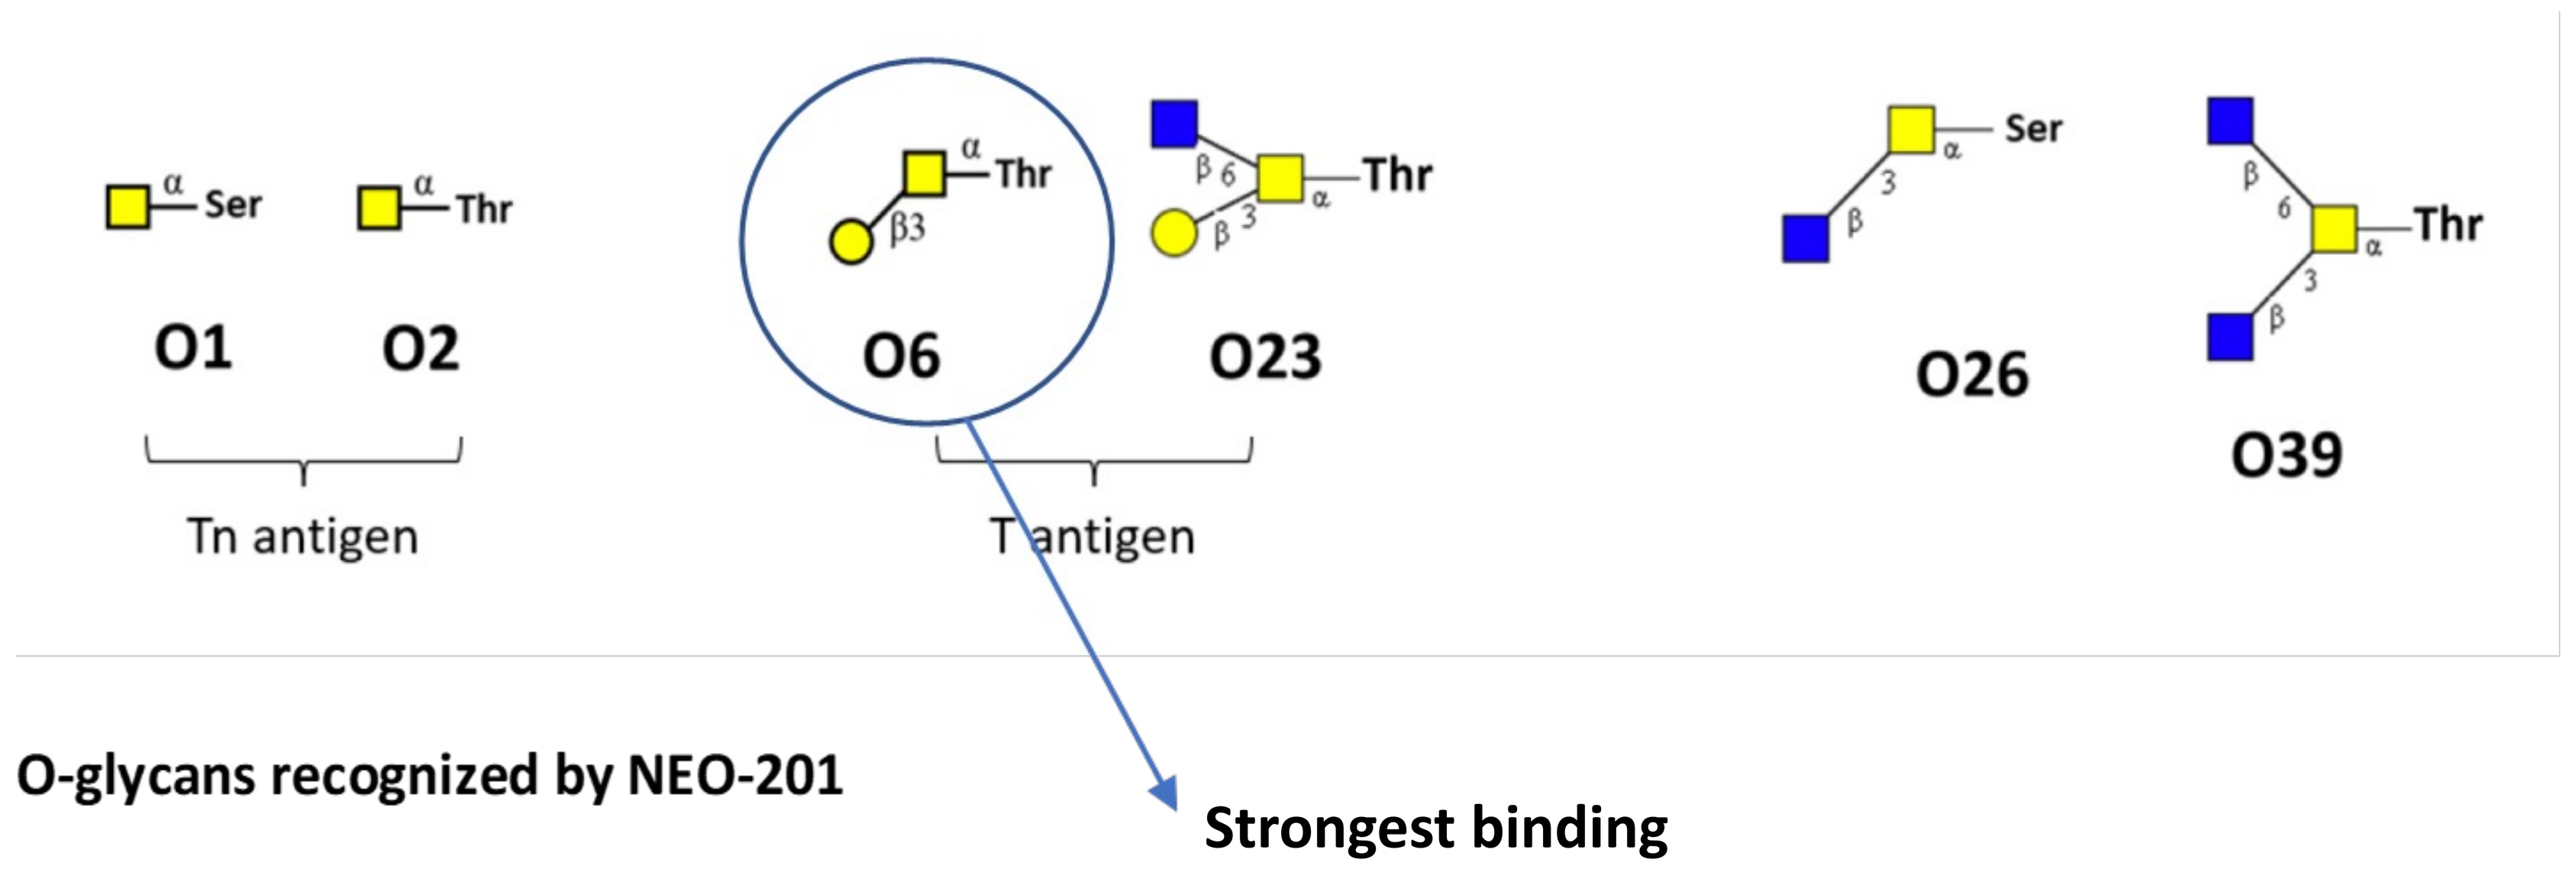

Ser
Thr
O1
O2
O6
O23
O26
O39
Tn antigen
T antigen
O-glycans recognized by NEO-201
Strongest binding

The 06 is core 1 O-glycan and has the strongest binding to NEO-201

01 and 02 are Tn antigens. 023 is core 2, 026 is core 3 and 039 is core 4

2. NEO-201 recognizes naïve Tregs in PBMCs

Flow cytometry analysis of the expression of NEO-201 in Tregs from PBMC isolated from healthy donors

Donor	% of Fr I (Naïve Treg)	% NEO201+ cells in Fr I	% of Fr II (Effector Treg)	% NEO201+ cells in Fr II
24	2.21%	0.54%	3.33%	0.0%
86	0.77%	0.0%	2.60%	0.0%
62	3.07%	1.75%	1.15%	0.69%
819	1.24%	0.0%	2.00%	0.0%
129	1.41%	0.13%	0.98%	0.0%
238T	7.26%	0.13%	1.59%	0.0%
238C	4.57%	0.31%	3.05%	0.0%

Flow cytometry analysis of the expression of NEO-201 in Tregs from PBMC isolated from cancer patients

Cancer patient	% of Fr I (Naïve Treg)	% NEO201+ cells in Fr I	% of Fr II (Effector Treg)	% NEO201+ cells in Fr II
1	8.87%	16.44%	0.36%	0.0%
5	9.34%	52.87%	0.43%	12.5%
6	9.44%	63.29%	2.63%	77.27%
7	13.40%	53.49%	0.93%	0.0%
8	9.16%	32.61%	1.00%	0.0%
11	5.14%	77.63%	0.07%	0.0%

3. NEO-201 recognizes naïve Tregs in PBMCs: flow cytometry plots

Flow Cytometry of PBMCs from a healthy donor

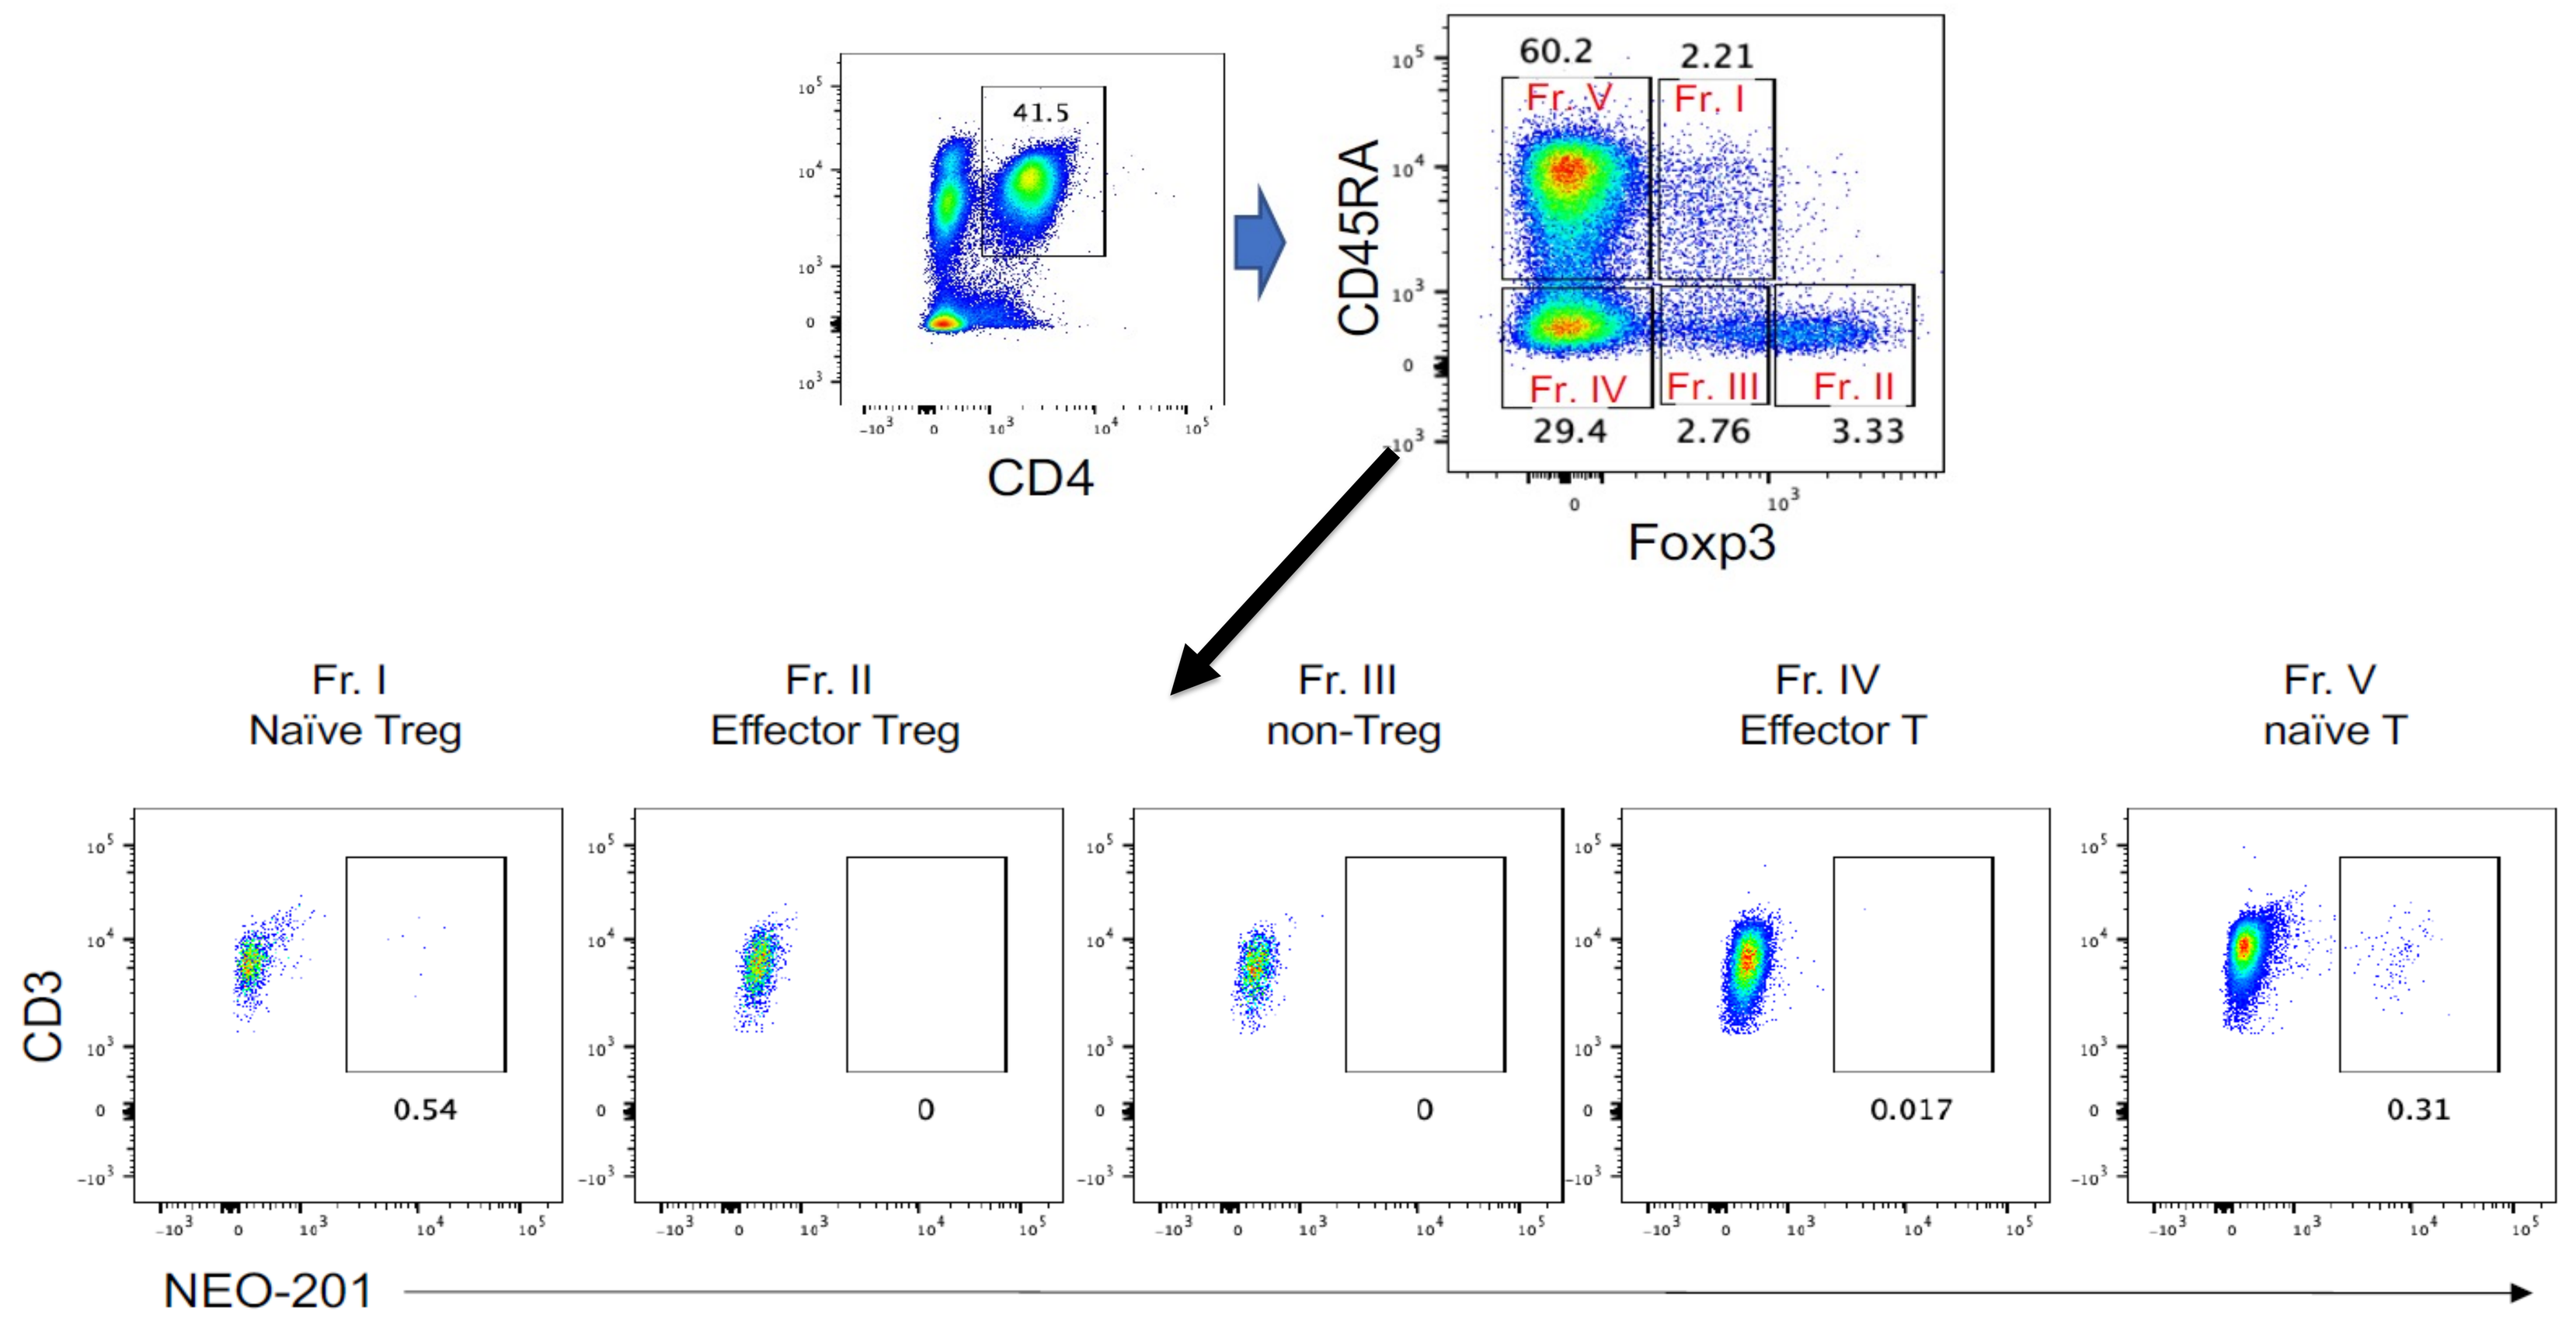

41.5
CD4
CD45RA
60.2
2.21
Fr. V
Fr. I
Fr. IV
Fr. III
Fr. II
29.4
2.76
3.33
Foxp3
Fr. I
Naïve Treg
Fr. II
Effector Treg
Fr. III
non-Treg
Fr. IV
Effector T
Fr. V
naïve T
CD3
0.54
0
0
0.017
0.31
NEO-201

Flow Cytometry from PBMCs from a cancer patient

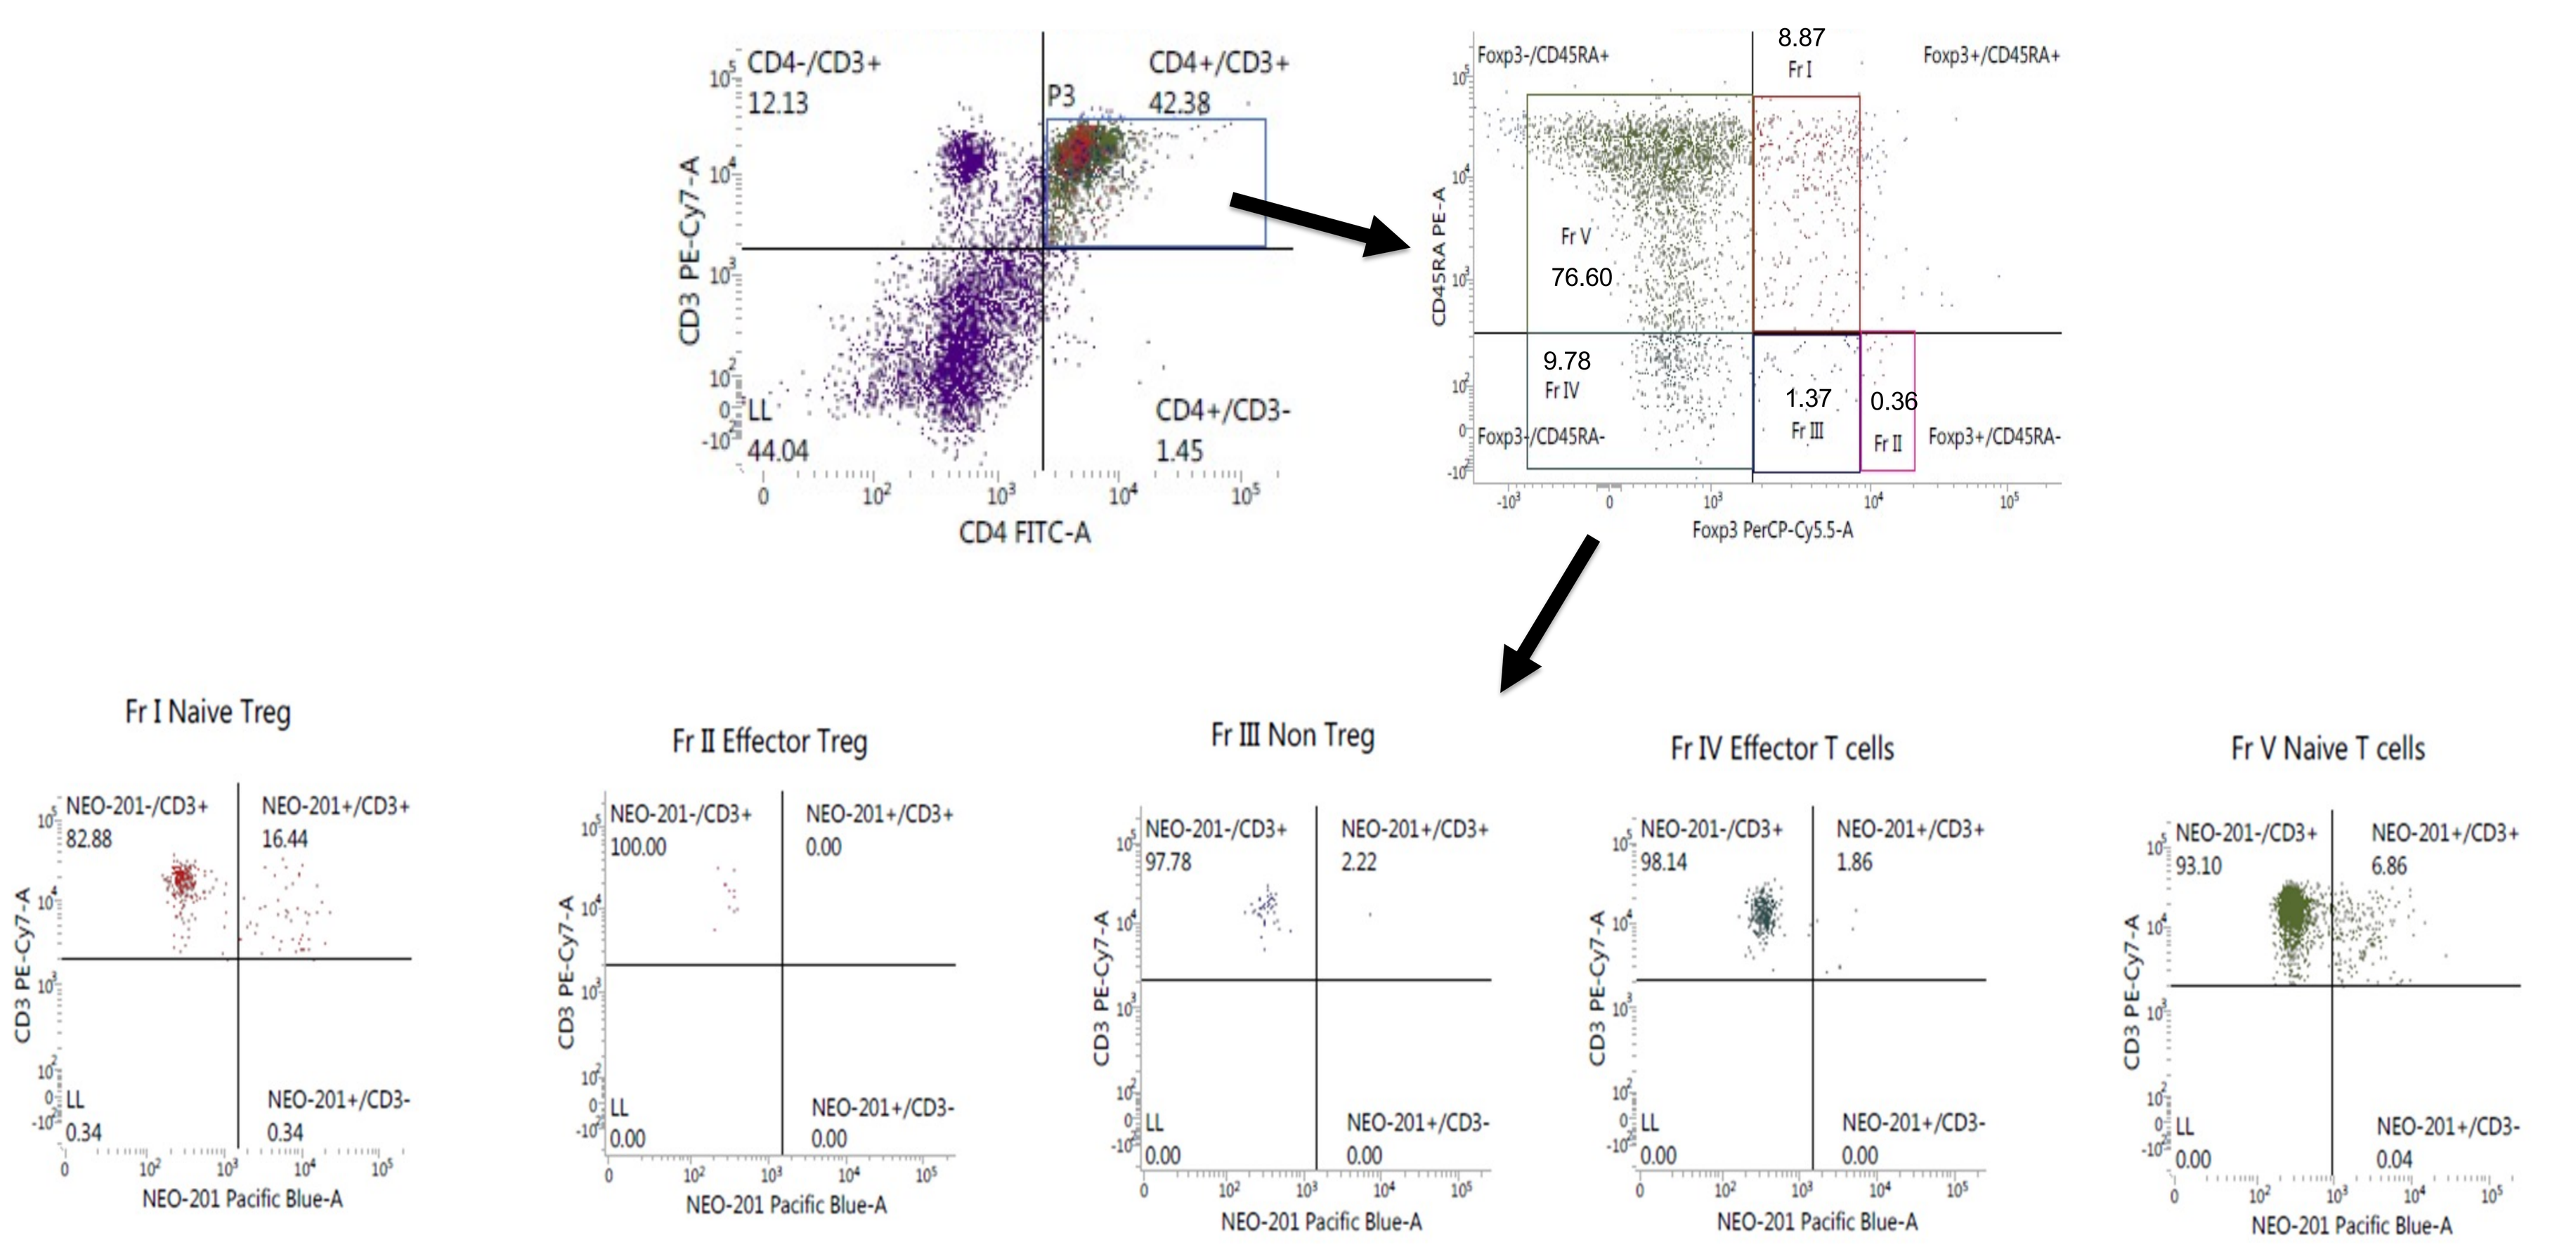

CD4-/CD3+
12.13
CD4+/CD3+
42.38
P3
LL
44.04
CD4+/CD3-
1.45
CD3 PE-Cy7-A
CD4 FITC-A
Foxp3-/CD45RA+
8.87
Fr I
Foxp3+/CD45RA+
Fr V
76.60
9.78
Fr IV
1.37
Fr III
0.36
Fr II
Foxp3-/CD45RA-
Foxp3+/CD45RA-
CD45RA PE-A
Foxp3 PerCP-Cy5.5-A
Fr I Naive Treg
NEO-201-/CD3+
82.88
NEO-201+/CD3+
16.44
NEO-201+/CD3-
0.34
Fr II Effector Treg
100.00
0.00
Fr III Non Treg
97.78
2.22
Fr IV Effector T cells
98.14
1.86
Fr V Naive T cells
93.10
6.86
0.04
NEO-201 Pacific Blue-A

Example of flow cytometry analysis of PBMCs from one healthy donor (#24) and one cancer patient (#1). Flow cytometry analysis revealed that NEO-201 recognizes naïve Tregs (nTregs, Fraction I: CD3+/CD4+/CD45RA+/Foxp3^{low} cells) while it does not bind to effector Tregs (eTregs, fraction II: CD3+/CD4+/CD45RA-/Foxp3^{high} cells) in both HD and cancer patients. The % of nTregs in cancer patients was higher than HD. Preliminary results from the ongoing Phase II clinical trial showed that subjects with durable stable disease vs progressive disease had a reduction of circulating nTregs after treatment with NEO-201 compared to baseline levels.

Conclusions: NEO-201 binds to human Tregs with significantly higher % of binding to Fr I and a greater % of Tregs expressing NEO-201 target antigen in cancer patients compared to HD. It is conceivable that naïve Tregs in cancer patients express high levels of Core 1 O-glycans. Furthermore, when subjected to TCR stimulation, naïve Tregs undergo proliferation and differentiate into eTregs. eTregs can then infiltrate into tumor microenvironment (TME). These data suggest that depletion of circulating nTregs in cancer patients after NEO-201 treatment may prevent the differentiation of nTregs into eTregs and their accumulation in the TME. This study suggests the potential use of NEO-201 to reduce the Treg-mediated suppression of anticancer immunity.

References:

Tsang KY et al. Cancer 14 (20) 4999, 2022.

Fantini M et el. Front. Immunol 8:1889, 2018

Cole CB et al. J Exp Clin Cancer Res 42 (11):76, 2023

Tanaka A et al. Cell Research 27:109, 2017.

www.PosterPresentations.com