O-glycan array to test the binding activity of NEO-201 to 94 different O-glycan structures (Figure S2).

Identification of the O-glycan epitope targeted by the anti-human carcinoma monoclonal antibody (mAb) NEO-201

Kwong Y. Tsang[1], Massimo Fantini[1], Anjum Zaki[1], Sharon A. Mavroukakis[1], Christina M. Annunziata[2] and Philip M Arlen[1]

[1]Precision Biologics, Inc., Bethesda MD, USA; [2]*Women's Malignancies Branch, Center for Cancer Research, National Cancer Institute, National Institutes of Health, Bethesda, MD, USA*

Introduction

NEO-201 is an IgG1 monoclonal antibody (mAb) reactive against multiple human cancers but not against most normal epithelial tissues. NEO-201 can mediate antitumor activity through multiple mechanisms such as antibody-dependent cellular cytotoxicity (ADCC), complement dependent cytotoxicity (CDC), and blockade of the CEACAM5/CEACAM1 immune checkpoint inhibitory pathway.
In addition to solid malignant tumors, flow cytometry analysis has demonstrated that NEO-201 binds to 98.9% of CD15+ granulocytes, human regulatory T cells as well as various human hematological neoplastic cell lines.
However, NEO-201 does not bind to other immune subsets, such as B cells, NK cells, monocytes, or CD8$^+$ T cells, and to the majority of CD4+ T cells.
In previous studies, we observed that NEO-201 binds to cancer cells expressing tumor-variant forms of CEACAM5 and/or CEACAM6. Since CEACAMs are highly glycosylated proteins and human regulatory T cells and some hematological neoplastic cell lines (HL-60 and U937) recognized by NEO-201 do not express CEACAM5 or CEACAM6, we hypothesized that the actual target of NEO-201 could be a glycan attached to proteins of cancer cells.
Glycosylation is an important post-translation modification of proteins and is affected by oncogenesis. The expression of incomplete/truncated O-glycans in cancer cells occurs in both solid and liquid tumors and is correlated with poor prognosis and tumor progression.
This study was designed to focus on the identification of the O-glycan binding epitope of NEO-201 and to evaluate if NEO-201 target cells express O-glycans recognized by NEO-201

Experimental Design

Mammalian and bacterial recombinant human (rh) CEACAM6 was used as a model to evaluate if post-translational modifications made by mammalian cells on proteins are crucial for NEO-201 binding. An O-glycan array consisting of 94 O-glycans was used to identify the O-glycans targeted by NEO-201. Expression of O-glycans recognized by NEO-201 was evaluated in NEO-201 target cells, including human pancreatic cancer cell line (CFPAC-1), human hematological neoplastic cell lines (HL60, U937, K562) and human neutrophils. These cells were also used as target cells to prove the specific killing by NEO-201 through ADCC.

Results

1. NEO-201 Binds Specifically to Human Tumor Tissues

Normal vs. Tumor Tissue Microarray IHC

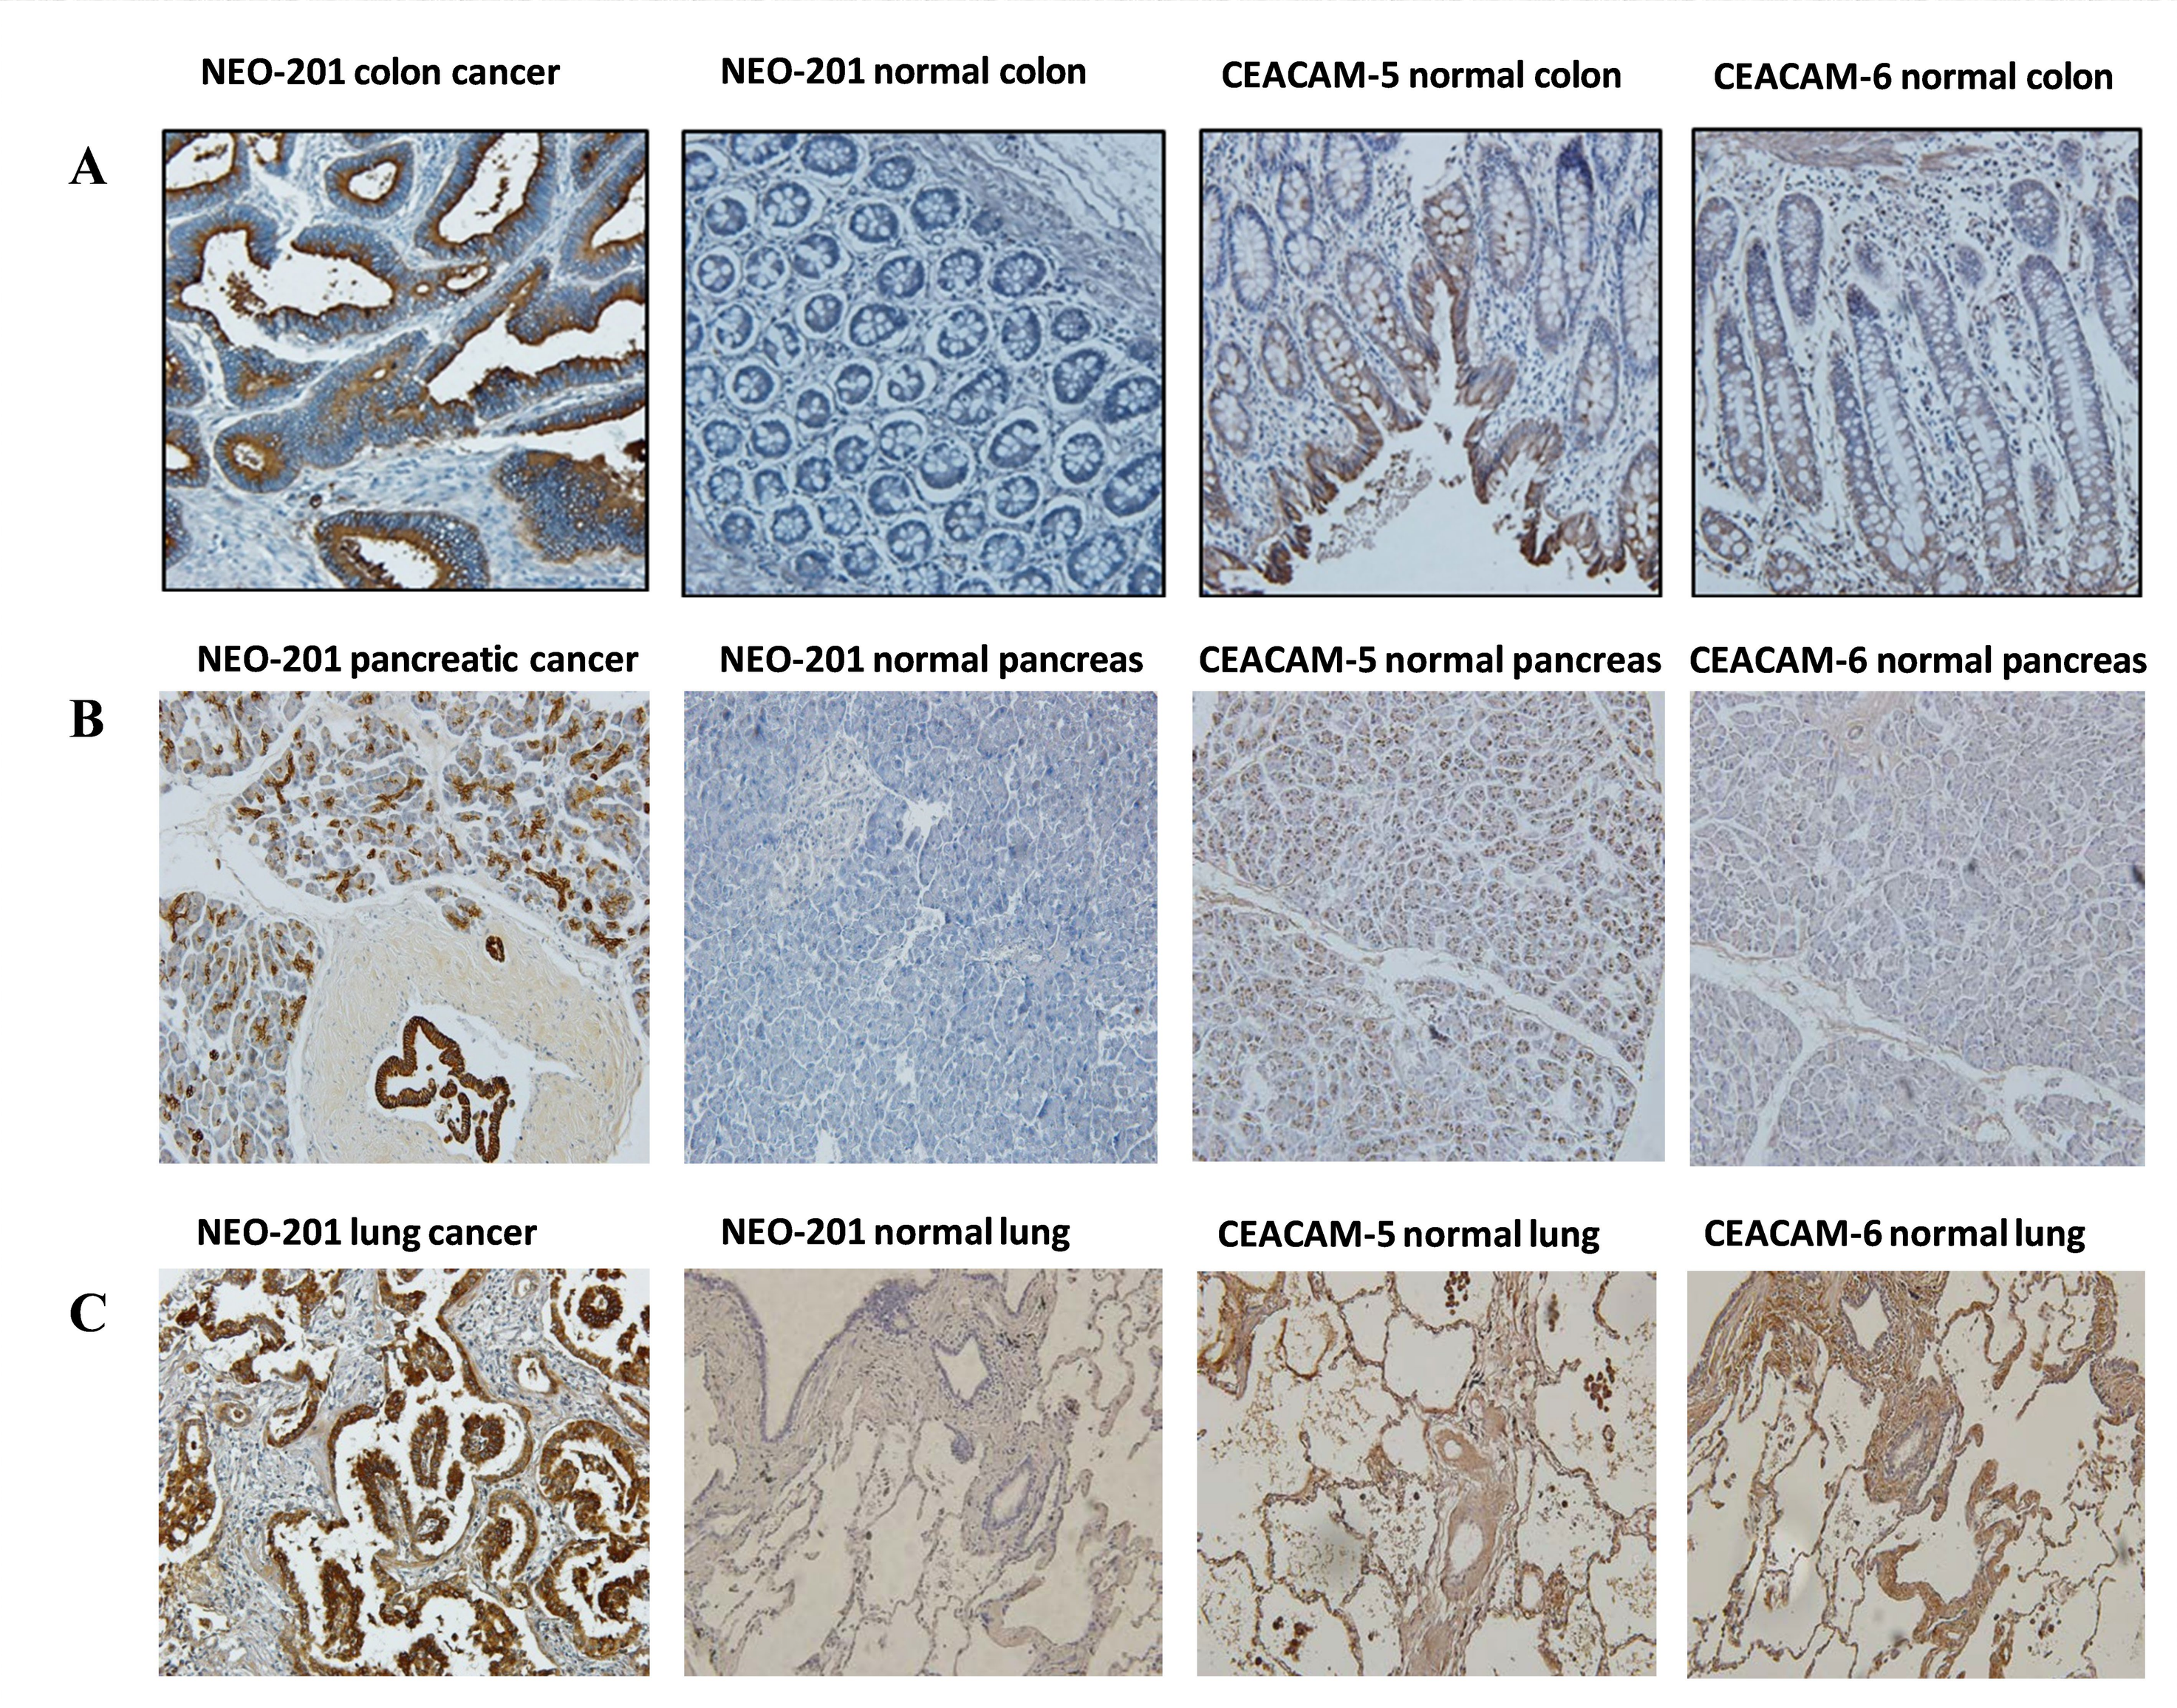

NEO-201 colon cancer
NEO-201 normal colon
CEACAM-5 normal colon
CEACAM-6 normal colon
A
NEO-201 pancreatic cancer
NEO-201 normal pancreas
CEACAM-5 normal pancreas
CEACAM-6 normal pancreas
B
NEO-201 lung cancer
NEO-201 normal lung
CEACAM-5 normal lung
CEACAM-6 normal lung
C

2. NEO-201 Targets Human CD15$^+$ Granulocytes and Different Hematological Neoplastic Cell Lines

PMBCs and Tumor Cell Line Flow Cytometry

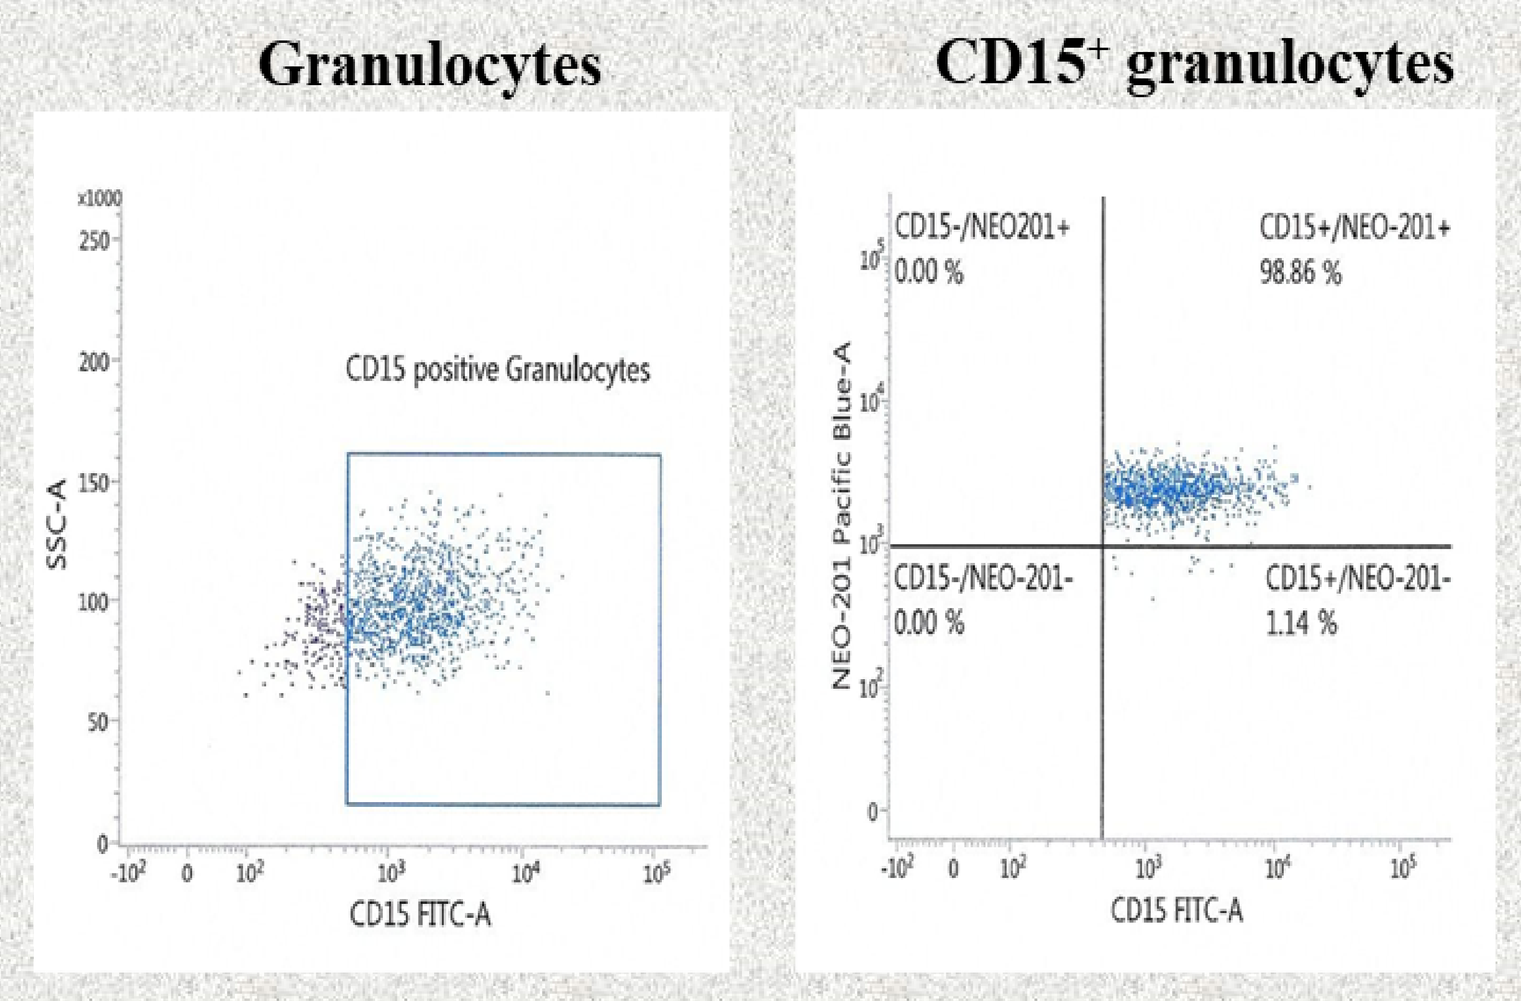

Granulocytes
CD15+ granulocytes
CD15 positive Granulocytes
SSC-A
CD15 FITC-A
NEO-201 Pacific Blue-A
CD15-/NEO201+ 0.00 %
CD15+/NEO-201+ 98.86 %
CD15-/NEO-201- 0.00 %
CD15+/NEO-201- 1.14 %

NEO-201 positivity was defined as % positive >10%.

Cell line	Tumor Type	% positive	MFI
HL60	AML	**47.0**	4,864
U937	AML	**99.5**	5,985
MOLM13	AML	**100**	28,278
AML2	AML	**100**	17,736
IMS-M2	AML	**97.8**	4,640

NEO-201 positivity was defined as % positive >5%.
AML: Acute Myeloid Leukemia

3. NEO-201 binds to mammalian-expressed rhCEACAM6 but not to bacterial-expressed rhCEACAM6 by ELISA

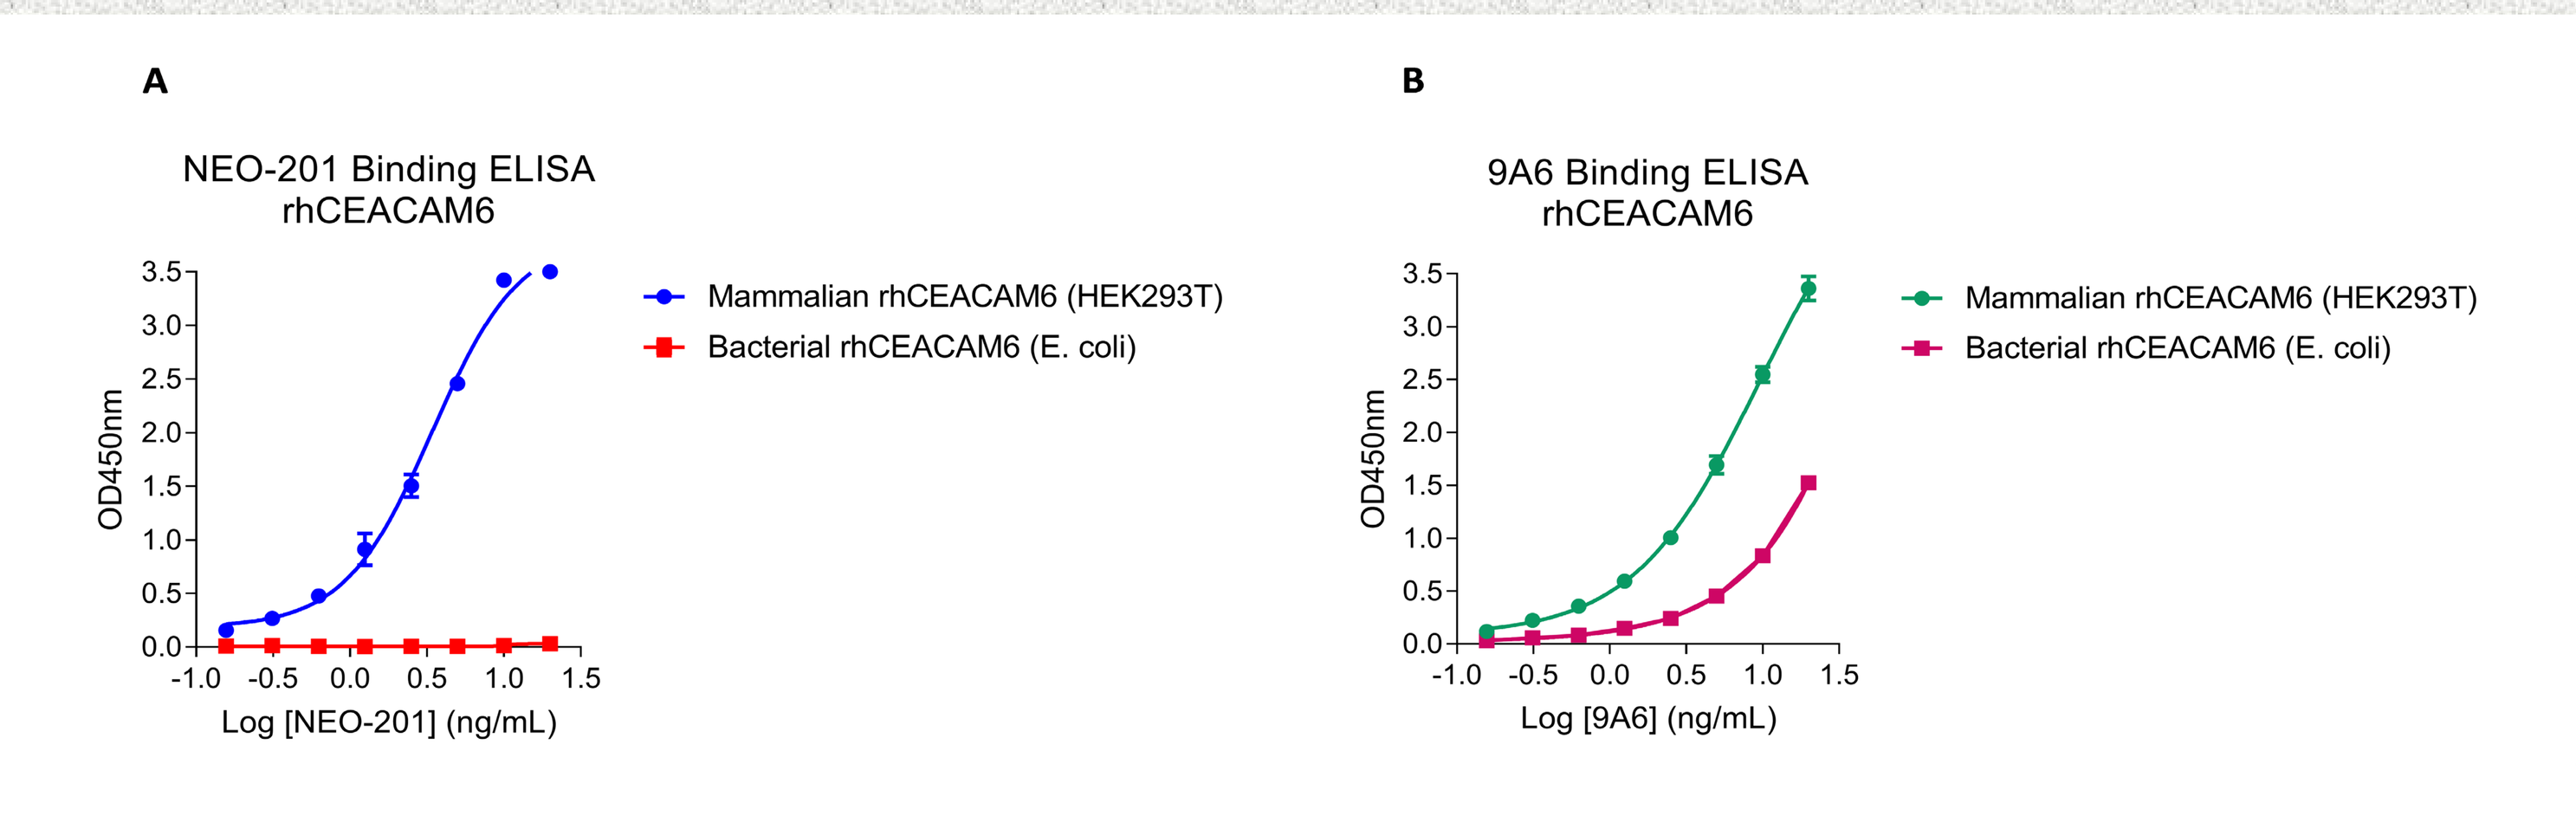

A
NEO-201 Binding ELISA
rhCEACAM6
Mammalian rhCEACAM6 (HEK293T)
Bacterial rhCEACAM6 (E. coli)
OD450nm
Log [NEO-201] (ng/mL)
B
9A6 Binding ELISA
rhCEACAM6
Mammalian rhCEACAM6 (HEK293T)
Bacterial rhCEACAM6 (E. coli)
OD450nm
Log [9A6] (ng/mL)

ELISA comparing the binding of NEO-201 and the commercial anti-CEACAM6 mAb 9A6 to HEK293 (mammalian, post-translational modification-competent) expressed rhCEACAM6 or E. coli (bacterial, post-translational modification-incompetent) expressed rhCEACAM6.

rhCEACAM6 was used at 400 ng/mL. NEO-201 (A) and 9A6 (B) were used at two-fold dilutions down from 20 ng/mL.

4. NEO-201 Binds to O-Glycans

O-Glycan Array to Test the Binding of NEO-201 to 94 Different O-Glycans

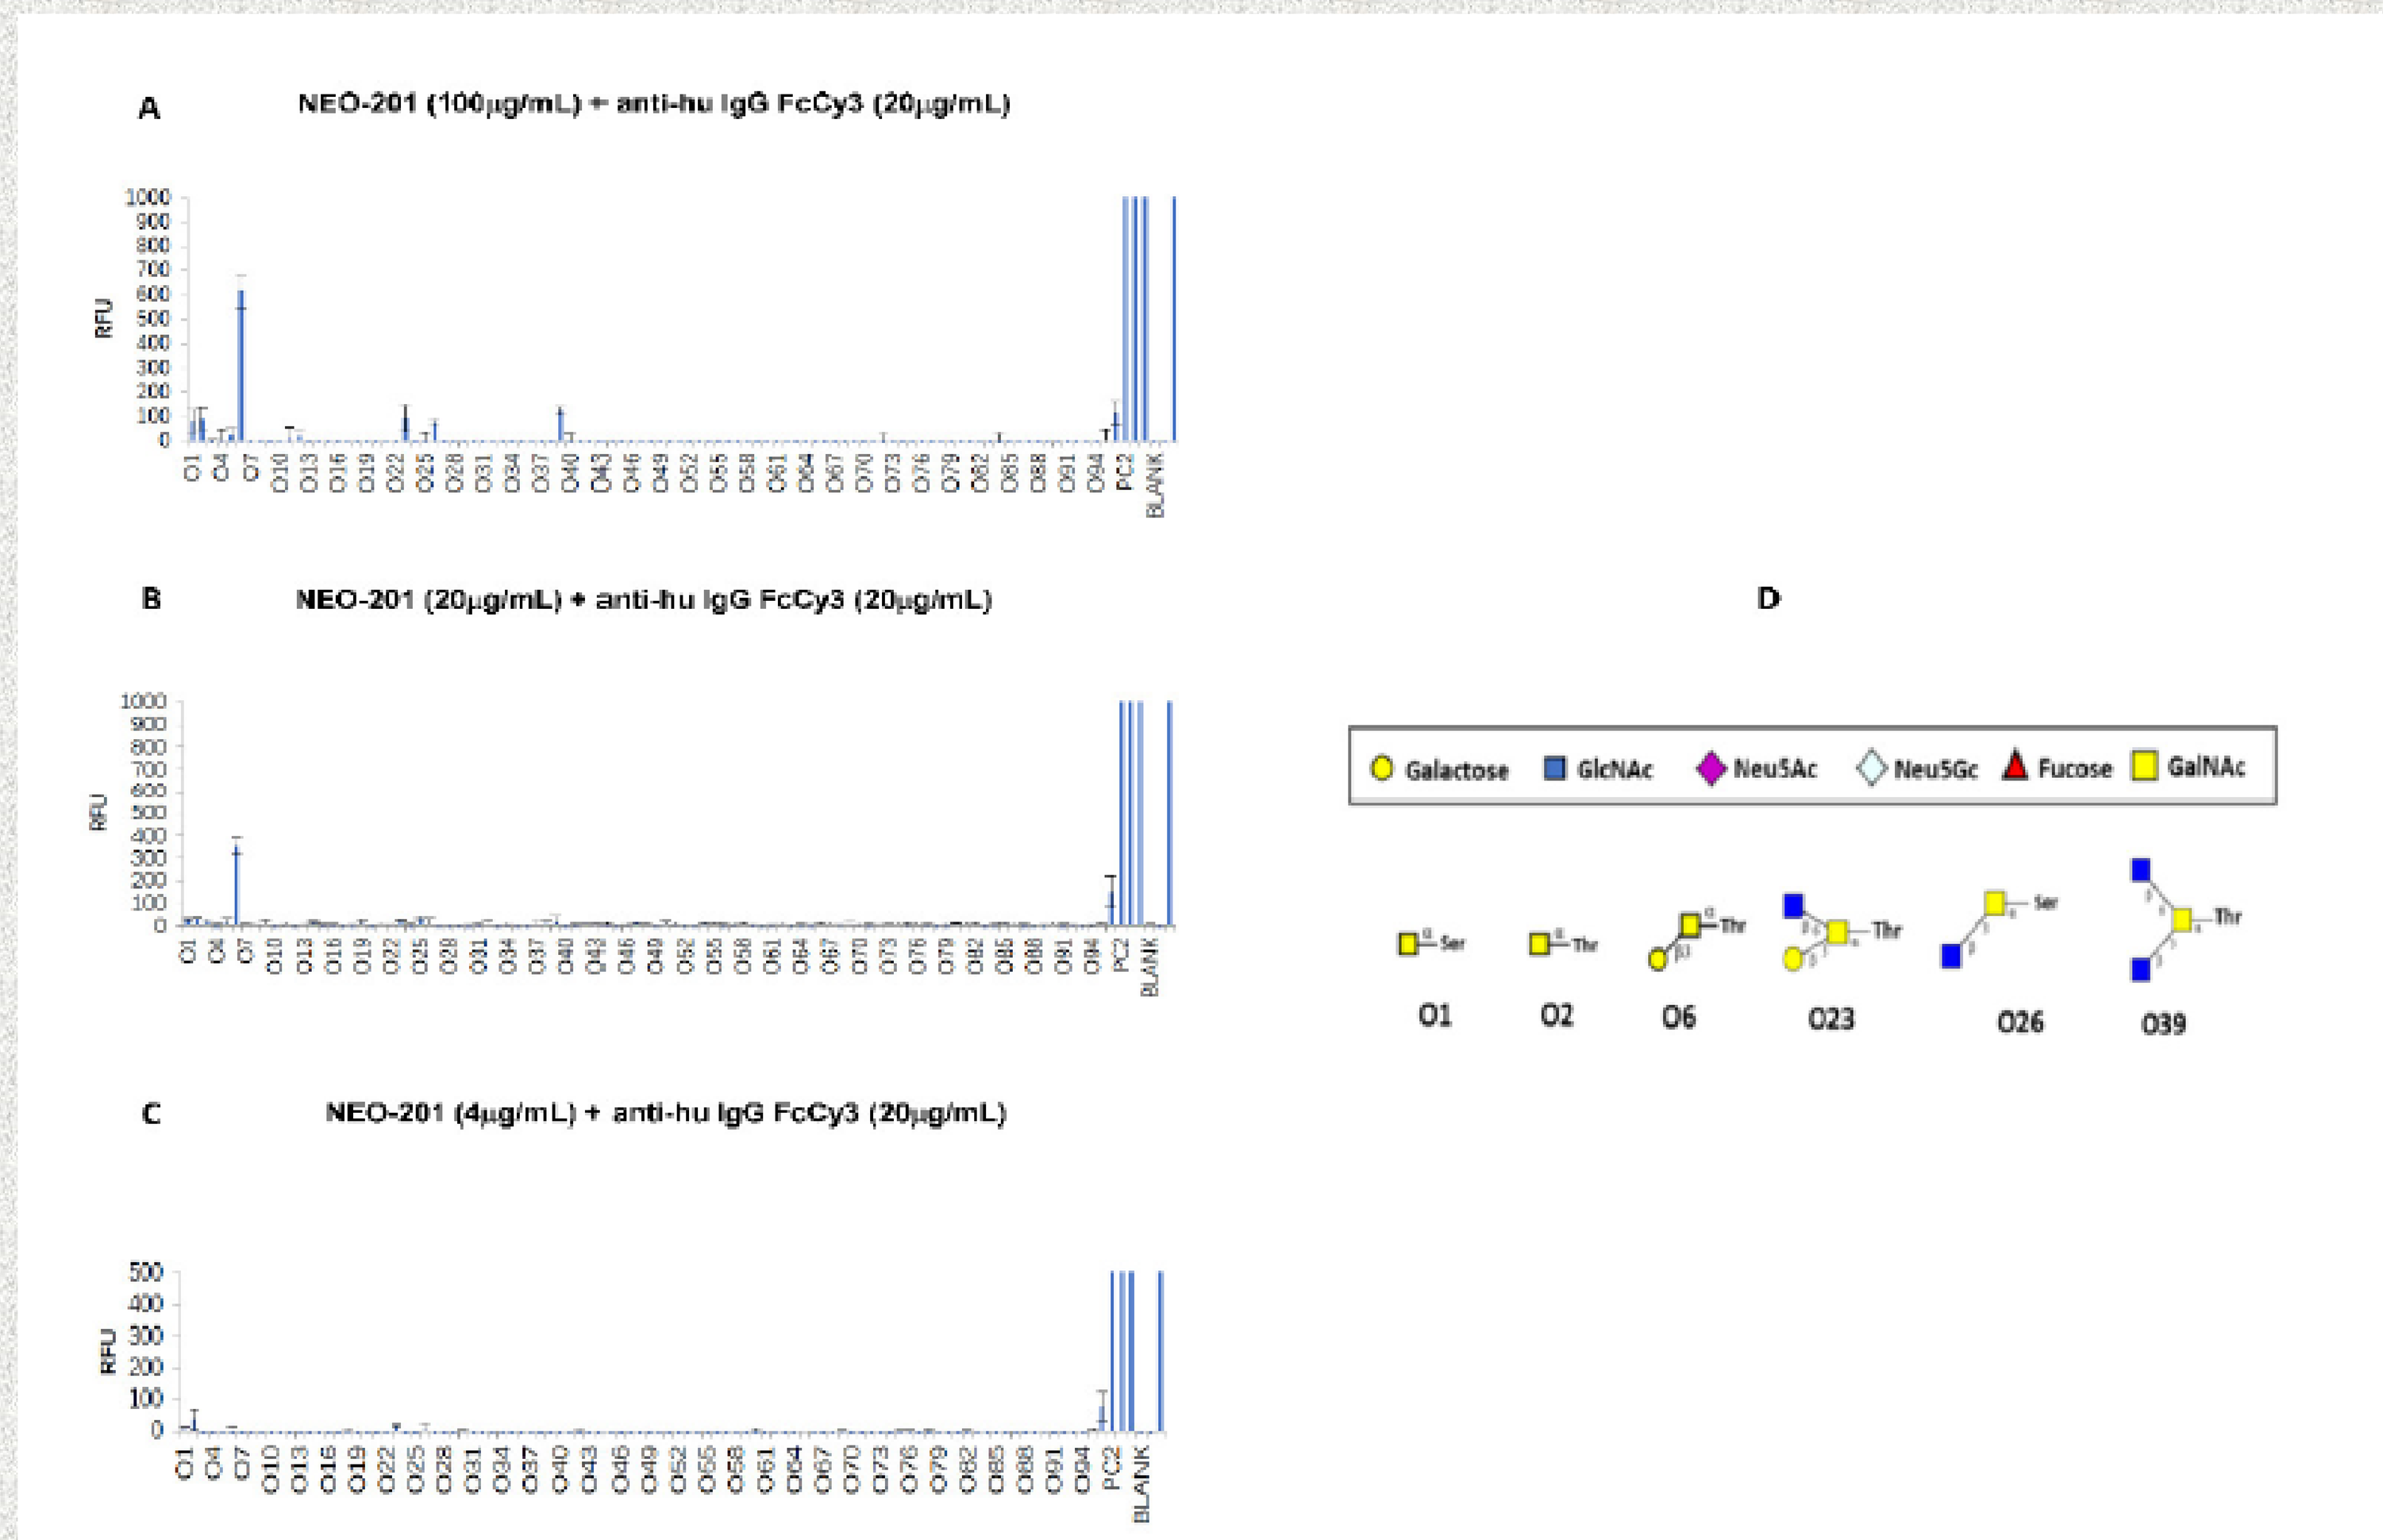

A
NEO-201 (100μg/mL) + anti-hu IgG FcCy3 (20μg/mL)
B
NEO-201 (20μg/mL) + anti-hu IgG FcCy3 (20μg/mL)
C
NEO-201 (4μg/mL) + anti-hu IgG FcCy3 (20μg/mL)
RFU
D
Galactose
GlcNAc
Neu5Ac
Neu5Gc
Fucose
GalNAc
01
02
06
023
026
039

01, 02, 06, 023, 026 and 039 O-glycans showed binding to NEO-201 in a dose-dependent manner. **The 06 binding interaction was the strongest of any observed**. 01 and 02 are Tn antigens. **06 is core 1**, 023 is core 2, 026 is core 3 and 039 is core 4.

5. Human Cancer Cell Lines and Human Neutrophils Recognized by NEO-201 Express Core 1 and Extended Core 1 O-Glycans

Proposed Structure	Relative Abundance (%)	O-Glycan Profile
CFPAC-1 (human pancreatic cancer cell line)		
	74.38%	Sialyl-T antigen (Sialyl core 1)
	13.92%	T antigen (core 1)
Human neutrophils		
	33.08%	Sialyl-T antigen (Sialyl core 1)
	24.65%	T antigen (core 1)
HL-60 (human acute myeloid leukemia cell line)		
	100%	Extended core 1
U937 (human acute myeloid leukemia cell line)		
	73.05%	Extended core 1
	12.10%	

Mannose, Man
Galactose, Gal
N-acetylgalactosamine, GalNAc
Glucose, Glc
N-acetylglucosamine, GlcNAc
Fucose, Fuc
N-acetylehexosamine, HexNac
N-acetyleneuraminic acid, Neu5Ac, NeuAc
N-glycolylneuraminic acid, Neu5Gc, NeuGc

: Galactose
: Fucose
: *N*-acetyl glucosamine (GlcNAc)
: Mannose
: *N*-glycolyl neuraminic acid (NeuGc)
: *N*-acetyl neuraminic acid (NeuAc)
: *N*-acetyl hexosamine (HexNAc, undefined stereochemistry)
: Hexose (Hex, undefined stereochemistry)
P : Phosphate (Phos)
S : Sulfate (Sulf)

Data are presented as the percentage of the most representative O-glycans. The legend below the table depicts monosaccharides contained in each O-glycan structure reported in the table..

5. NEO-201 Mediates ADCC to Kill Target Cells Expressing Core 1 and Extended Core 1 O-Glycans

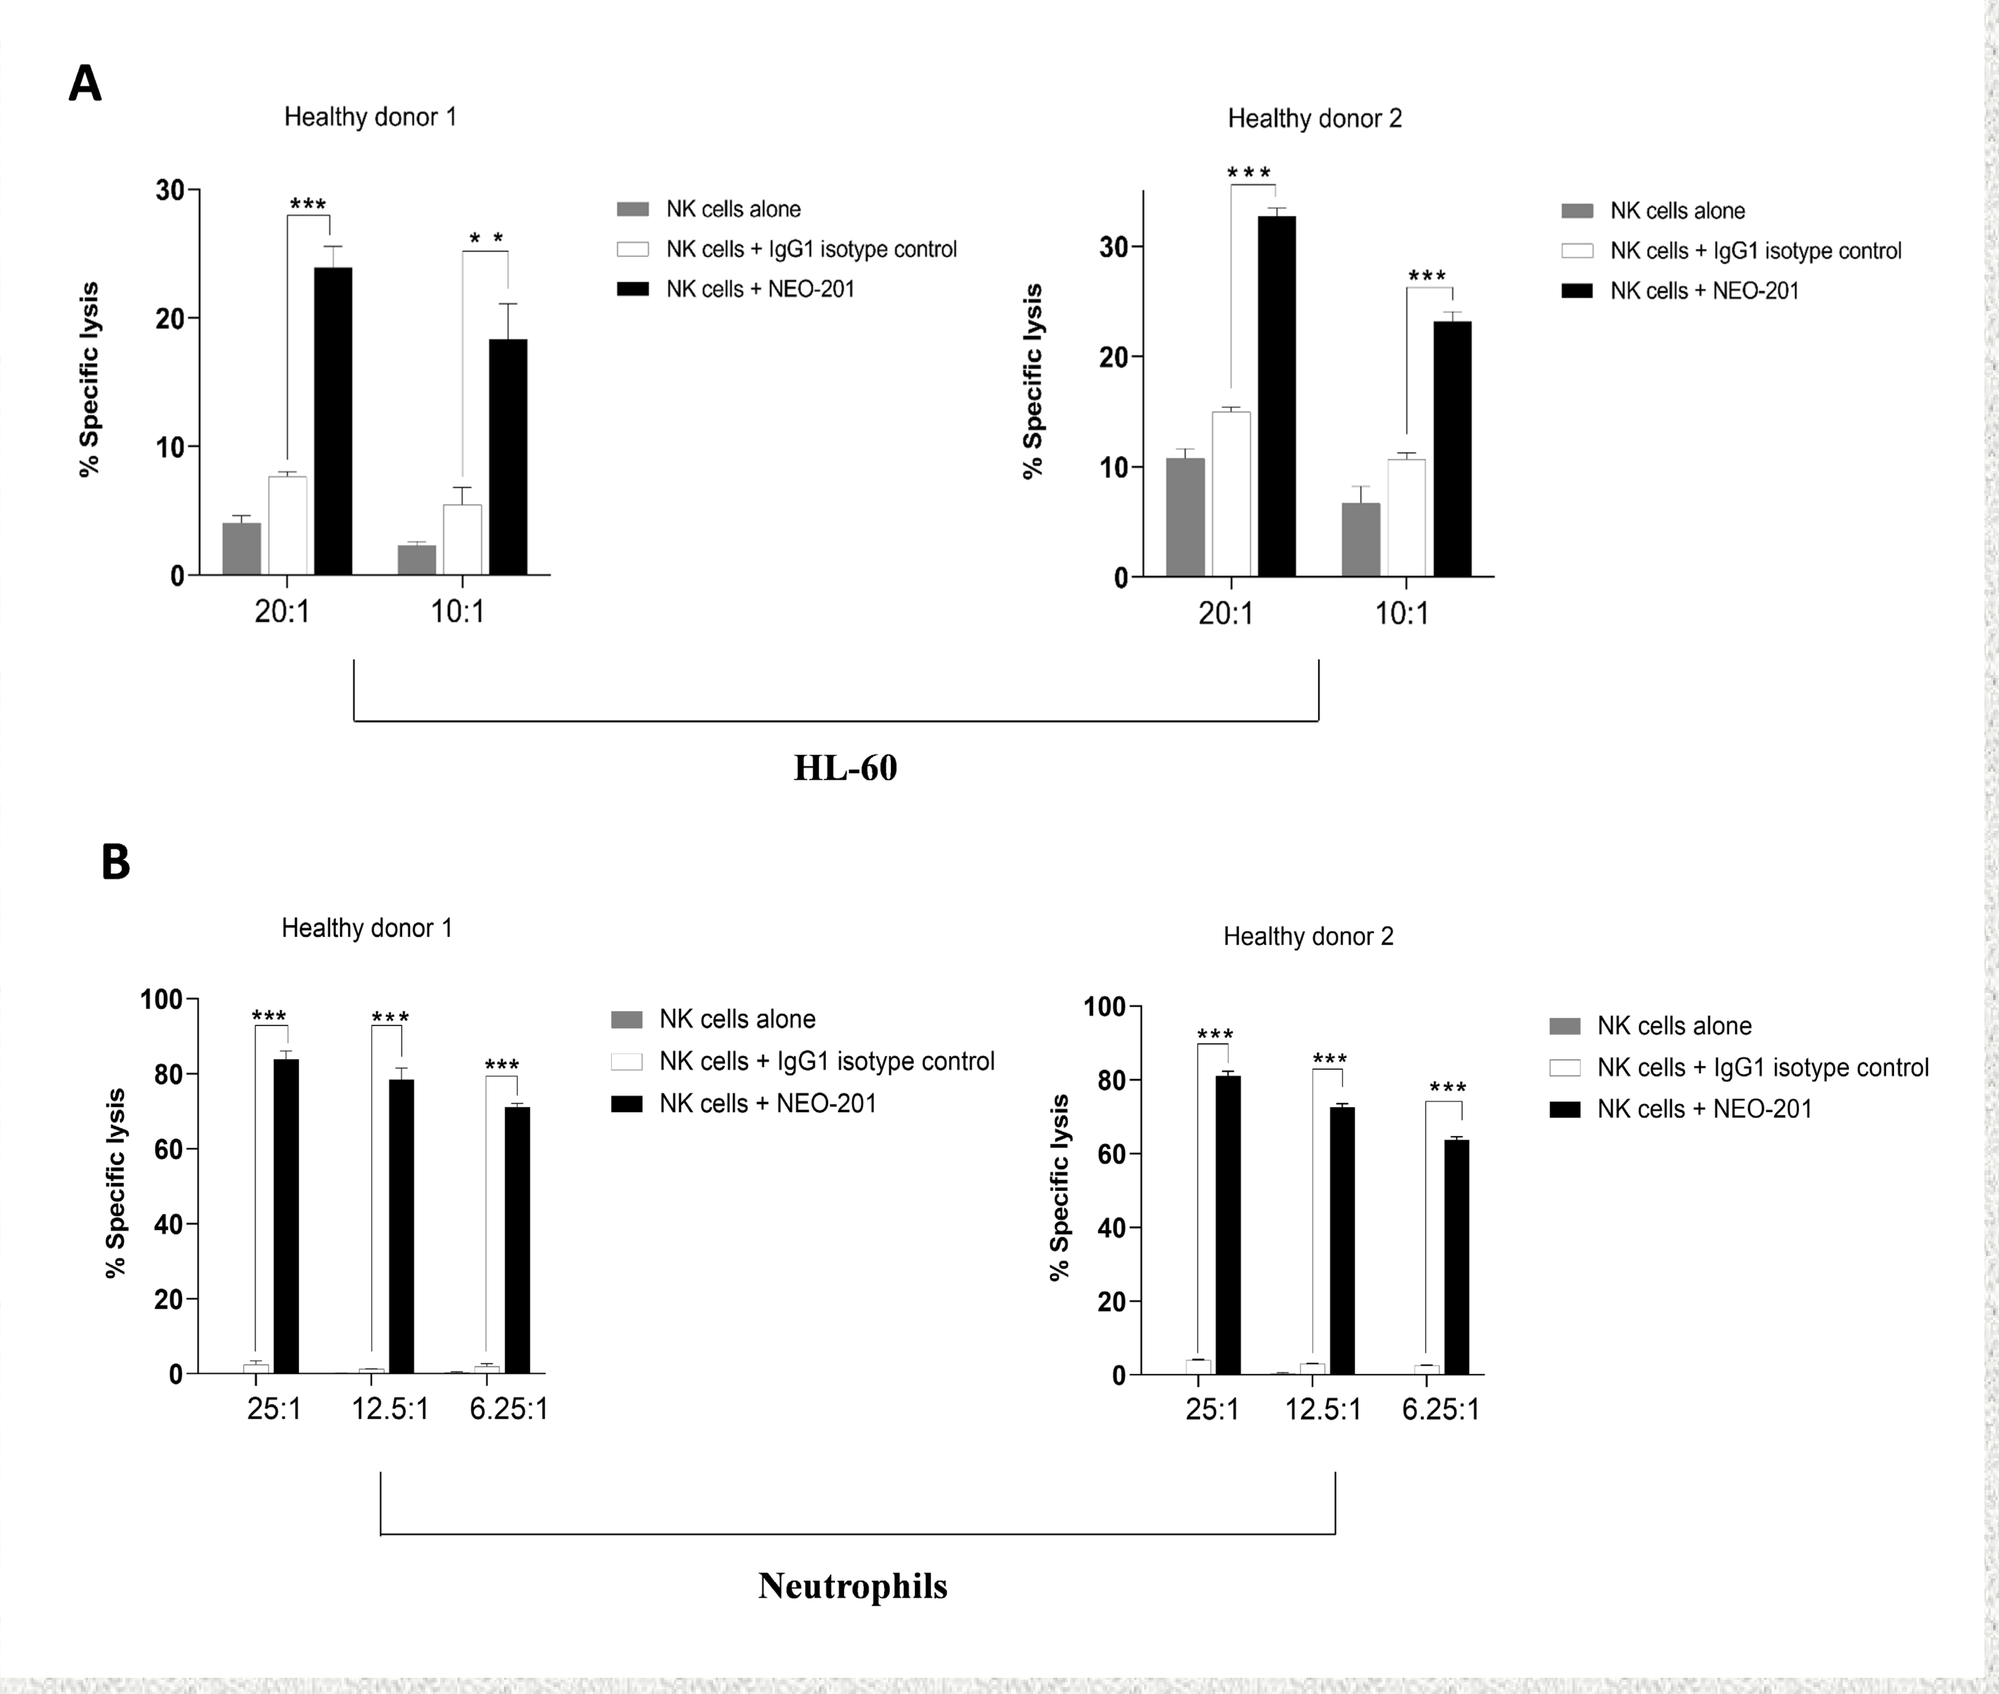

A
Healthy donor 1
Healthy donor 2
% Specific lysis
NK cells alone
NK cells + IgG1 isotype control
NK cells + NEO-201
20:1
10:1
HL-60
B
25:1
12.5:1
6.25:1
Neutrophils

The HL-60 cell line and human neutrophils have been used as target cells to evaluate the ADCC mediated by NEO-201 in the ADCC assay.

Cells were incubated with 10 μg/mL of NEO-201 or human IgG1 (negative control).

NK cells from two healthy donors were used as effector cells at the indicated E:T ratios.

Results are presented as the mean of % specific lysis ± SD (standard deviation) from three replicate wells in each experiment.

*** statistically significant ($p < 0.001$) by two-way ANOVA (NEO-201 + NK cells vs. IgG1+ NK cells).
** statistically significant ($p < 0.01$) by two-way ANOVA (NEO-201 + NK cells vs. IgG1+ NK cells).

Conclusion

This study demonstrated that NEO-201 recognizes core 1 and extended core 1 O-glycans expressed on NEO-201-reactive cells such as the pancreatic cancer cell line CFPAC-1, human neutrophils and the human hematological neoplastic cells HL60 and U937. These cells can be killed by NEO-201 through NK-mediated ADCC.
The aberrant expression of incomplete/truncated O-glycans in cancer cells occurs in both solid tumors and hematologic malignancies and is correlated with a poor prognosis and tumor progression. The employment of mAbs targeting truncated O-glycans in cancer cells, such as NEO-201, could represent a novel and promising immunotherapy approach for treating both solid and liquid tumors.