Reduction of circulating naïve Tregs and gMDSCs and low levels of soluble MICA are prognostic for efficacy of combined NEO-201 and pembrolizumab

Jamie Hur[1], Massimo Fantini[2], Kwong Y. Tsang[2], Kevin Conlon[1], Charalampos Floudas[3], Azam Ghafoor[4], Anjum Zaki[2], Sharon A. Mavroukakis[2], Ann McCoy[1], Chris Feierabend[4], Erica Redmond[3], Philip M. Arlen[2], Christina M. Annunziata[1]

[1]Women's Malignancies Branch, Center for Cancer Research, National Cancer Institute, National Institutes of Health, Bethesda, MD, USA; [2]Precision Biologics, Inc., Bethesda, MD, USA; [3]Center for Immuno-Oncology, Center for Cancer Research, National Cancer Institute, National Institutes of Health, Bethesda, MD, USA; [4]Thoracic and GI Malignancies Branch, Center for Cancer Research, National Cancer Institute, National Institutes of Health, Bethesda, MD, USA.

Abstract # 2530

Background

The humanized IgG1 monoclonal antibody NEO-201 binds to Core 1 and/or extended Core 1 O-glycans expressed by several human solid and blood tumors, as well as neutrophils, and mediates killing of cancer cells, neutrophils, regulatory T cells (Tregs) and granulocytic myeloid-derived suppressor cells (gMDSCs) via ADCC and CDC. Resistance to PD-1/PD-L1 blockade may be due to the accumulation of Tregs and gMDSCs in the tumor microenvironment (TME). NEO-201 was proven to bind and reduce the amount of circulating Tregs in cancer patients. This supports the rationale of the ongoing phase II clinical trial (NCT03476681) evaluating the activity of NEO-201 with pembrolizumab in adults with solid tumors resistant to prior checkpoint inhibitors. Furthermore, elevated serum levels of soluble MICA (sMICA) have been correlated with decreased NK cell activity and progression of several types of solid tumors.

Methods

Subjects with recurrent, locally advanced unresectable or metastatic Non-Small Cell Lung Cancer (NSCLC), Cervical Cancer, Head and Neck Squamous Cell Carcinoma (HNSCC), Uterine Carcinoma who have progressed during or after front-line standard of care treatment and have tumors resistant to checkpoint inhibitors are being enrolled in the ongoing phase II clinical trial (NCT03476681) and treated each cycle with 3 doses of NEO-201 1.5mg/kg every 2 weeks and pembrolizumab 400mg IV every 6 weeks and imaged for response every 2 cycles. PBMCs from cancer patients pre- and at multiple time points post-treatment, including C1D15 (14 days of the first infusion with NEO-201), C2D1 PRE (beginning of cycle 2), C3D1 PRE (beginning of cycle 3), were profiled by flow cytometry to evaluate if the modulation of the percentage of circulating gMDSCs and Tregs after treatment was correlated with clinical response. Serum levels of sMICA were compared pre- and and at multiple time points post-treatment by ELISA to evaluate if high serum levels of sMICA were correlated with a poor clinical response.

Schema of Treatment: Expansion Cohorts

Table 1: Enrollment to two-stage design per expansion cohorts

Disease Cohort	Initial Stage	Acceptable Response Rate % (N/Total)	Second Stage	Sufficiently interesting Response Rate
NSCLC	12	20% (≥1/12)	21	14.3% (≥3/21)
HNSCC	16	25% (≥2/16)	31	19.4% (≥6/31)
Endometrial	16	25% (≥2/16)	31	19.4% (≥6/31)
Cervical	13	25% (≥1/13)	20	15% (≥3/20)

The sample size for the expansion cohorts are calculated based on the desirable ORR (Simon minimax two-stage phase II design) for each cohort.

Results

Reduction of quantity of circulating naïve Tregs post treatment is associated with stabilization of the disease

Flow cytometry analysis of PBMCs revealed that NEO-201 recognizes circulating naïve Tregs (**nTregs: $CD3^+/CD4^+/CD45RA^+/Foxp3^{low}$ cells**) but not circulating effector Tregs (**eTregs: $CD3^+/CD4^+/CD45RA^-/Foxp3^{high}$ cells**). Based on this observation, we compared the percentage of nTregs population from PBMCs in three cancer patients with stable disease (SD) and three cancer patients with progressive disease (PD) pre and post treatment, and we correlated the modulation of levels of circulating naïve Tregs with the clinical response.

General downtrend of circulating naïve Tregs in patients with SD

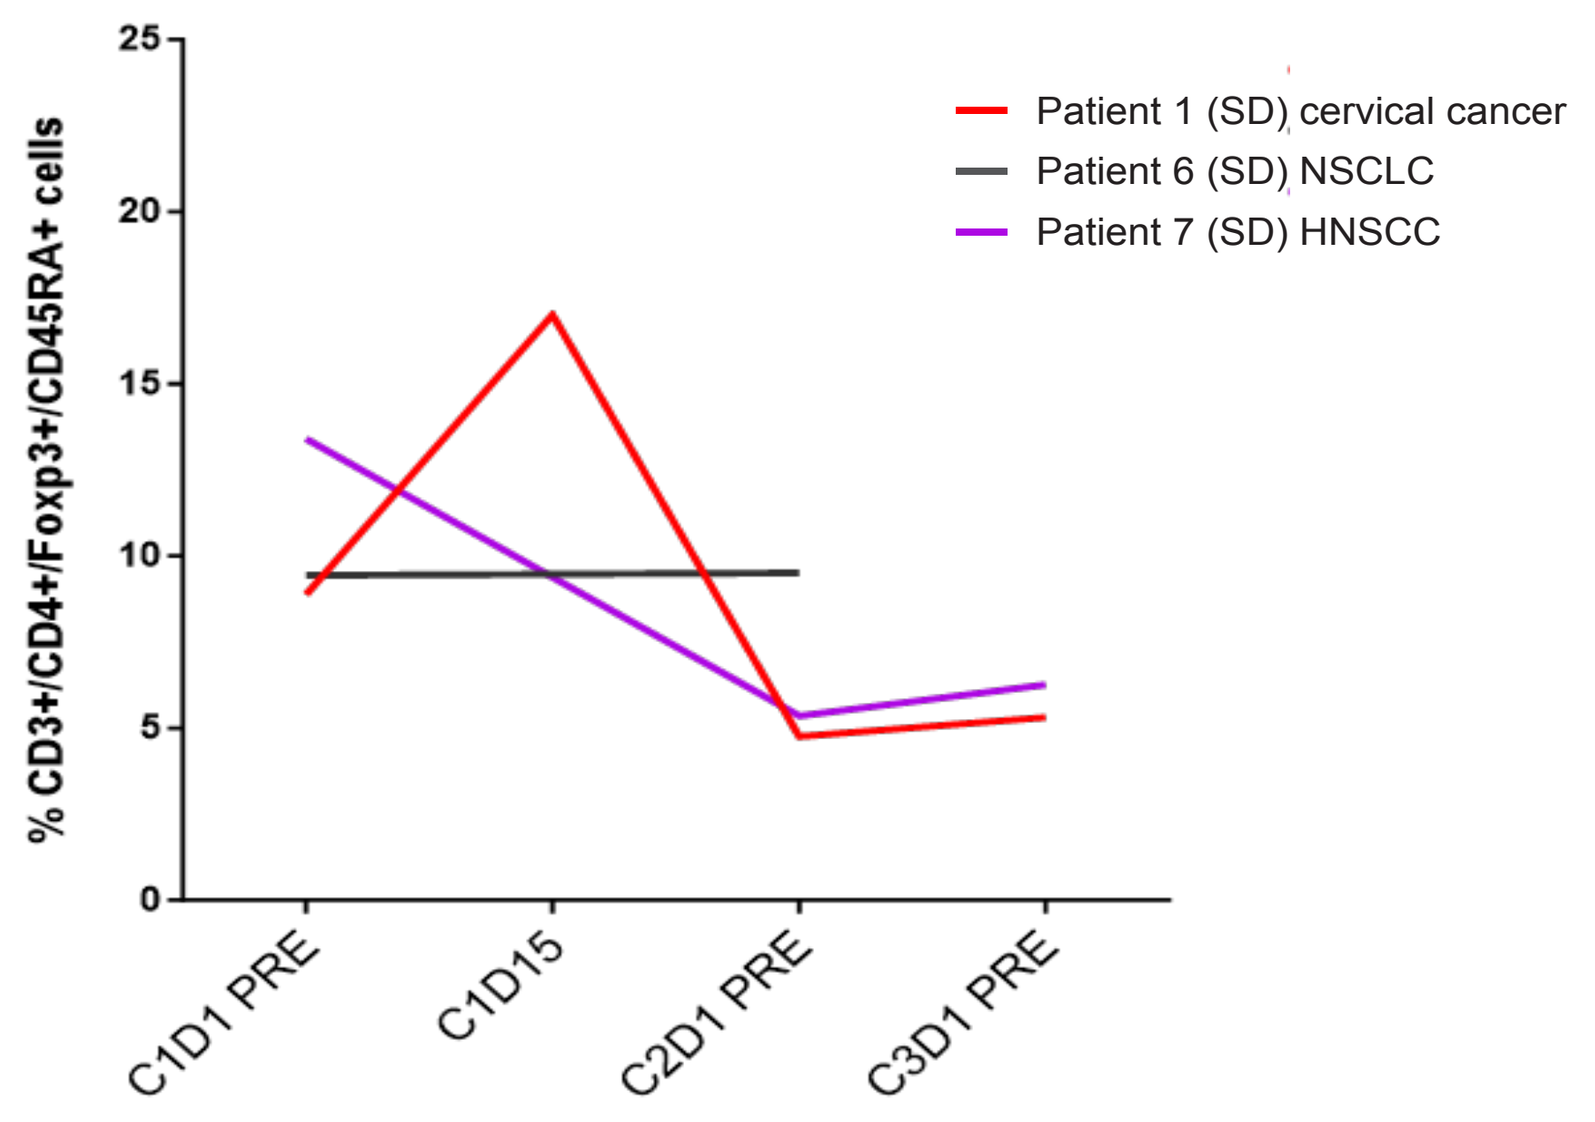
Patient 1 (SD) cervical cancer
Patient 6 (SD) NSCLC
Patient 7 (SD) HNSCC
% CD3+/CD4+/Foxp3+/CD45RA+ cells
C1D1 PRE
C1D15
C2D1 PRE
C3D1 PRE

General uptreand of circulating naïve Tregs in patients with PD

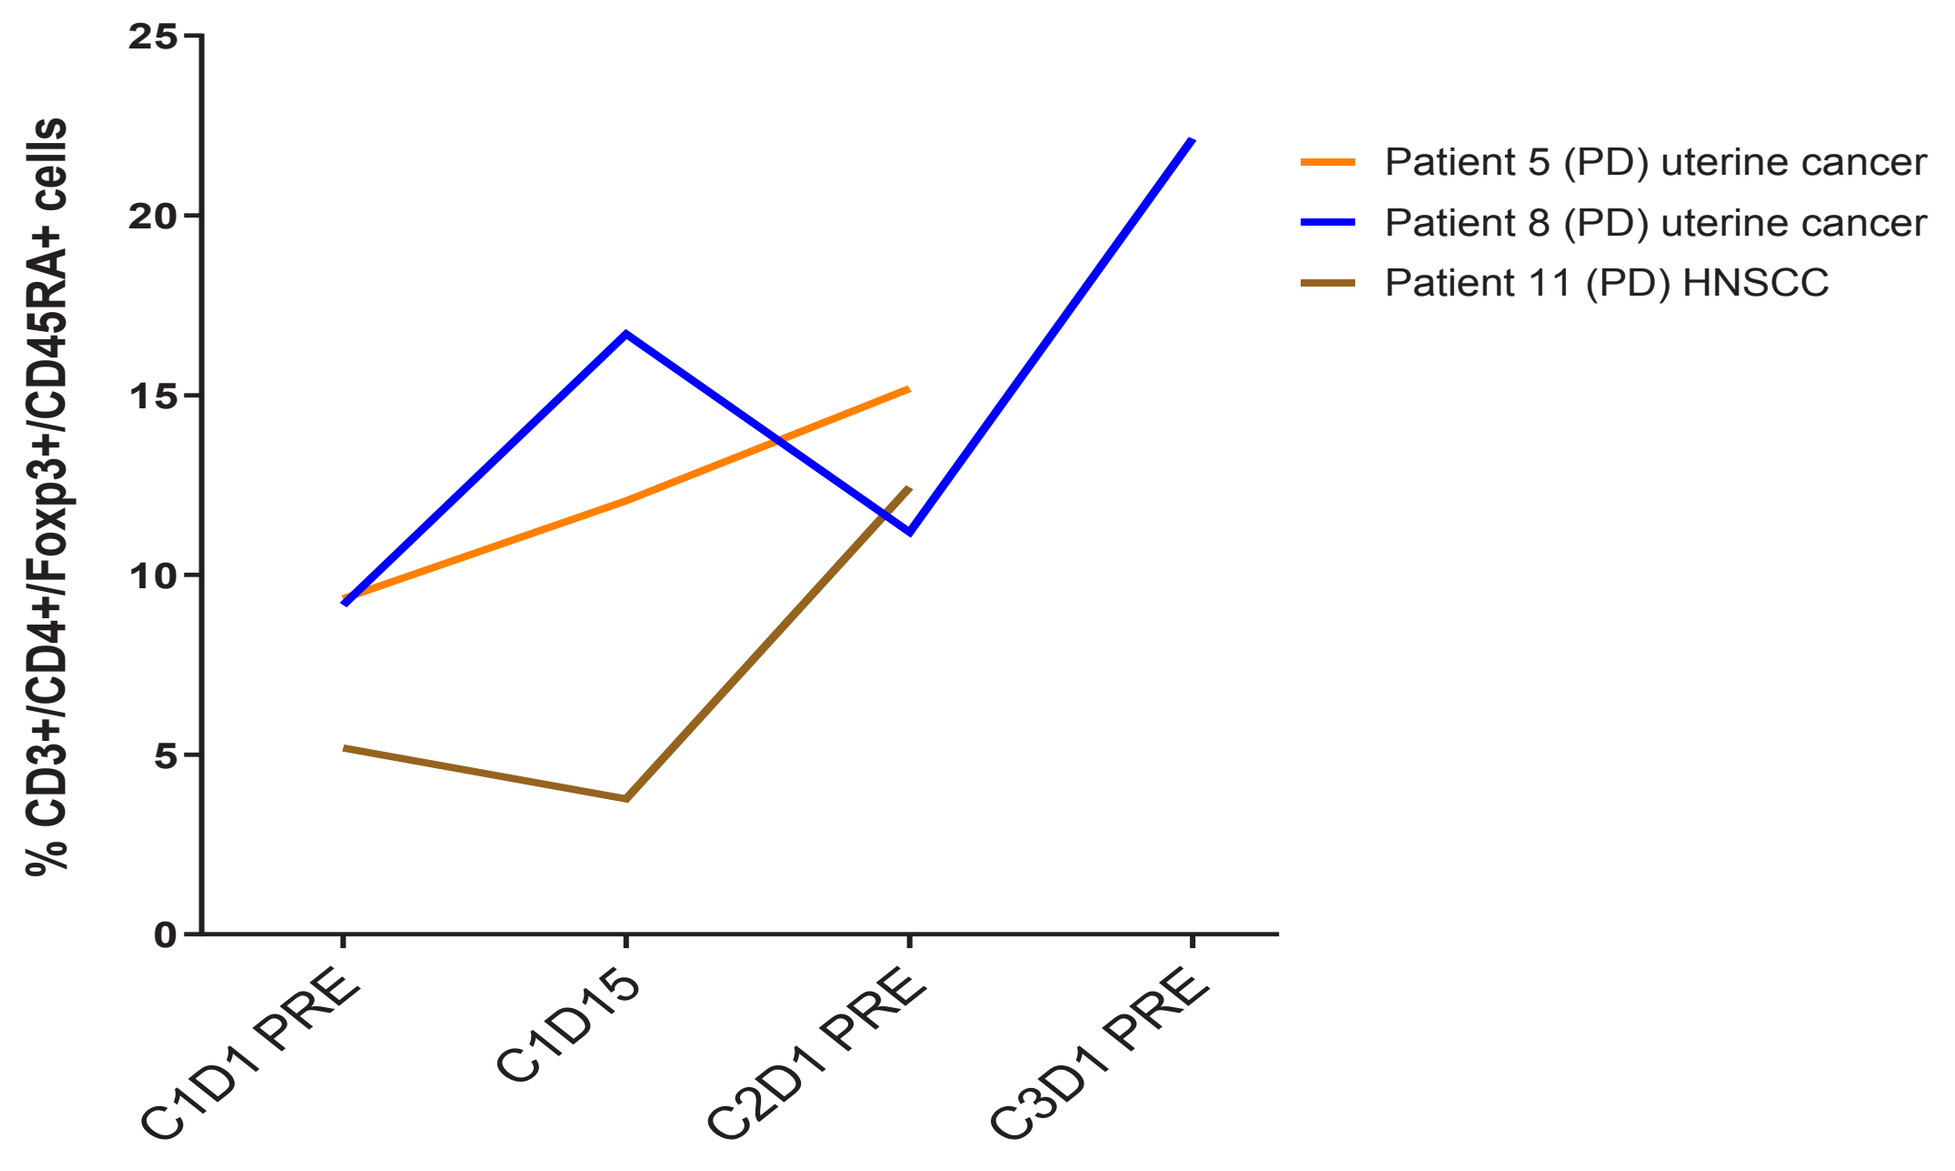
Patient 5 (PD) uterine cancer
Patient 8 (PD) uterine cancer
Patient 11 (PD) HNSCC
% CD3+/CD4+/Foxp3+/CD45RA+ cells
C1D1 PRE
C1D15
C2D1 PRE
C3D1 PRE

Reduction of quantity of circulating gMDSCs post treatment is associated with durable stabilization of the disease

Flow cytometry analysis of PBMCs revealed that NEO-201 recognizes circulating gMDSCs (defined as **$CD33^+/HLA\text{-}DR^{neg}/CD15^+/CD14^{neg}/CD66b^+$ cells**). Based on this observation, we compared the percentage of gMDSCs from PBMCs of three cancer patients with stable disease (SD) and four cancer patients with progressive disease (PD) pre and post treatment. Then, we correlated the modulation of levels of circulating gMDSCs with the clinical response.

General downtrend of circulating gMDSCs in patients with SD

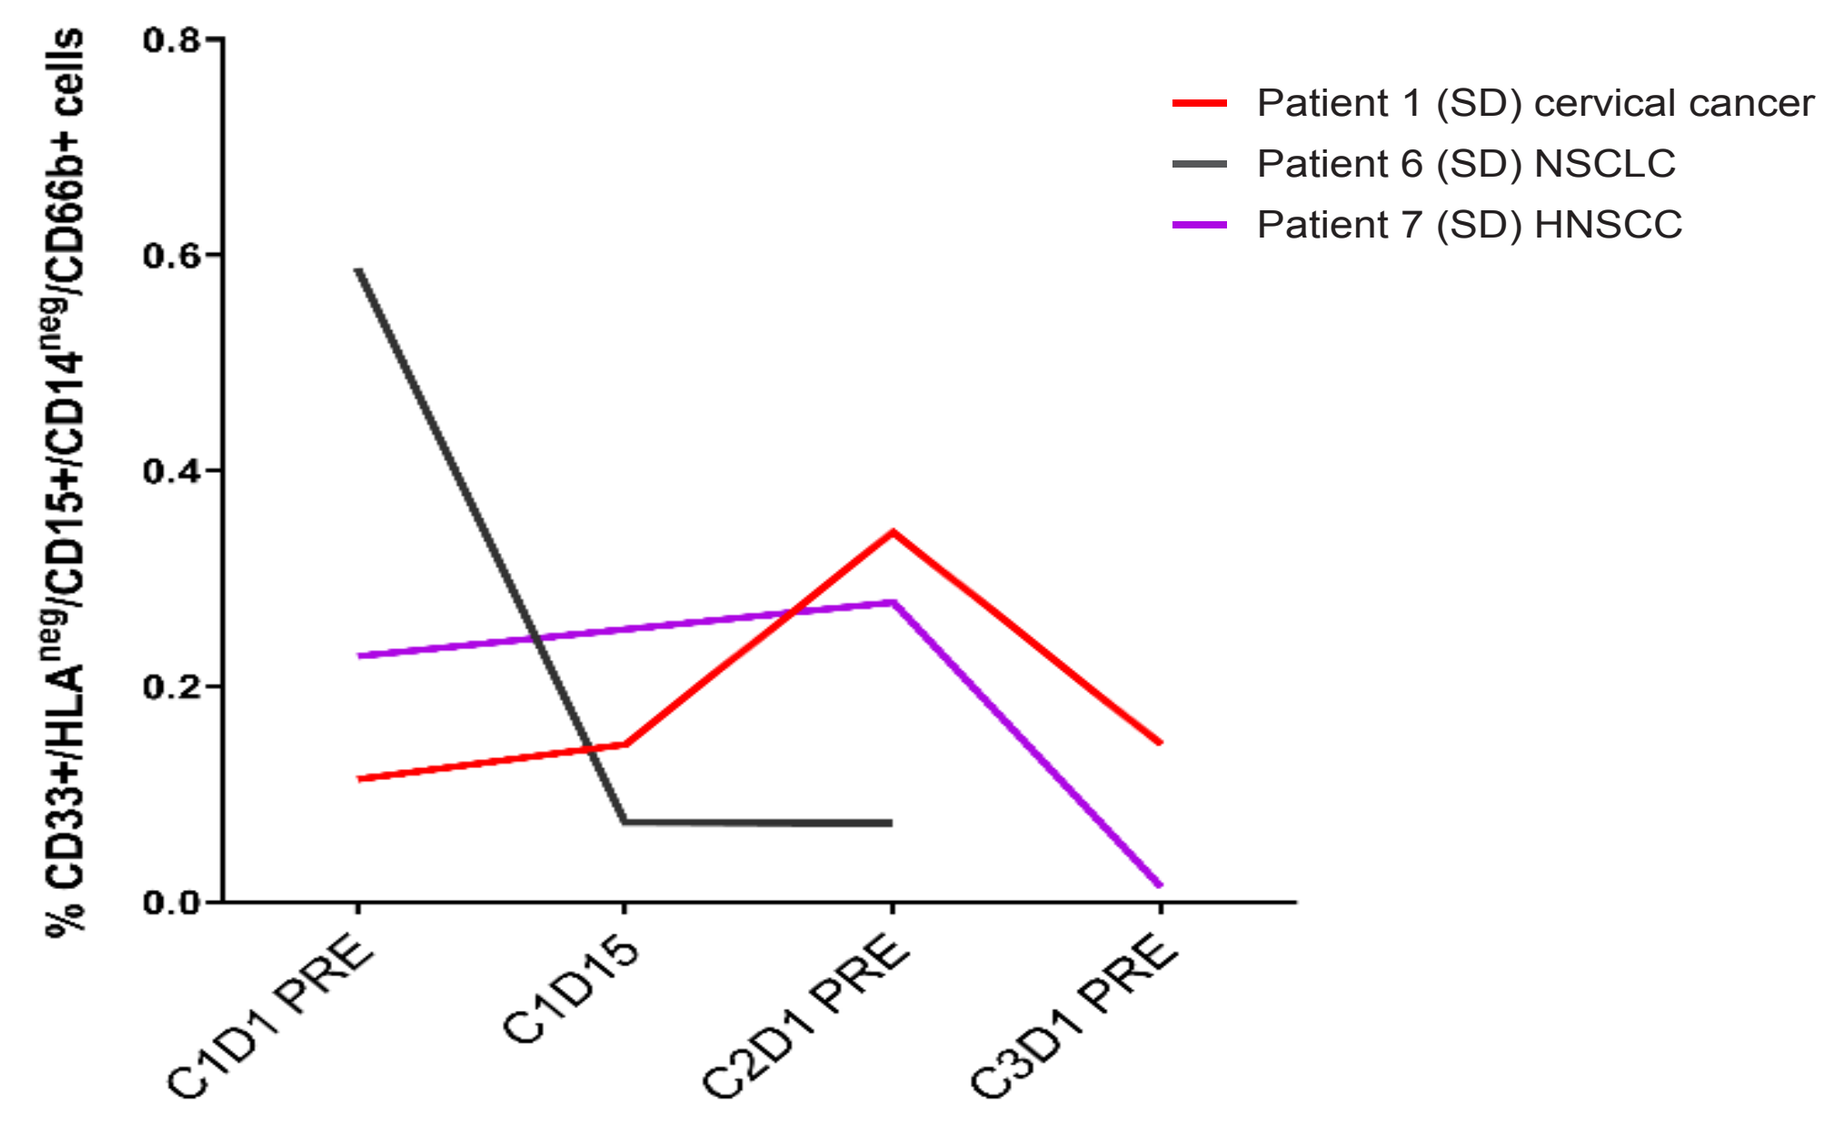
Patient 1 (SD) cervical cancer
Patient 6 (SD) NSCLC
Patient 7 (SD) HNSCC
% CD33+/HLAneg/CD15+/CD14neg/CD66b+ cells
C1D1 PRE
C1D15
C2D1 PRE
C3D1 PRE

General uptreand of circulating gMDSCs in patients with PD

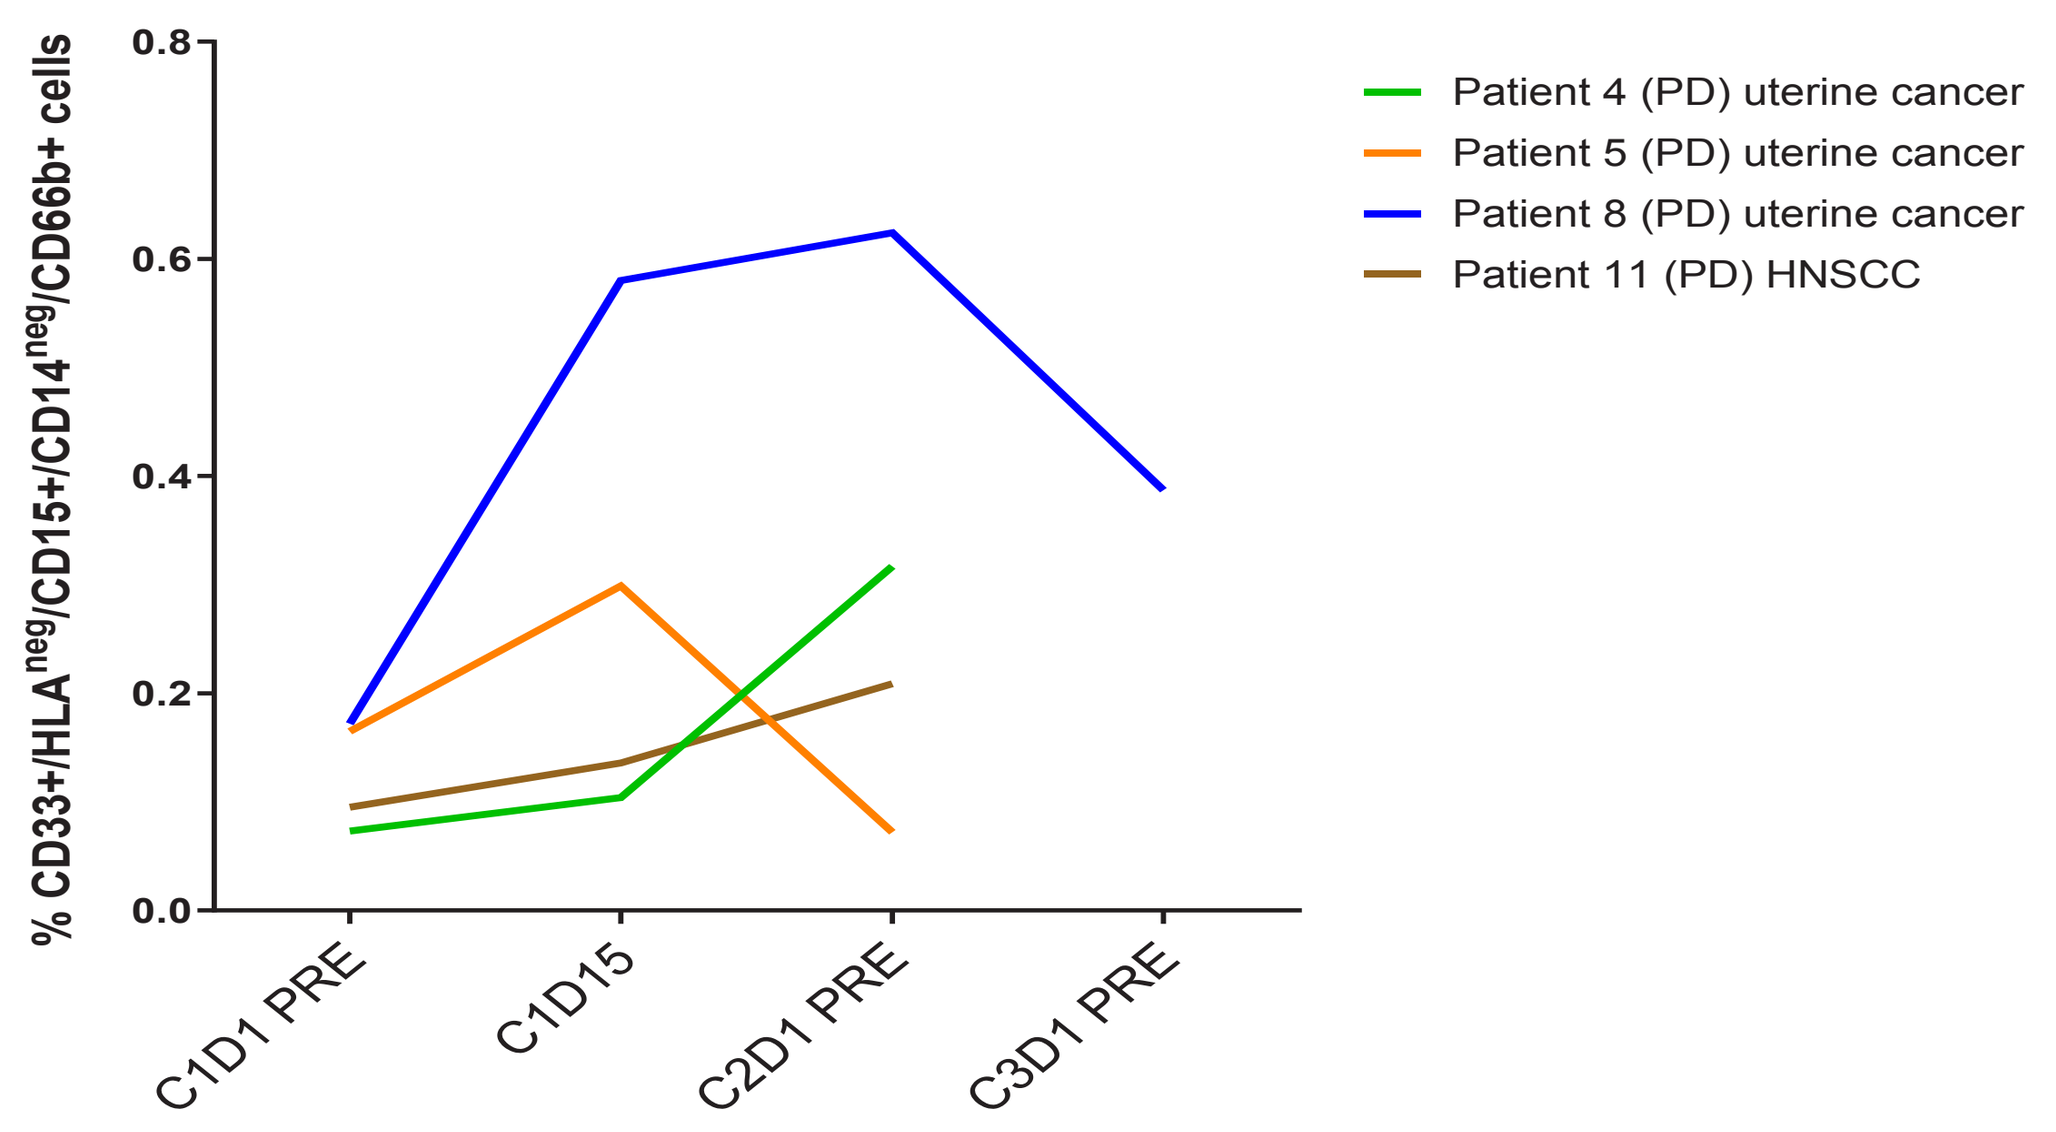
Patient 4 (PD) uterine cancer
Patient 5 (PD) uterine cancer
Patient 8 (PD) uterine cancer
Patient 11 (PD) HNSCC
% CD33+/HLAneg/CD15+/CD14neg/CD66b+ cells
C1D1 PRE
C1D15
C2D1 PRE
C3D1 PRE

Patients with durable SD: a patient with HNSCC (SD >11 months) showed >50% reduction of nTregs and >90% of gMDSCs at C3D1 (beginning of cycle 3) compared to baseline (C1D1 PRE). One patient with cervical cancer (SD >8 months) showed 40% reduction of nTregs at C3D1 compared to baseline, while gMDSCs trended down to baseline levels at C3D1 after initial increase. The patient with NSCLC showed a marked reduction of gMDSCs (87.42%) starting at C1D15 compared to baseline, while no difference in percentage of nTregs were observed between baseline and post-treatment.

Cancer patients with SD had lower median serum levels of sMICA compared to cancer patients with PD

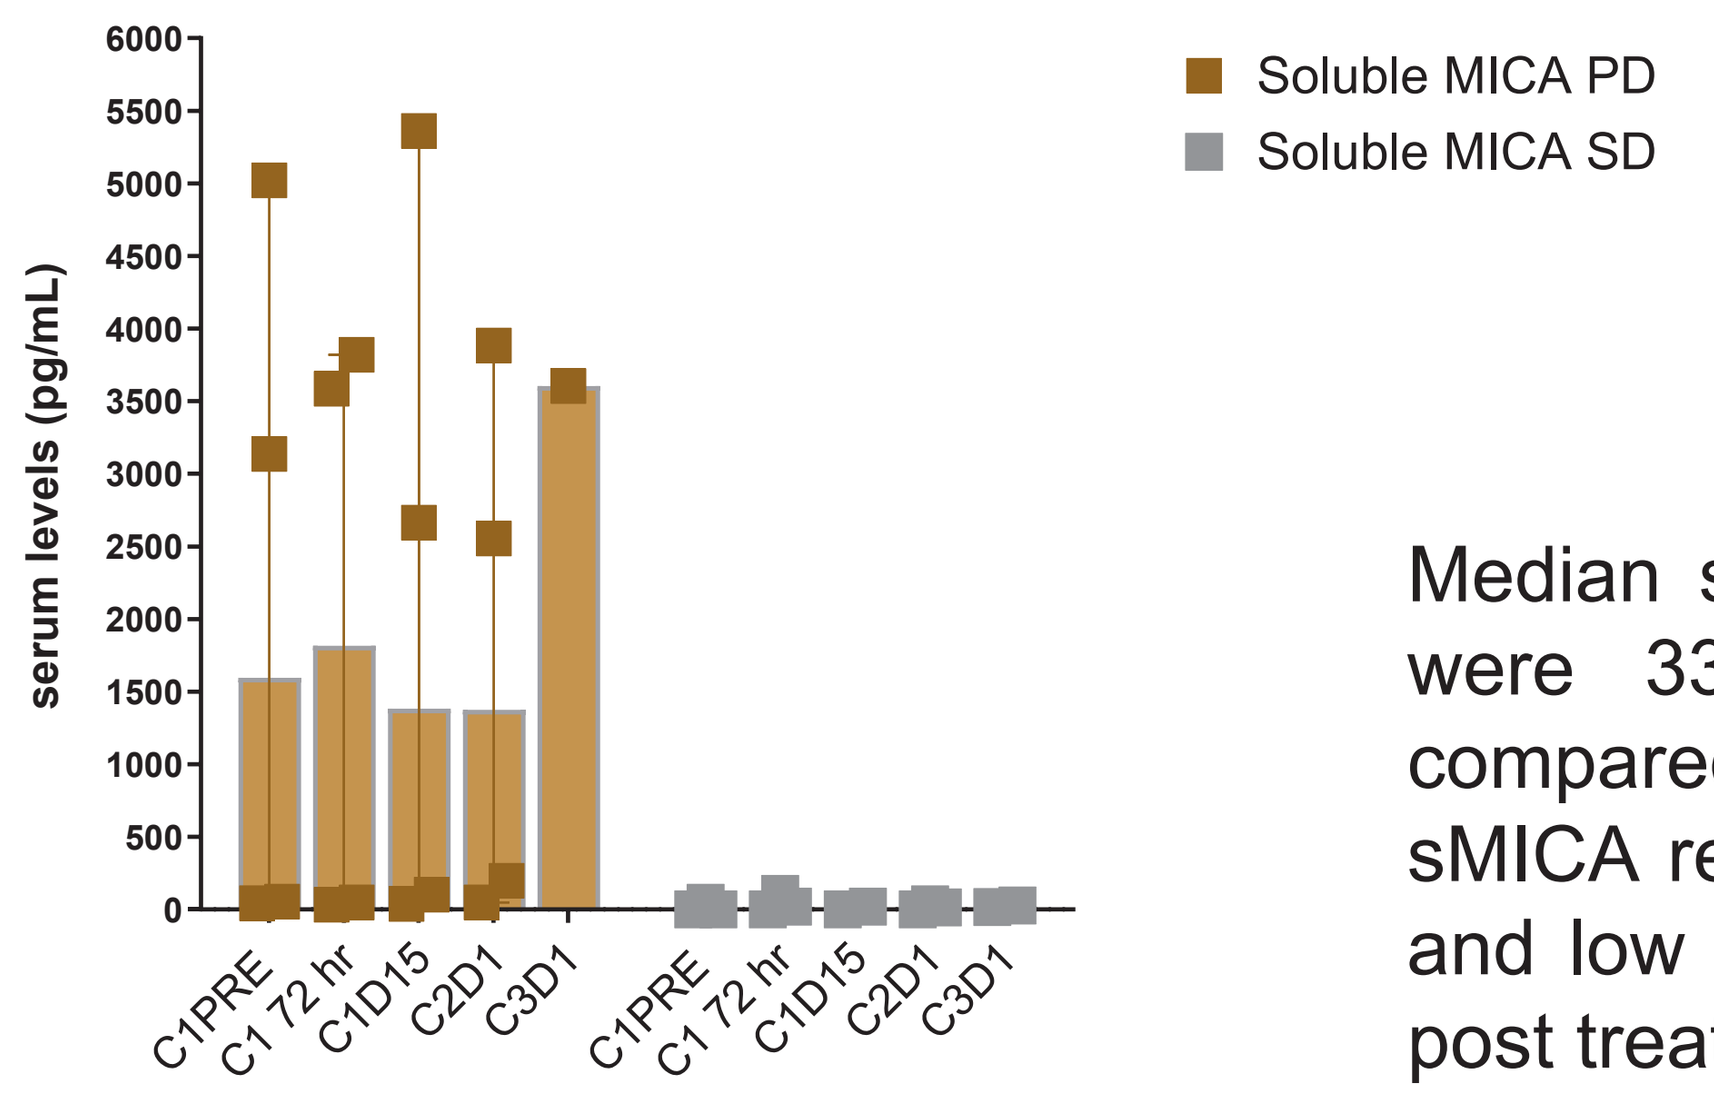
Soluble MICA PD
Soluble MICA SD
serum levels (pg/mL)
C1PRE
C1 72 hr
C1D15
C2D1
C3D1

Comparison of median serum levels of sMICA between three cancer patients with SD and four cancer patients with PD.

Median serum levels of sMICA pre-treatment were 33-fold higher in patients with PD compared to patients with SD. Levels of sMICA remained elevated in patients with PD and low in patients with SD at all time points post treatment

Characteristics of cancer patients with SD

Cervical cancer patient:

- 50 years old with chemo-resistant metastatic mucinous carcinoma.
- 80% tumor tissue positive for NEO-201 staining in IHC with 3+ intensity.
- Stable disease for more than 8 months after first infusion with both antibodies
- Patient qualified for debulking surgery requiring exiting the study

Head and neck squamous cell carcinoma patient:

- 72 years old with moderately differentiated carcinoma.
- 10-15% tumor tissue positive for NEO-201 staining in IHC with 3+ intensity.
- Stable disease for more than 11 months after first infusion with both antibodies

NSCLC patient:

- 69 years old with metastatic lung adenocarcinoma
- 90% tumor positive for NEO-201 staining in IHC with 3+ intensity
- Stable disease by RECIST on Day 84.
- Patient was taken off study per treating physician's decision.

Conclusions

Depletion of circulating Tregs and gMDSCs may prevent their accumulation in the TME and enhance the efficacy of pembrolizumab in subjects with tumors resistant to checkpoint inhibitors. The decrease in circulating naïve Tregs and gMDSCs after treatment with NEO-201 and pembrolizumab was associated with durable SD. High levels of sMICA can impair ADCC mediated by NEO-201, resulting in poor clinical response. Low levels of sMICA in patients with SD, together with the reduction of both circulating nTregs and gMDSCs, could be favorable prognostic markers for clinical benefit following treatment with NEO-201 and pembrolizumab. Ongoing enrollment in this clinical trial will validate these findings in larger cohorts.