Abstract #:1101

Affinity Maturation and Characterization of a Novel O-glycan Epitope Targeting Anti-Human Carcinoma Monoclonal Antibody (mAb) PB-223

Kwong Y. Tsang, Massimo Fantini, Anjum Zaki, Sharon A Mavroukakis and Philip M. Arlen

Precision Biologics, Inc. Bethesda, MD 20814

Introduction

Tumor-targeting mAbs can be leveraged to stimulate innate anti-cancer immunity. NEO-102 (Ensituximab) is a chimeric human IgG1 mAb targeting a glycosylated variant of MUC5AC with specificity to colorectal and pancreatic cancer. In a phase 2 study, NEO-102 showed promising results in pretreated patients with advanced, refractory colorectal cancer (Kim R. et al. Clin Cancer Res 2020). Enhancing binding affinity stands as a prominent avenue within the realm of antibody engineering, offering the potential to significantly augment the therapeutic efficacy of antibodies. Thus, the objective of this investigation is to enhance the binding affinity of NEO-102.

Experimental Design

This research entails the engineering of the VH and VL sequences of NEO-102 through Fast Screening for Expression Biophysical Properties and Affinity, with the aim to maintain the binding to target antigen while achieving a lower KD. To this end, we constructed a library for saturation mutagenesis into all residues in the VH and VL region of the antibody. Flow cytometry analysis was used to compare the binding affinity between the NEO-102 and the affinity maturation generated clone (AHF18095)(PB-223) to various tumor cell lines. O-glycan microarray was utilized to identify the O-glycan binding epitope of the PB-223. IHC tumor tissue microarrays, and antibody internalization were performed on PB-223.

RESEARCH POSTER PRESENTATION DESIGN © 2019
www.PosterPresentations.com

Results

1. Binding kinetics of antigen to antibodies by ELISA

Antibody	Antigen	Ka (1/Ms)	Kd (1/s)	KD (M)
NEO-102	BSM	9.63×10^{4}	5.40×10^{-4}	5.60×10^{-9}
PB-223	BSM	1.59×10^{5}	1.95×10^{-4}	1.23×10^{-9}

BSM=Bovine submaxillary mucin

2. PB-223 does not bind to healthy tissues by IHC

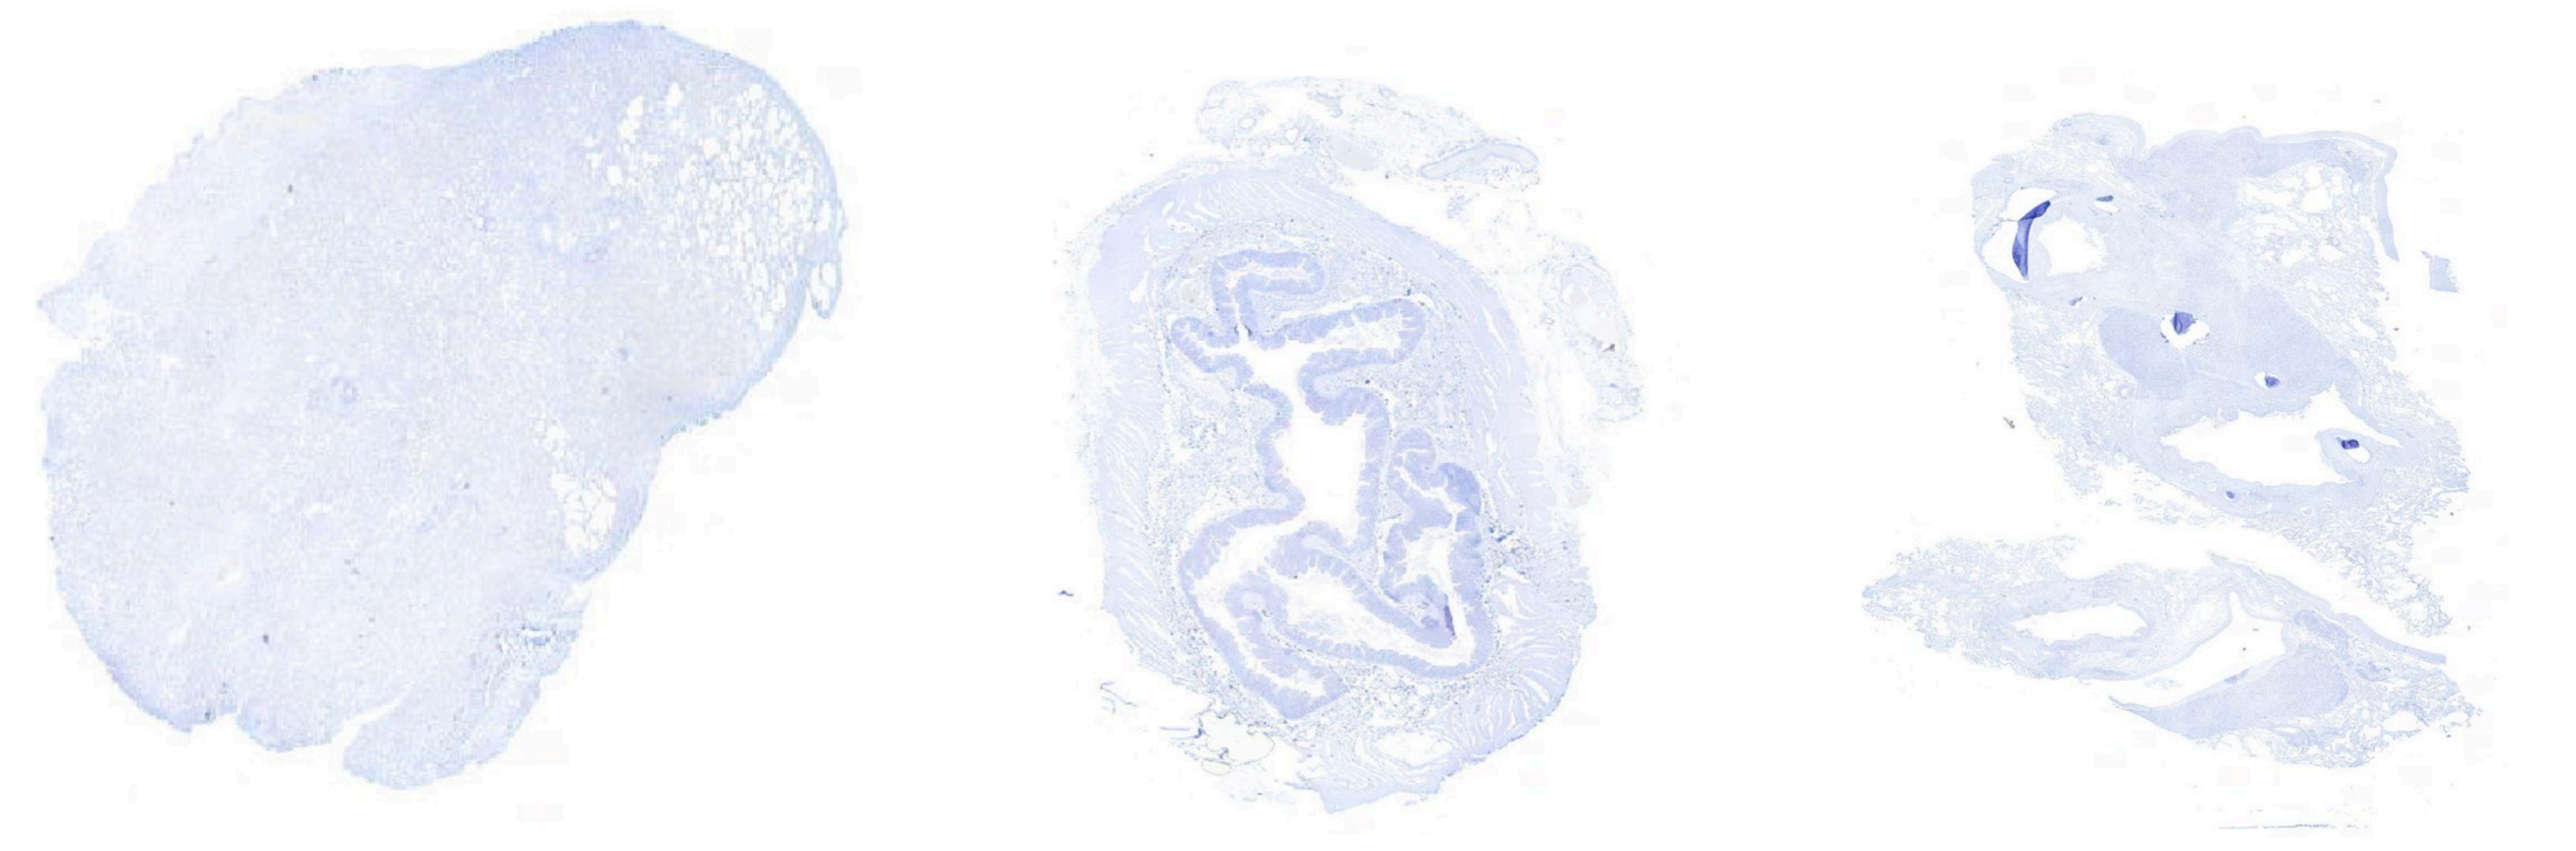

Healthy liver　Healthy colon　Healthy lung

Negative IHC images are represented by the absence of DAB chromogen staining

3. PB-223 binds to core 2 O-glycans

The binding of PB-223 to O-glycans was evaluated using an O-Glycan array containing 94 different O-glycans

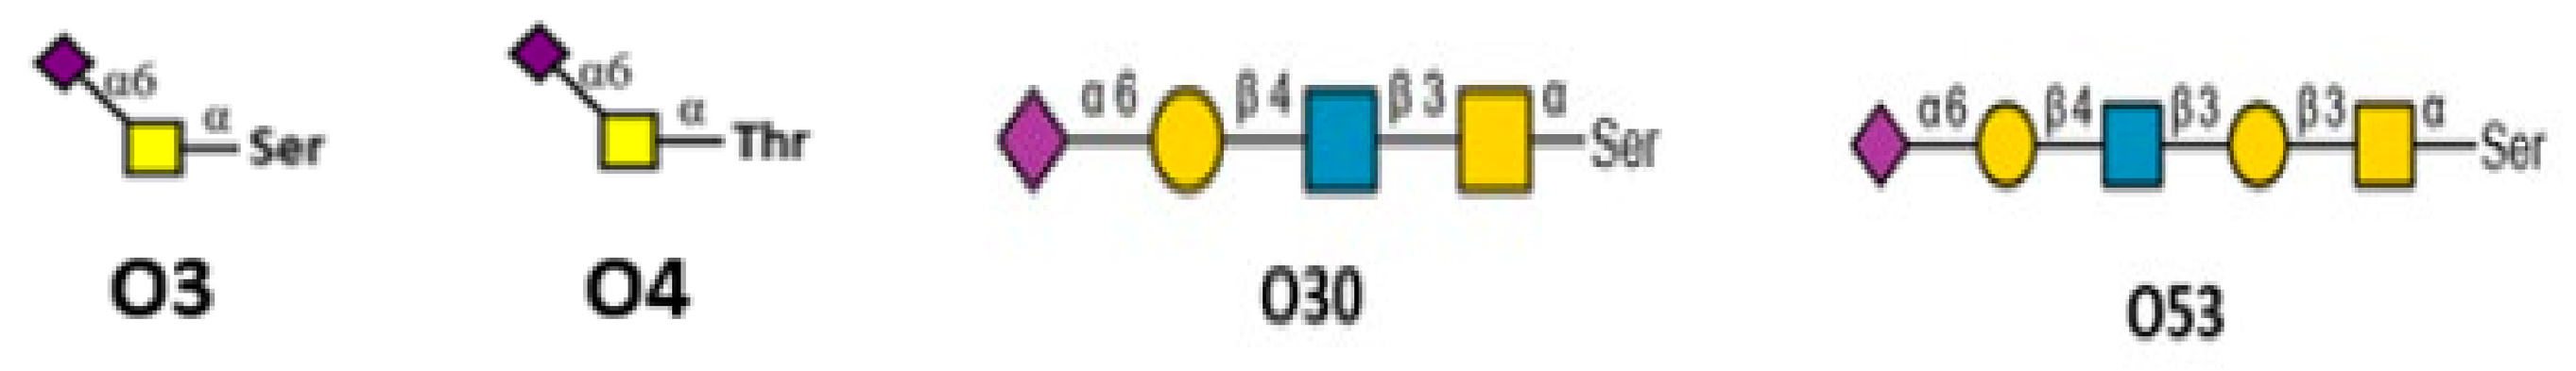
α6
α
Ser
O3
α6
α
Thr
O4
α6 β4 β3 α
Ser
030
α6 β4 β3 β3 α
Ser
053

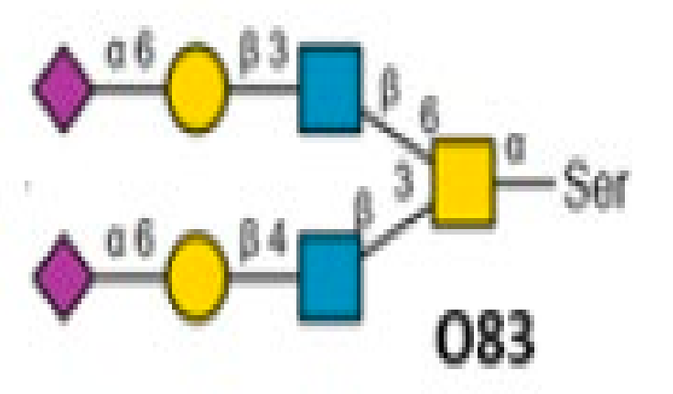
α6 β3
α6 β4
Ser
083

PB-223 binds to 03,04,030,053, 083 (core 2 O-glycans)

4. Flow cytometry: PB-223 shows a stronger binding than NEO-102 to cancer cell lines expressing core 2 O-glycans

Cell Line	Tumor Type	% NEO-102 Positive Cells (MFI)	% PB-223 Positive Cells (MFI)	Fold Increase	% Core 2 O-glycan expression
LoVo	Colorectal adenocarcinoma	0.90% (253)	1.01% (254)	1.12	053 (0.29%)
SW-403	Colorectal adenocarcinoma	**22.21%** (62)	**51.23%** (102)	**2.31**	N.T.
COLO-205	Colorectal adenocarcinoma	**10.30%** (48)	**41.05%** (78)	**3.98**	N.T.
HCC1937	Triple negative breast cancer	8.39% (86)	**42.08%** (134)	**5.02**	053 (6.32%)
OV-90	Ovarian adenocarcinoma	**24.66%** (392)	**43.12%** (469)	**1.75**	053 (6.76%)
PC-3	Prostate adenocarcinoma	8.81% (97)	**13.23%** (107)	1.50	N.T.

Table 1. Positive human cancer cell lines for NEO-102 and PB-223 appear in bold text. Positivity was defined as % positive cells >10%. Cancer cell lines in which we observed more than 1.5-fold increase in percentage of positive cells after staining with PB-223 compared to NEO-102 appear in bold and green. N.T.= Not Tested for glycans expression

5. PB-223 exhibits enhanced binding to human cancer tissues compared to NEO-102 by IHC

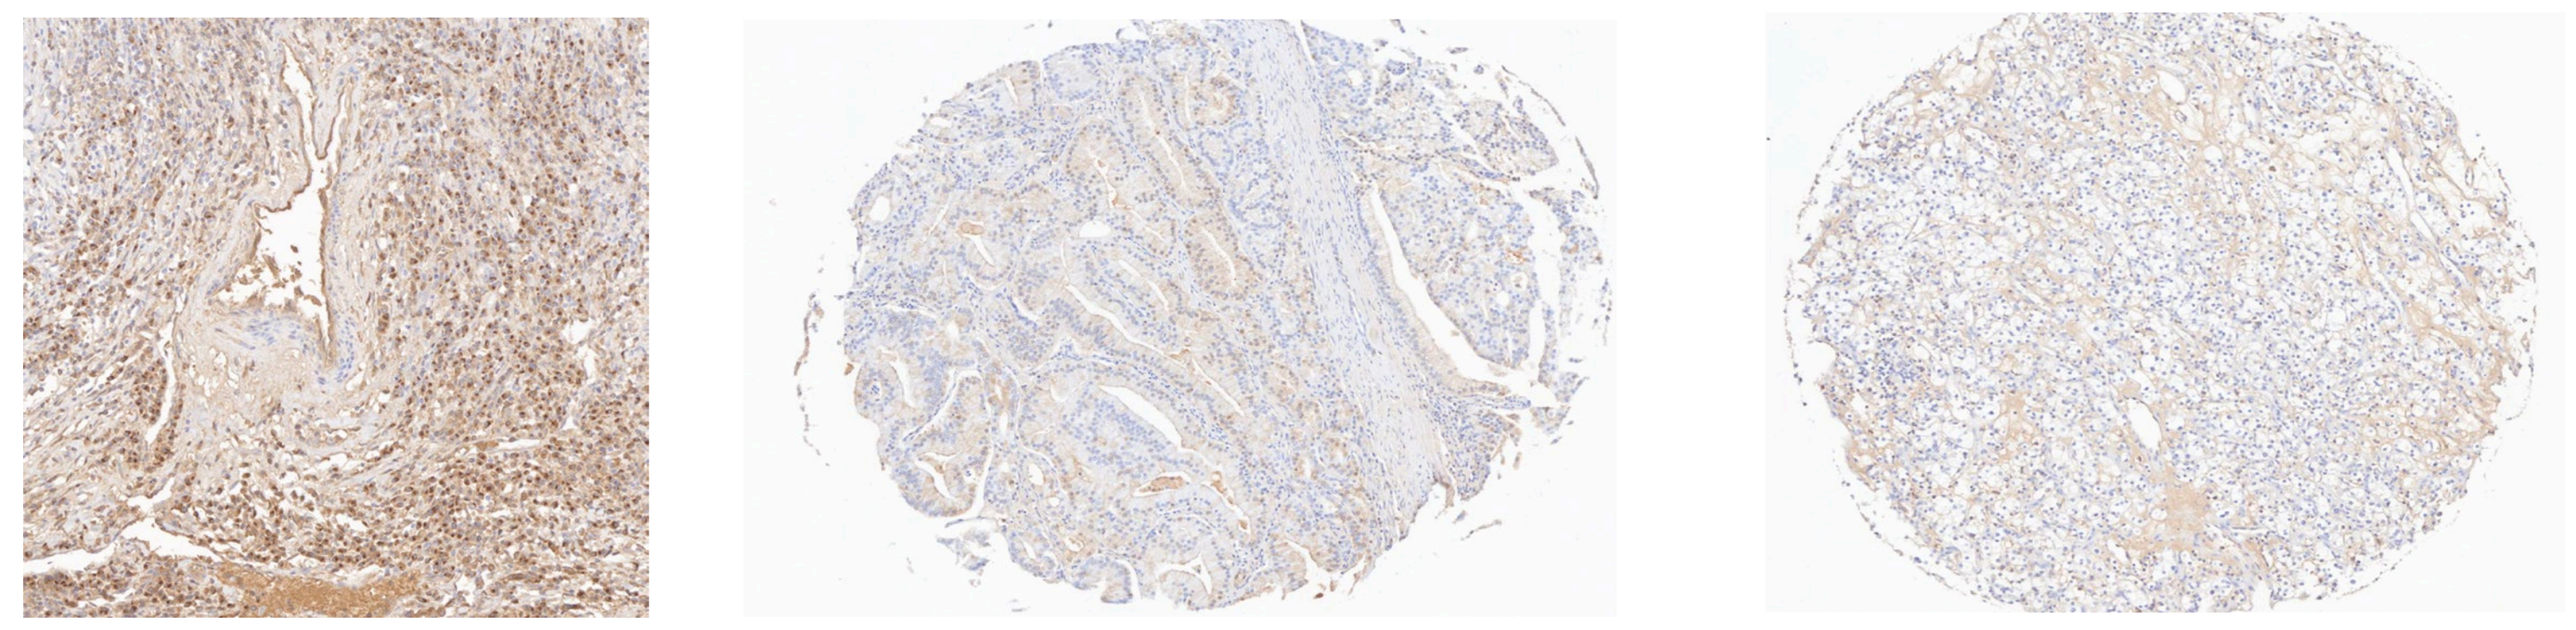

TNBC (PB-223)　Prostate (PB-223)　Kidney (PB-223)

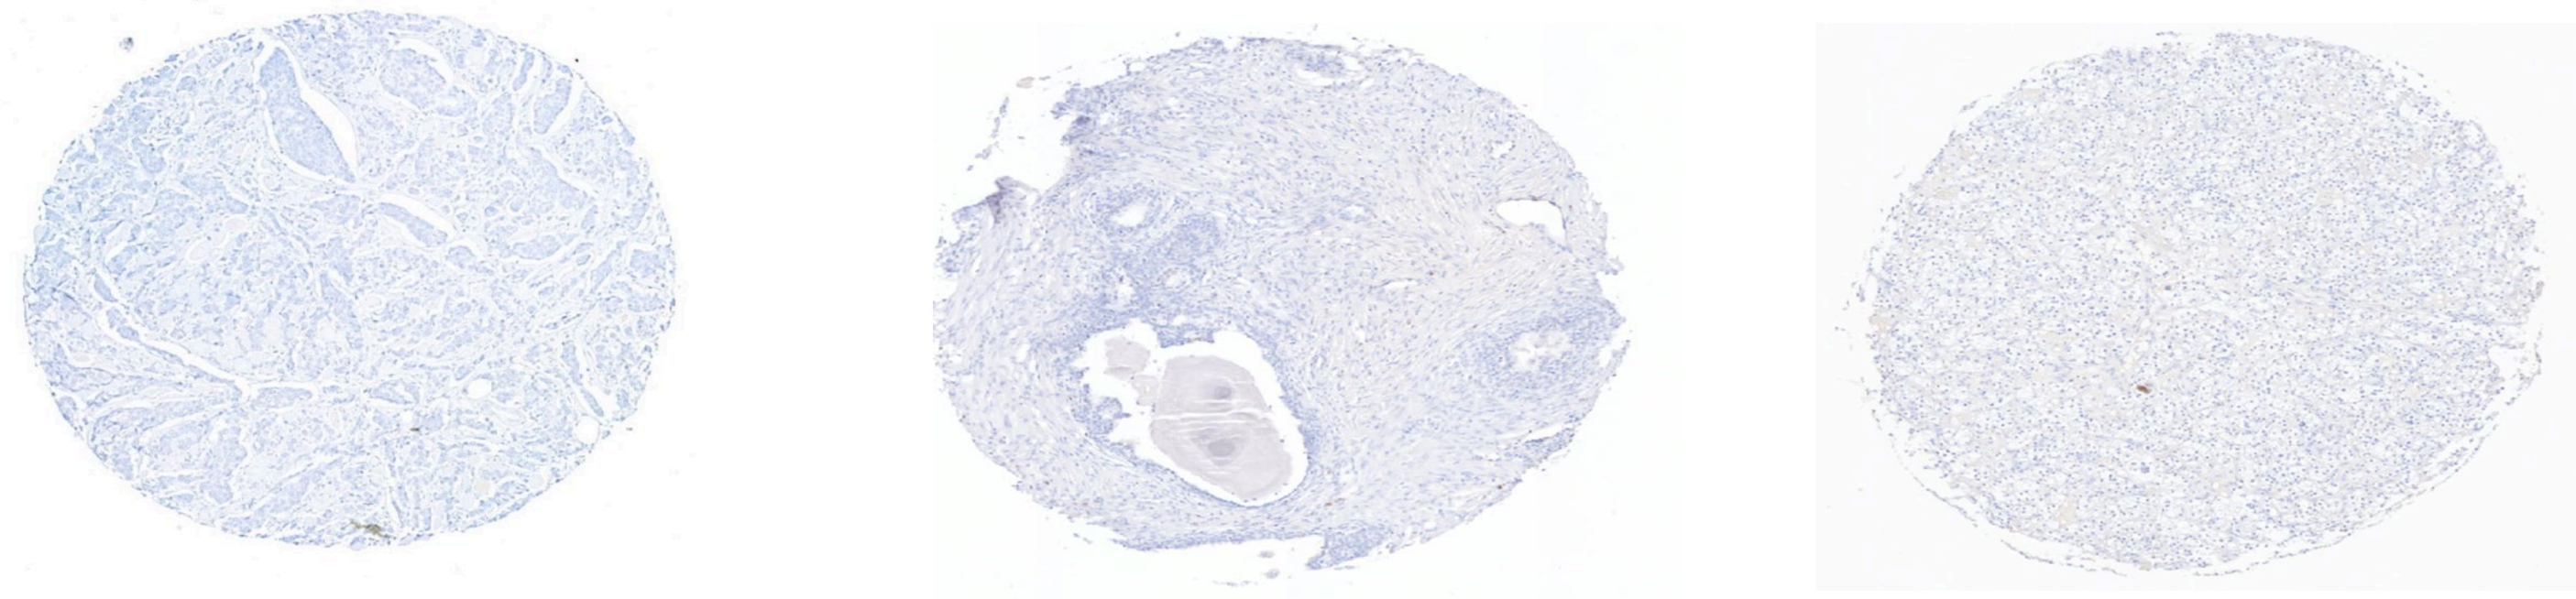

TNBC (NEO-102)　Prostate (NEO-102)　Kidney (NEO-102)

Positive tumor tissues for PB-223 and NEO-102 staining are depicted in brown color (DAB chromogen staining). Negative IHC images are represented by the absence of DAB chromogen staining (absence of brown color). TNBC=Triple Negative Breast Cancer

PB-233 also binds to other tumor tissues not reactive with NEO-102, including head and neck, kidney, liver, bladder cancer.

6. PB-223 internalizes into OV-90

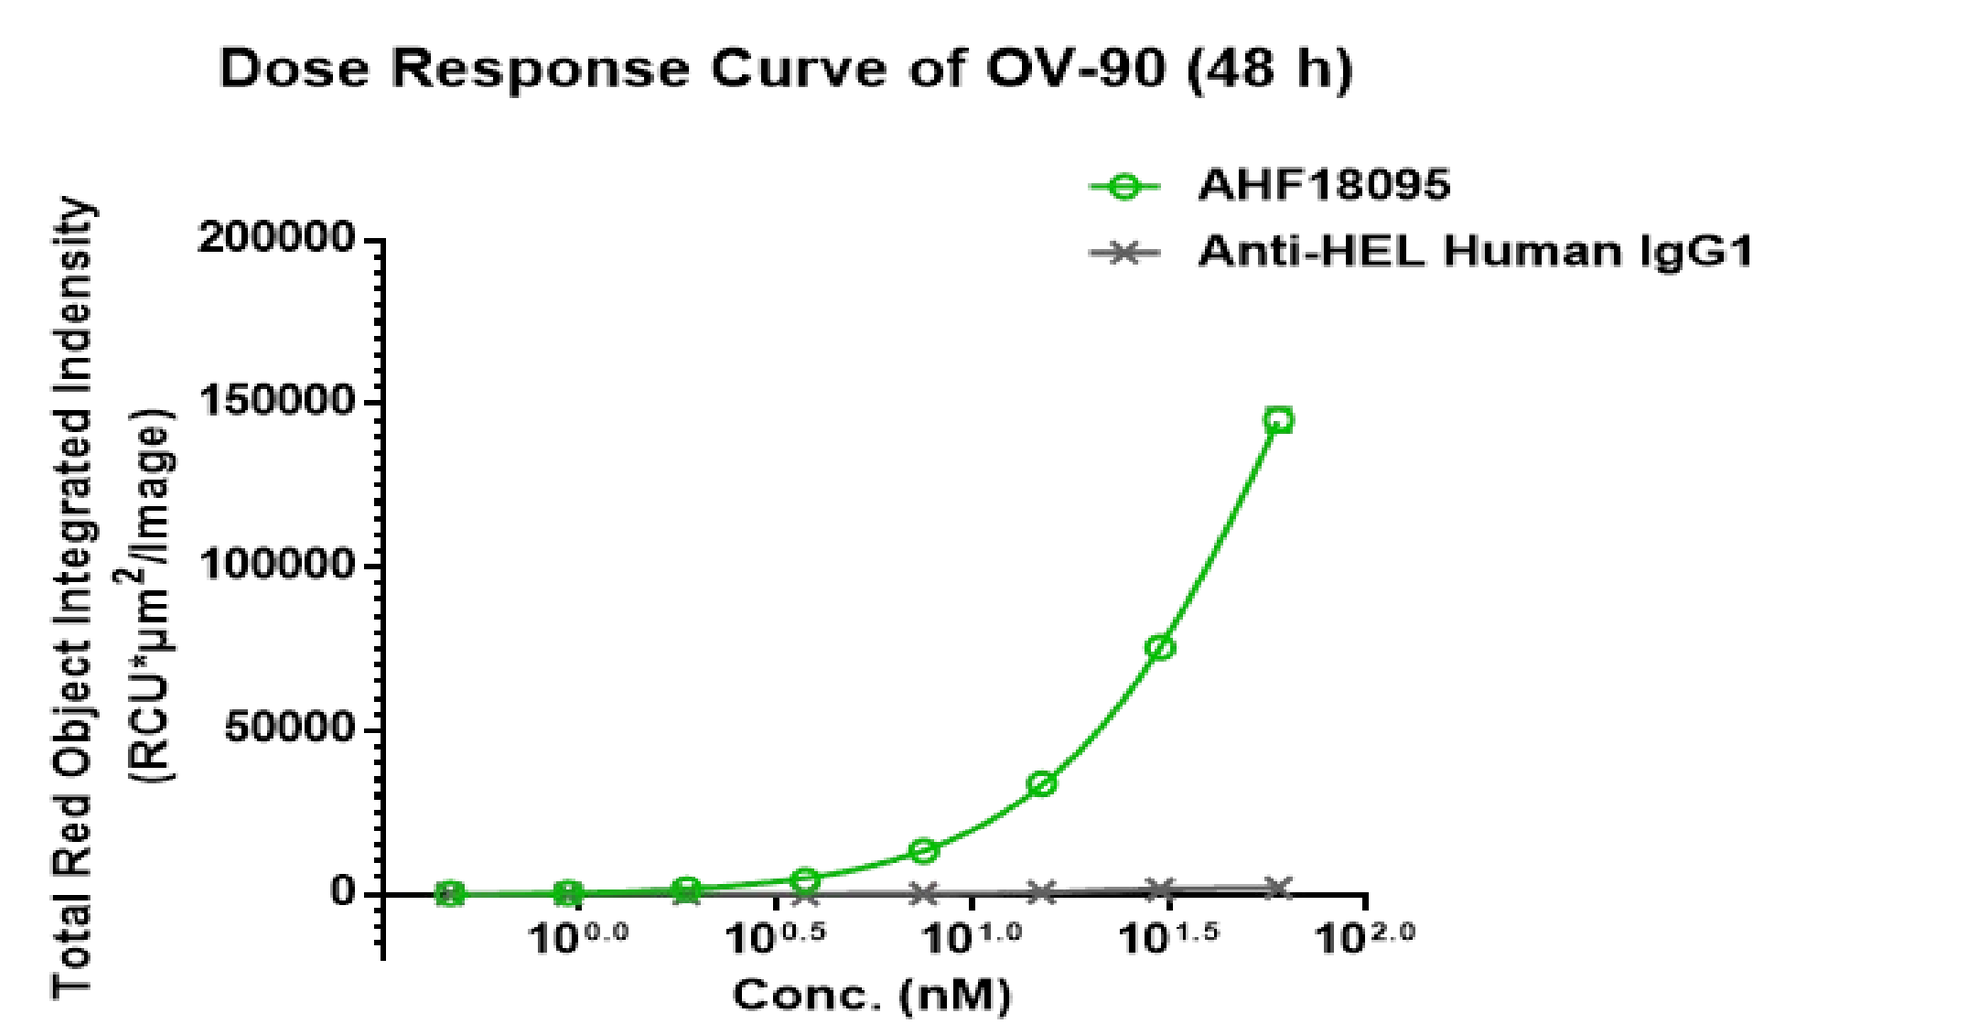
Dose Response Curve of OV-90 (48 h)
AHF18095
Anti-HEL Human IgG1
Total Red Object Integrated Indensity (RCU*µm²/Image)
200000
150000
100000
50000
0
10^0.0
10^0.5
10^1.0
10^1.5
10^2.0
Conc. (nM)

48 hours incubation with PB-223 and the isotype control antibody, anti-HEL human IgG1

Conclusions

The high affinity clone PB-223 showed a KD of 4.5-fold less than NEO-102. Flow cytometry and IHC analysis suggest that PB-223 can bind to a wider spectrum of tumor types than NEO-102 but not to healthy tissues. We identified the Core-2 O-glycan as the binding epitope of PB-223. In addition,PB-223 can be internalized in the cell line OV-90, expressing core 2 O-glycans. PB-223 may be a potential candidate for the development of antibody-drug conjugates for treatment of various human carcinomas.