An anti-carcinoma monoclonal antibody (mAb) NEO-201 can also target human Acute Myeloid Leukemia (AML) cell lines *in vitro*

Alessandra Romano[1], Nunziatina Parrinello[1], Sara Marino[1], Enrico La Spina[1], Massimo Fantini[2], Philip M Arlen[2], Kwong Y Tsang[2], Francesco Di Raimondo[1]

[1]Dipartimento di Chirurgia e Specialità Medico Chirurgiche, Universita di Catania, Catania, Italy; [2]Precision Biologics, Inc., Bethesda, MD, USA

Abstract

Background: NEO-201 is a humanized IgG1 monoclonal antibody (mAb) that recognizes tumor-associated variants of CEACAM5/6. This mAb is remarkably tumor-specific in its staining profile and demonstrated its ability to react to wide range of human carcinoma cell lines by flow cytometry and tumor tissues by immunohistochemistry. NEO-201 exhibited both ADCC and complement-dependent cellular cytotoxicity (CDC) activity against human carcinoma cell *in vitro* and counteracted the growth of human pancreatic xenograft tumors *in vivo*. A recent Phase 1 clinical trial at the NCI has determined both safety and recommended Phase 2 dosing. We have also seen the expression of the NEO-201 target on hematologic cells, specifically Tregs and neutrophils. Due to epitope being expressed both on malignant epithelial cells as well as several hematologic cells, we designed this study to explore the reactivity of NEO-201 against human hematological neoplastic cells *in vitro,* such as Acute Myeloid Leukemia (AML), Multiple Myeloma (MM), Acute Lymphoblastic Leukemia (ALL), Mantel Cell Lymphoma (MCL) cells.

Methodology: Flow cytometry analysis was used to profile a panel of human hematologic neoplastic cell lines for NEO-201 binding. Cell lines used were six AML (HL60, U937, MOLM13, AML2, IMS-M2 and OCL-AML3), two MM (OPM2, MM1.S), two ALL (SUP-B15, RPMI8402) and four MCL (Jeko-1, Z138, JVM2 and JVM13). Markers used for flow cytometry analysis were CD15, CD45, CD38, CD138, CD14, CD19 and NEO-201. The ability of NEO-201 to mediate ADCC activity against hematological neoplastic cell lines was assessed through a non-radioactive ADCC assay, using PBMCs or isolated human NK cells as effector cells at different E:T ratios.

Results: NEO-201 was found to react with AML and MM cell lines. 5 of 6 AML cell lines tested bound to NEO-201 and the % of positive cells were 47%, 99.5%,100%,100% and 97.8% for HL60, U937, MOLM13, AML2 and IMS-M2, respectively. The % of positive cells in the two MM cell line were 99% and 18% for OPM2 and MM1.S, respectively. NEO-201 did not react against the two ALL and the four MCL cell lines tested. Functional analysis has demonstrated that NEO-201 can mediate ADCC activity against the AML cell line HL60 and against neutrophils expressing its target antigen.

Conclusions: This study demonstrates that NEO-201 target antigen is expressed in most of the AML and in MM cell lines tested *in vitro.* In addition, we have shown that NEO-201 can mediate ADCC against HL60 cell line and neutrophils *in vitro.* Together, these findings provide a rationale for further investigation of the role of NEO-201 in AML as well as MM, further exploring patient PBMCs and bone marrow samples.

1. NEO-201 binds to human carcinoma cell lines and tumor tissues

Tumor Cell Line Flow Cytometry

CELL LINE	TUMOR TYPE	% POSITIVE	MFI
COLO 205	**Colon**	**10.33**	**245**
HT-29	**Colon**	**38.40**	**352**
LS174T	**Colon**	**46.46**	**345**
SW1116	Colon	2.36	194
SW1463	Colon	1.23	278
SW480	Colon	1.70	575
ASPC-1	**Pancreatic**	**79.26**	**8927**
BxPC-3	**Pancreatic**	**97.25**	**2584**
CAPAN-2	**Pancreatic**	**29.69**	**327**
CFPAC-1	**Pancreatic**	**97.79**	**9281**
PANC-1	Pancreatic	3.29	289
H441	**NSCLC (adenocarcinoma)**	**69.16**	**675**
H522	NSCLC (adenocarcinoma)	1.38	238
HCC4006	**NSCLC (adenocarcinoma)**	**99.27**	**9899**
HCC827	**NSCLC (adenocarcinoma)**	**77.46**	**692**
SK-LU-1	NSCLC (adenocarcinoma)	1.77	685
CALU-1	NSCLC (squamous)	4.22	571
H1703	NSCLC (squamous)	4.16	111
H226	NSCLC (squamous)	4.83	209
H520	**NSCLC (squamous)**	**61.78**	**443**
AU-565	**Breast (HER2+)**	**50.04**	**227**
BT-474	**Breast (PR+/HER2+)**	**68.79**	**591**
HCC1500	Breast (ER+/PR+)	1.53	597
SK-BR-3	Breast (HER2+)	1.61	329
T-47D	Breast (ER+/PR+)	8.00	161
ZR-75-1	**Breast (ER+/PR+/HER2+)**	**68.80**	**550**
BT-549	Breast (ER-/PR-/HER2-)	1.47	477
HCC1937	**Breast (ER-/PR-/HER2-)**	**19.14**	**510**
HCC38	Breast (ER-/PR-/HER2-)	2.15	226
MDA-MB-468	Breast (ER-/PR-/HER2-)	6.33	344

NEO-201 is reactive against a broad range of *in vitro* cultured tumor cell lines. NEO-201 positive cell lines appear in bold text. NEO-201 positivity was defined as % positive >10%.

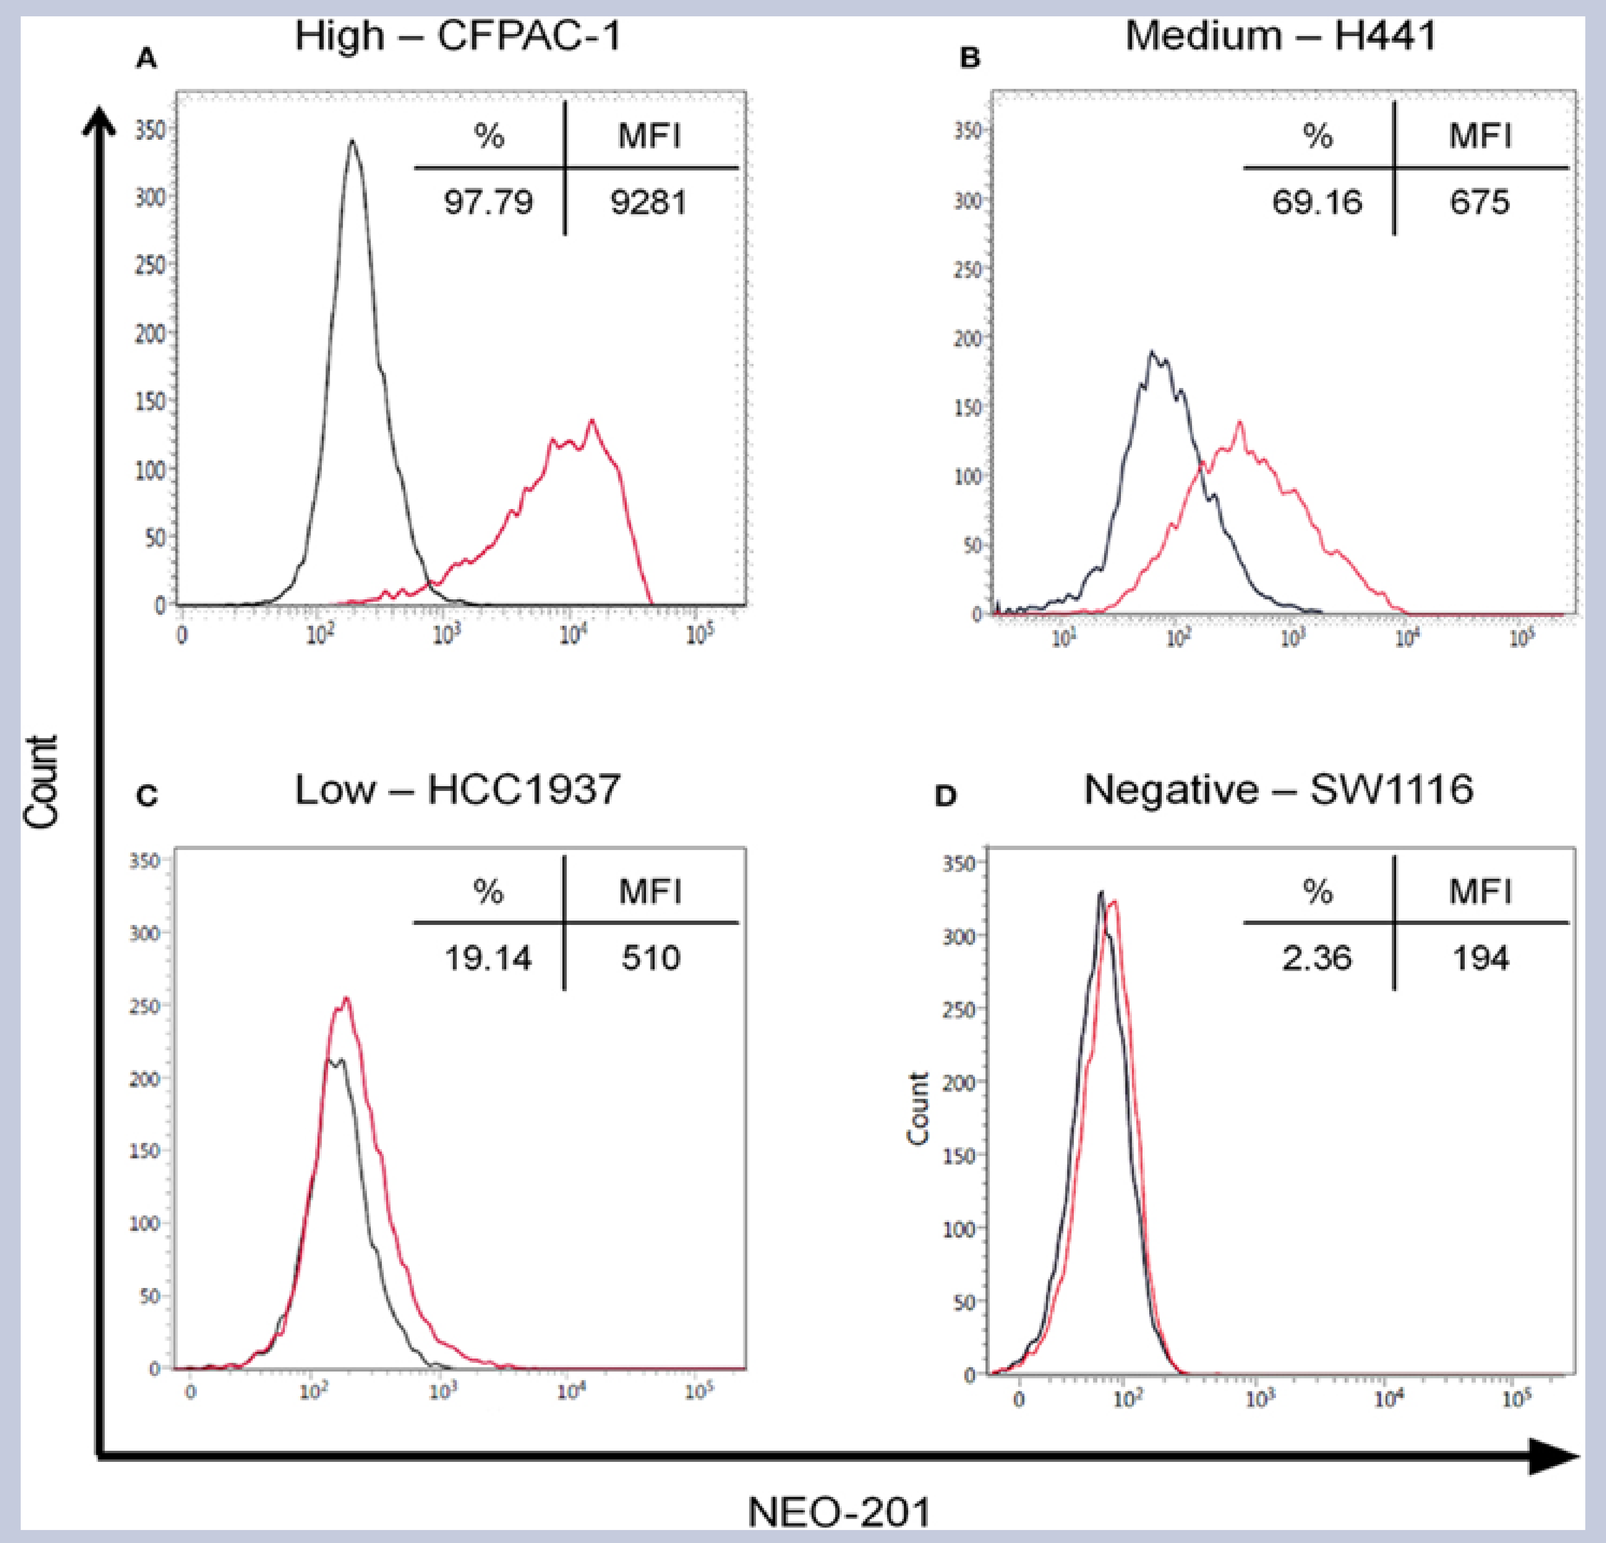
A
High – CFPAC-1
% | MFI
97.79 | 9281
B
Medium – H441
% | MFI
69.16 | 675
C
Low – HCC1937
% | MFI
19.14 | 510
D
Negative – SW1116
% | MFI
2.36 | 194
Count
NEO-201

Positivity was determined using fluorescence minus one (FMO) controls. Positive cell lines were ranked according to their quantified expression level (% positive × MFI), and then sorted into groups of low (<200), medium (200-1000), and high (<1000) expression.

Normal vs. Tumor Tissue Microarray IHC

CEACAM 5 CEACAM 6 NEO-201

Normal Colon

Colon Cancer

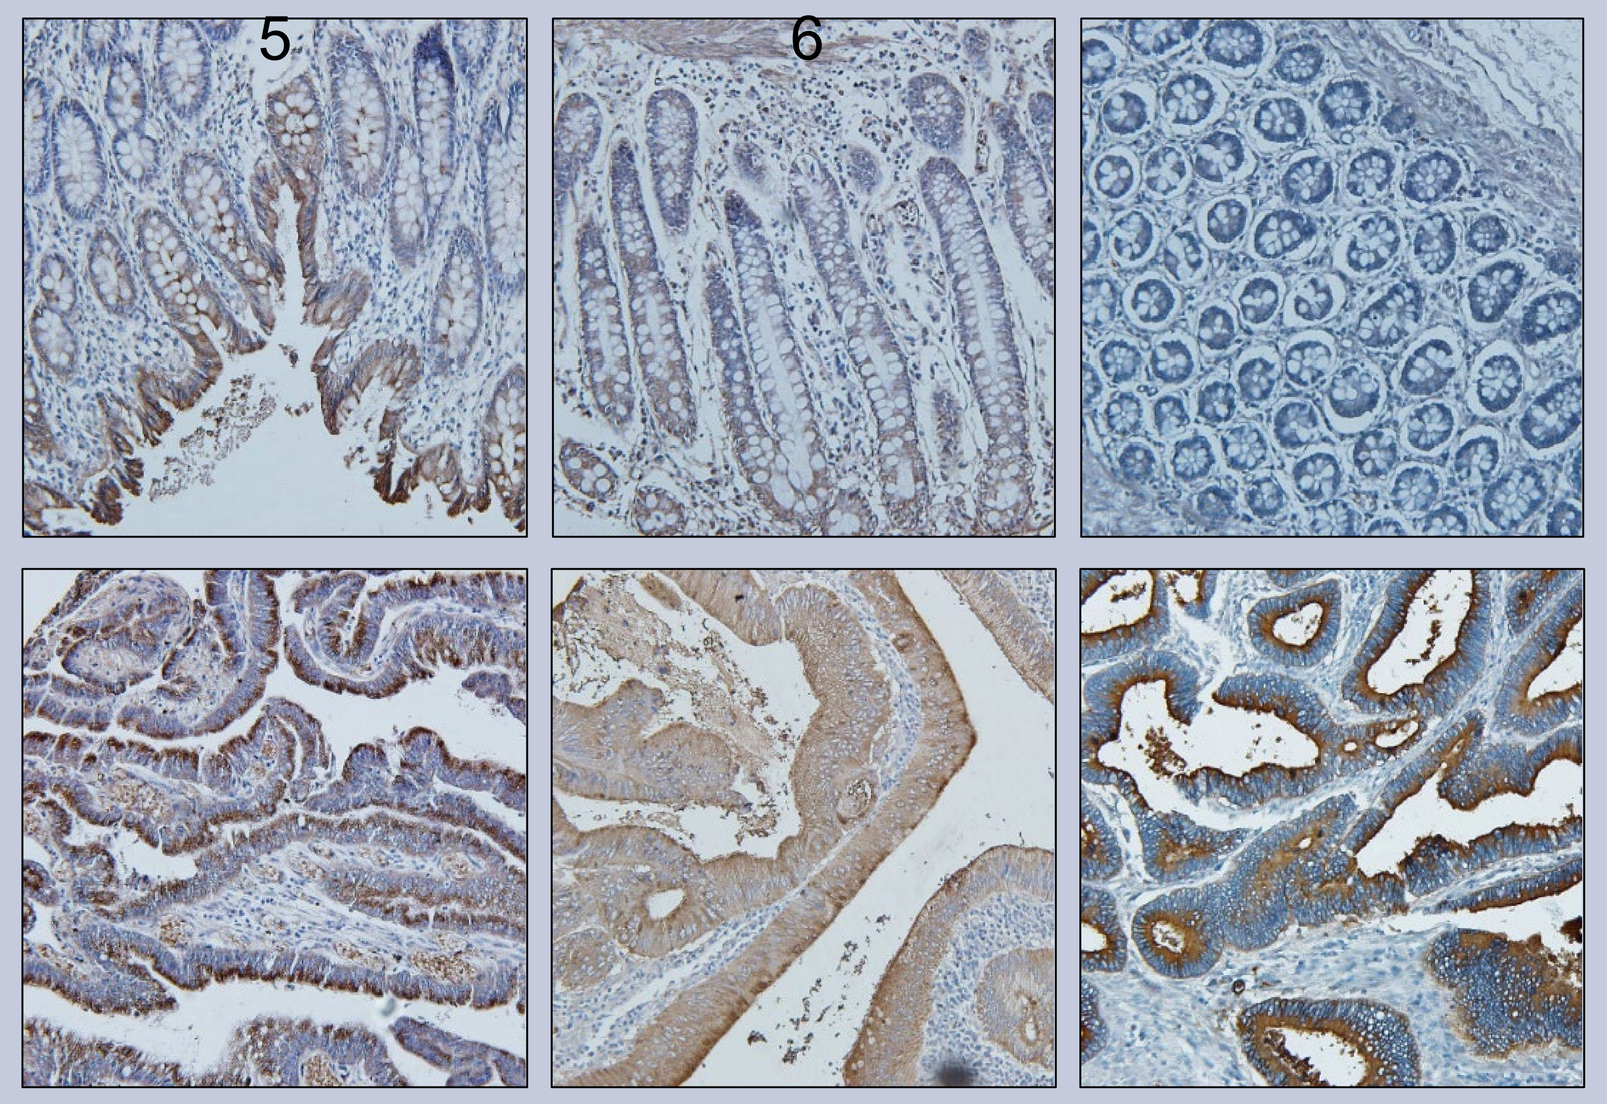

Quantification of Staining

Staining Pattern			Normal Colon (Cases, %)	Colon Cancer (Cases, %)
NEO-201	CEACAM5	CEACAM6		
-	-	-	**2/31 (6%)**	**1/32 (3%)**
+	-	-	0/31 (0%)	0/32 (0%)
-	+	-	0/31 (0%)	0/32 (0%)
-	-	+	**1/31 (3%)**	0/32 (0%)
+	+	-	0/31 (0%)	0/32 (0%)
+	-	+	0/31 (0%)	0/32 (0%)
-	+	+	**28/31 (90%)**	**3/32 (9%)**
+	+	+	0/31 (0%)	**28/32 (88%)**

Similar results observed from normal and cancerous pancreas and lung tissues.

2. NEO-201 binds to various human hematological neoplastic cell lines

Tumor Cell Line Flow Cytometry

Cell line	Tumor Type	% positive	MFI
HL60	AML	**47.0**	4,864
U937	AML	**99.5**	5,985
MOLM13	AML	**100**	28,278
AML2	AML	**100**	17,736
IMS-M2	AML	**97.8**	4,640
OCL-AML3	AML	Neg	NA
OPM2	MM	**99.0**	21,082
MM1.S	MM	**18.0**	2,380

Cell line	Tumor Type	% positive	MFI
SUP-B15	ALL	Neg	NA
RPMI-8402	ALL	Neg	NA
Jeko-1	MCL	Neg	NA
Z138	MCL	Neg	NA
JVM2	MCL	Neg	NA
JVM13	MCL	Neg	NA

NEO-201 positive cell lines appear in bold text. NEO-201 positivity was defined as % positive >5%. AML: Acute Myeloid Leukemia; MM: Multiple Myeloma; ALL: Acute Lymphocytic Leukemia; MCL: Mantle Cell Lymphoma. Neg: < 5% positive.

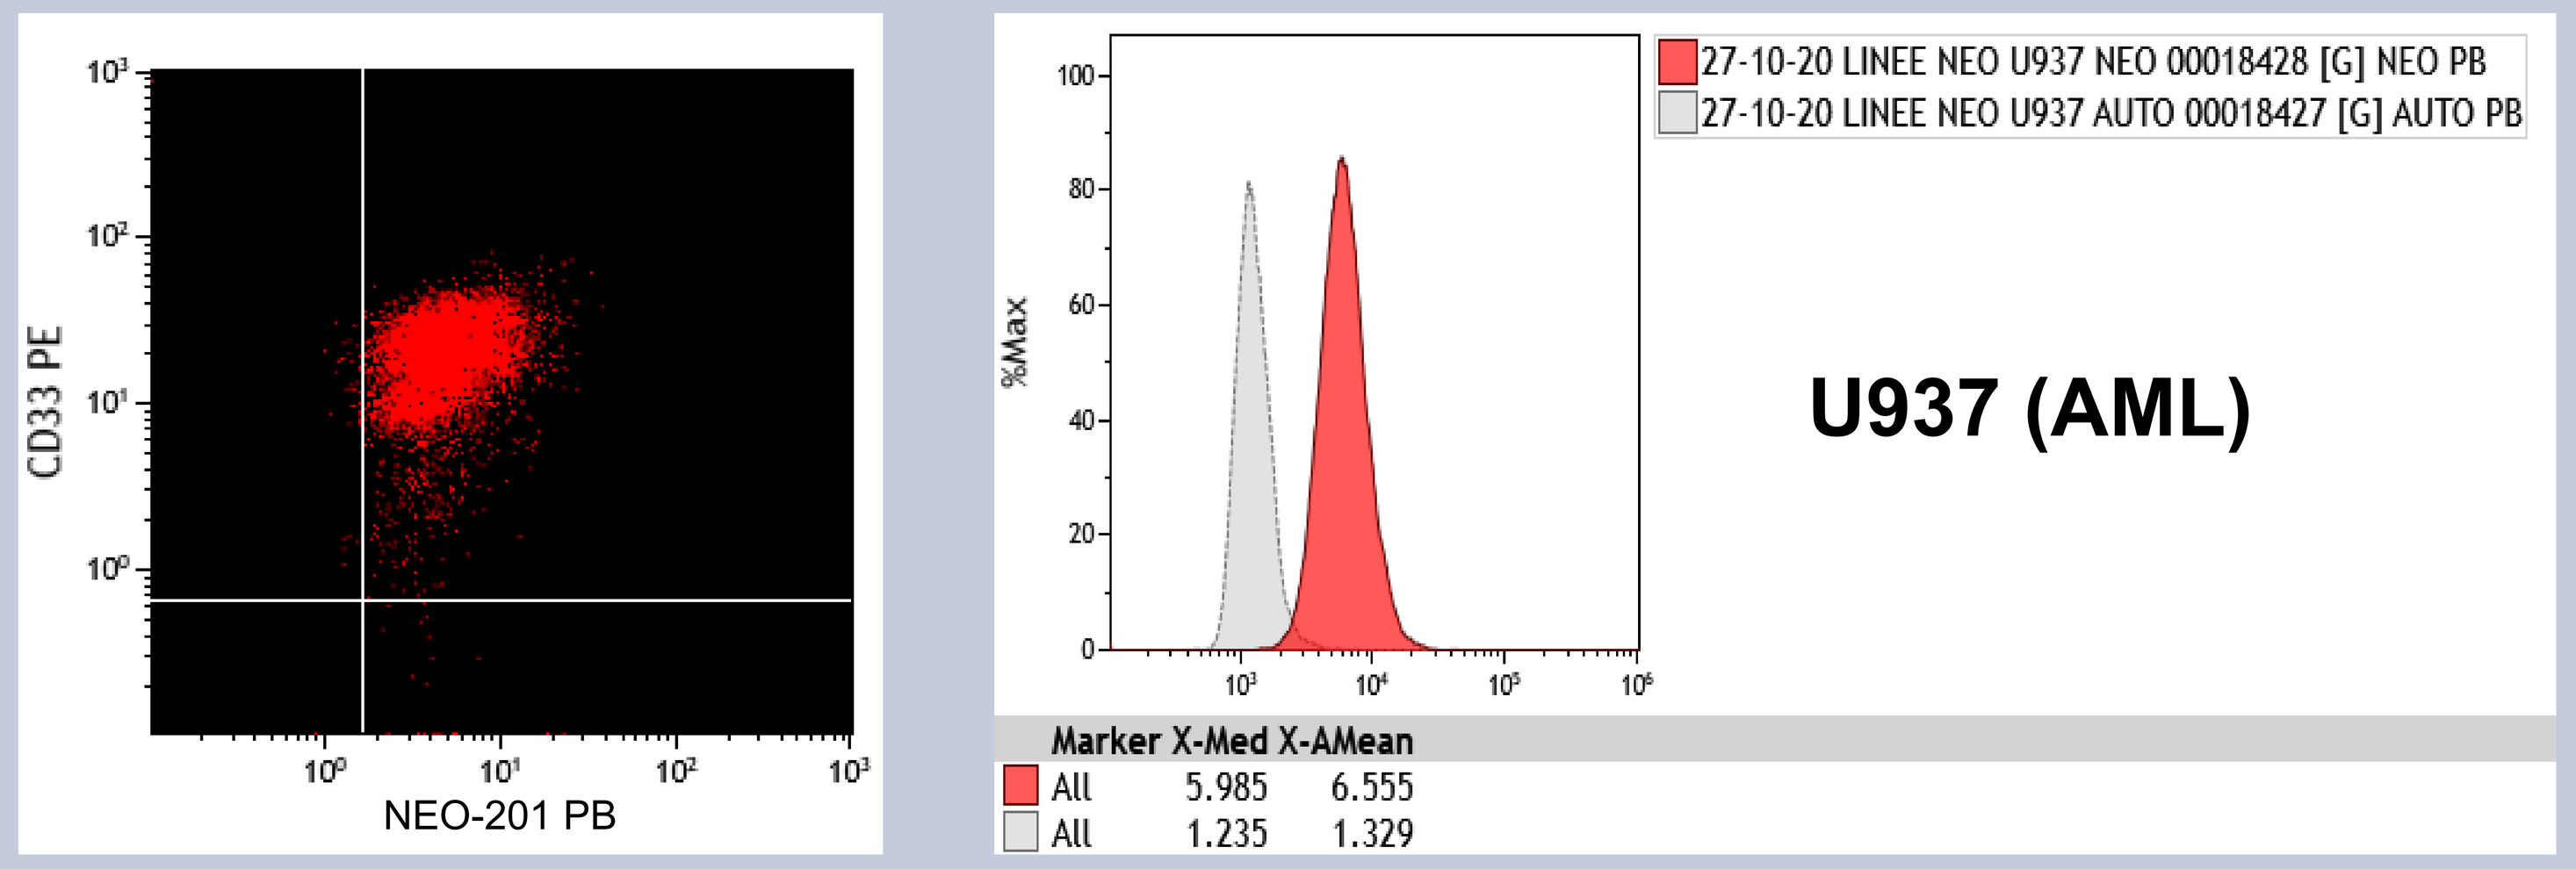
CD33 PE
NEO-201 PB
27-10-20 LINEE NEO U937 NEO 00018428 [G] NEO PB
27-10-20 LINEE NEO U937 AUTO 00018427 [G] AUTO PB
%Max
U937 (AML)
Marker X-Med X-AMean
All 5.985 6.555
All 1.235 1.329

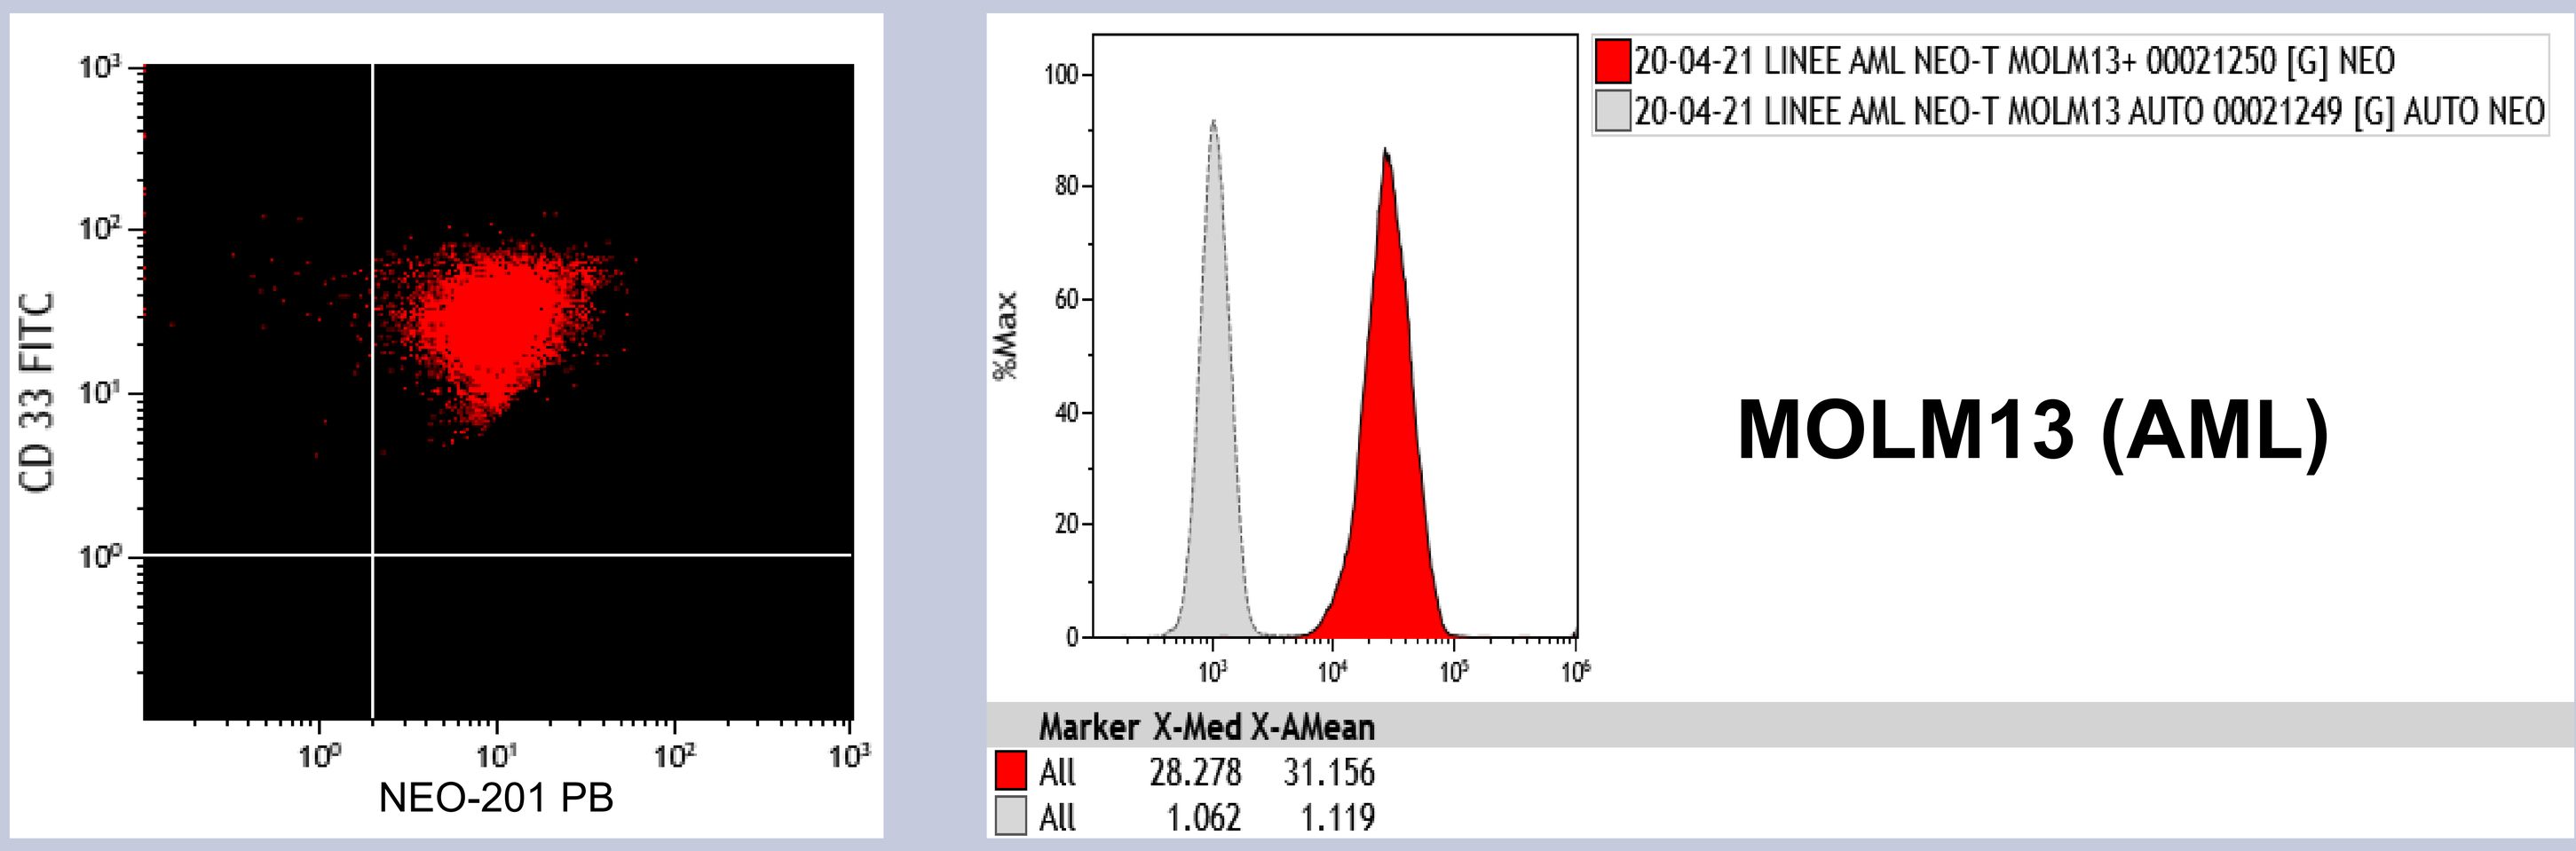
CD 33 FITC
NEO-201 PB
20-04-21 LINEE AML NEO-T MOLM13+ 00021250 [G] NEO
20-04-21 LINEE AML NEO-T MOLM13 AUTO 00021249 [G] AUTO NEO
%Max
MOLM13 (AML)
Marker X-Med X-AMean
All 28.278 31.156
All 1.062 1.119

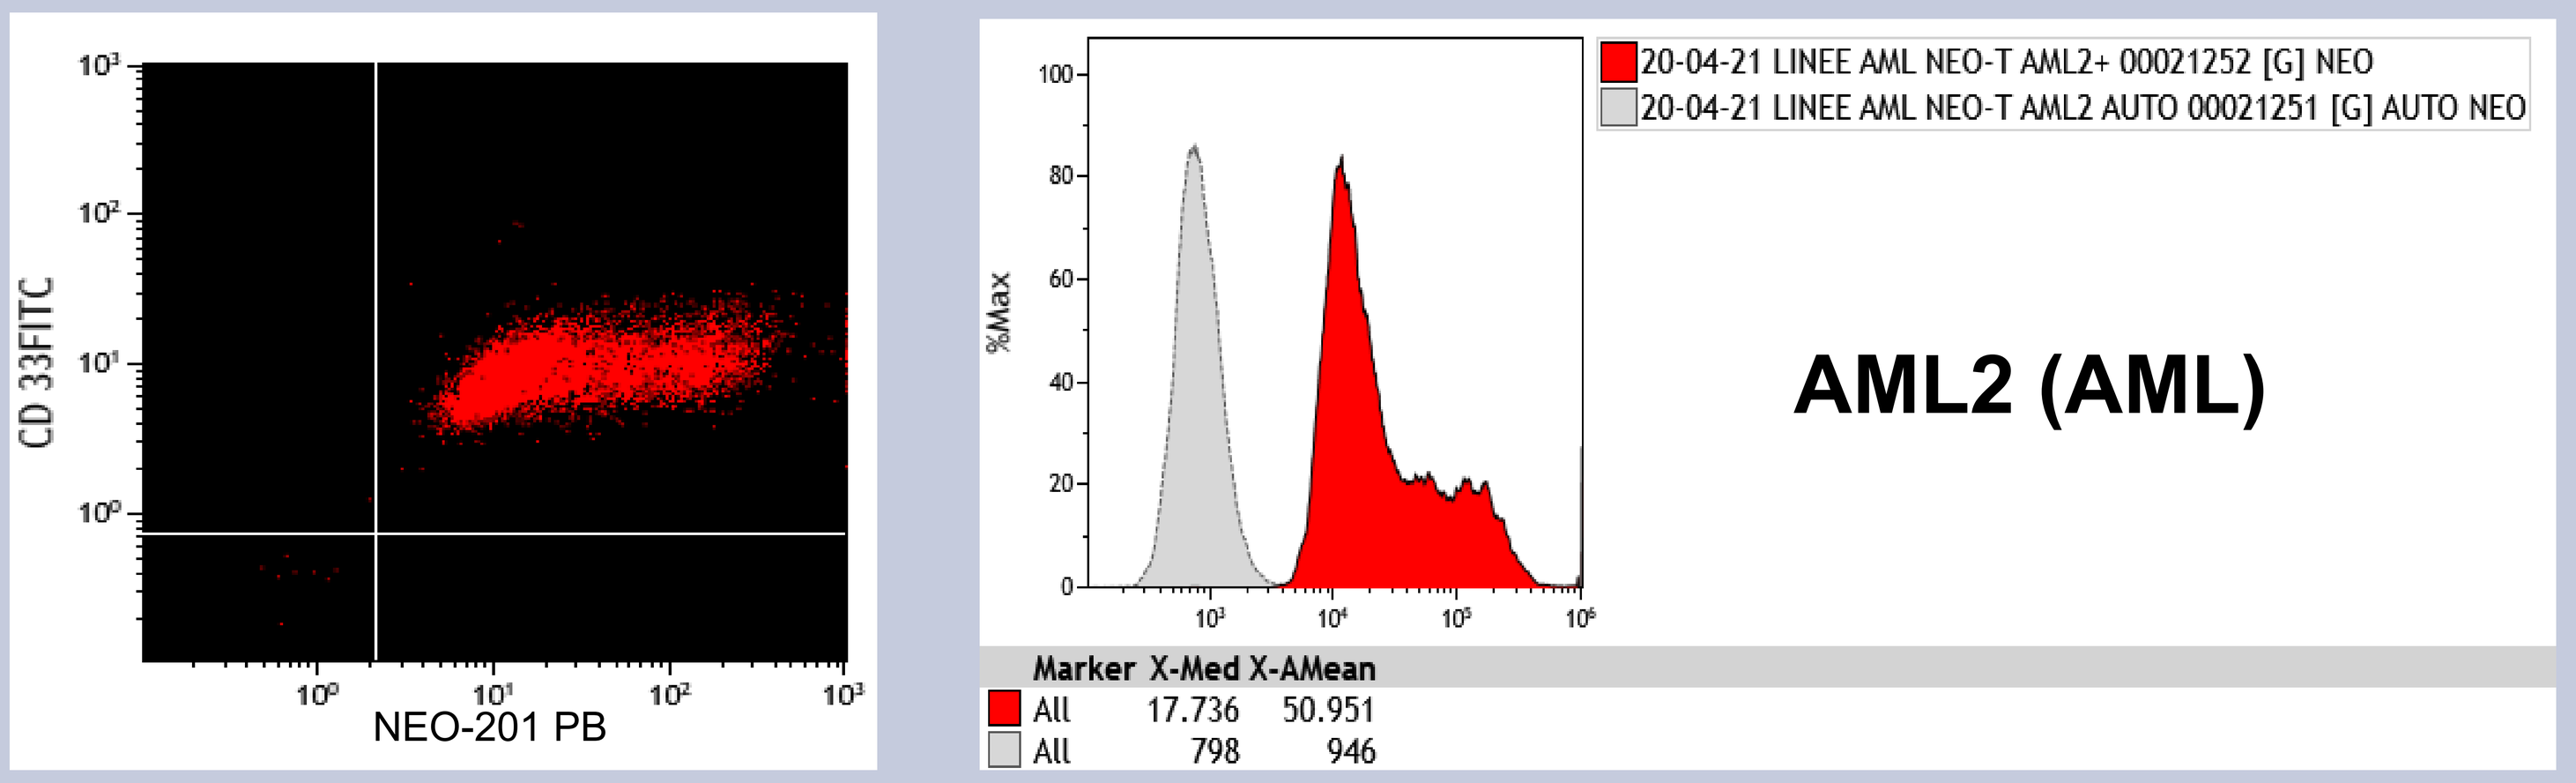
CD 33FITC
NEO-201 PB
20-04-21 LINEE AML NEO-T AML2+ 00021252 [G] NEO
20-04-21 LINEE AML NEO-T AML2 AUTO 00021251 [G] AUTO NEO
%Max
AML2 (AML)
Marker X-Med X-AMean
All 17.736 50.951
All 798 946

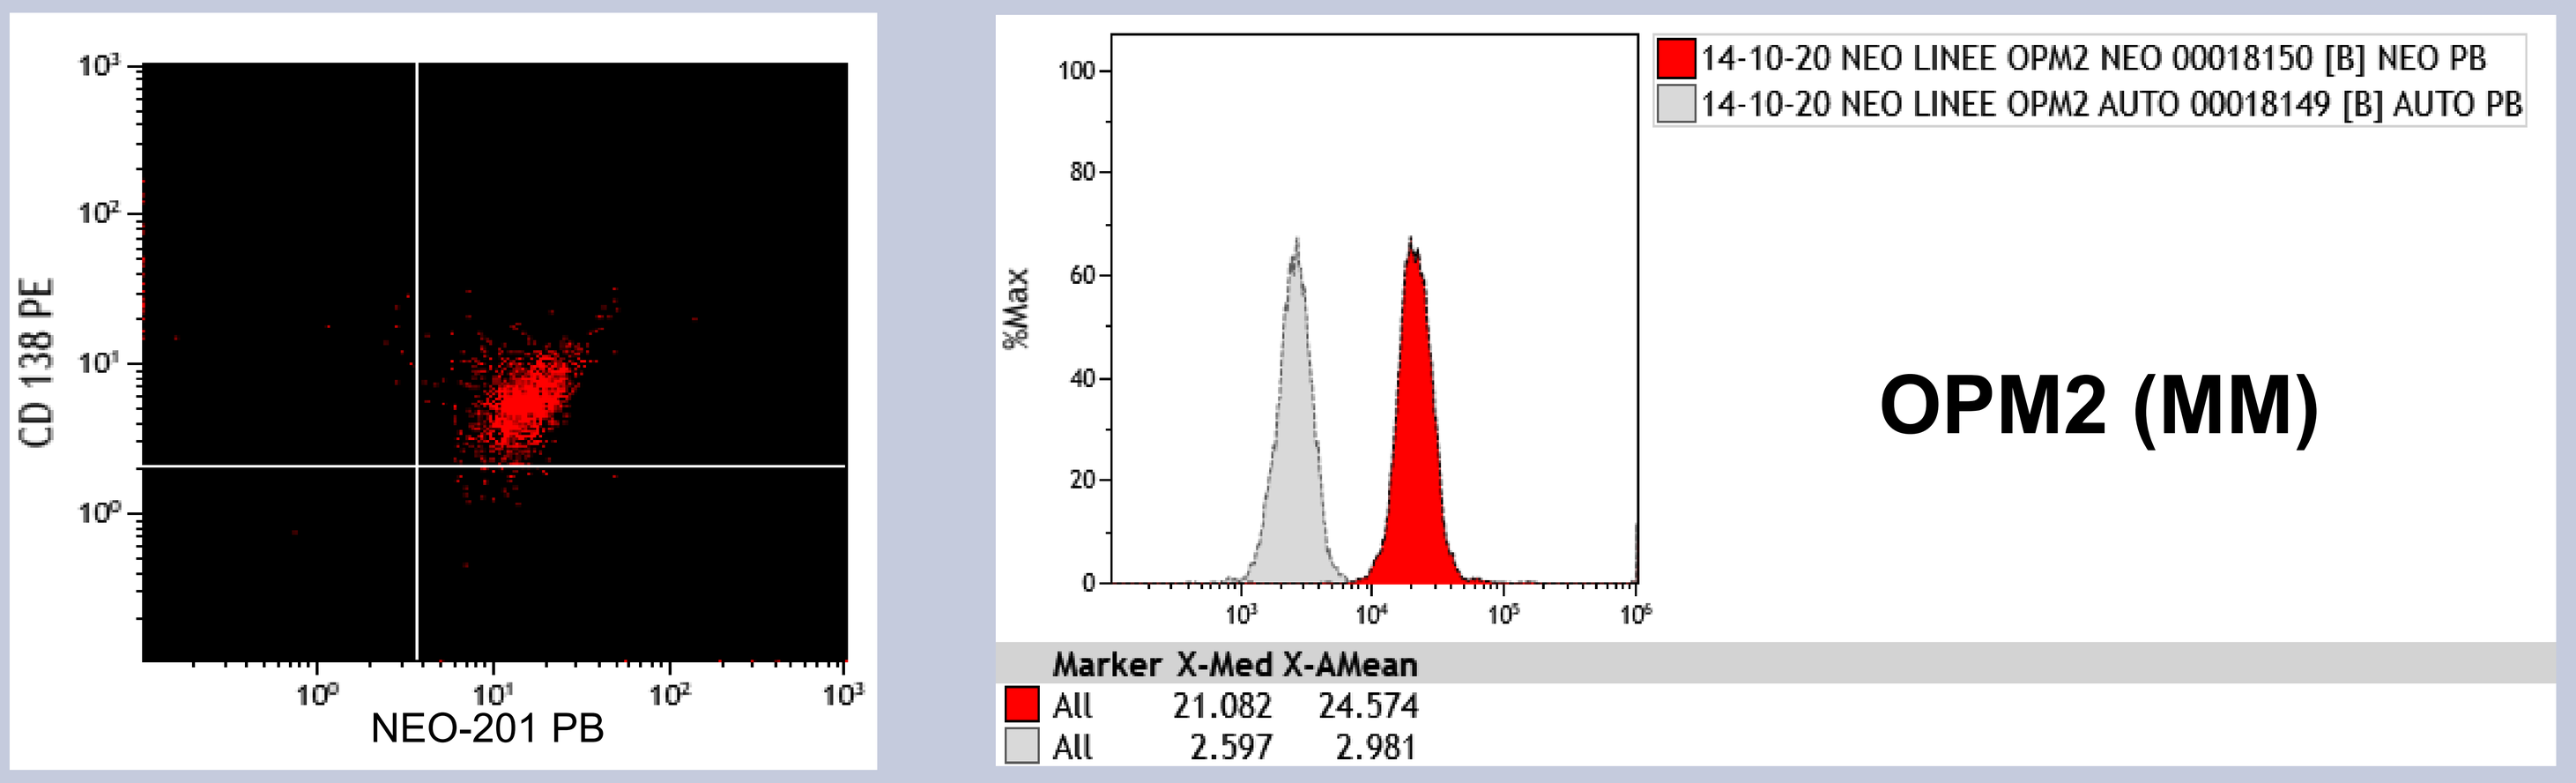
CD 138 PE
NEO-201 PB
14-10-20 NEO LINEE OPM2 NEO 00018150 [B] NEO PB
14-10-20 NEO LINEE OPM2 AUTO 00018149 [B] AUTO PB
%Max
OPM2 (MM)
Marker X-Med X-AMean
All 21.082 24.574
All 2.597 2.981

3. NEO-201 mediates ADCC against the AML cell line HL60

Flow Cytometry

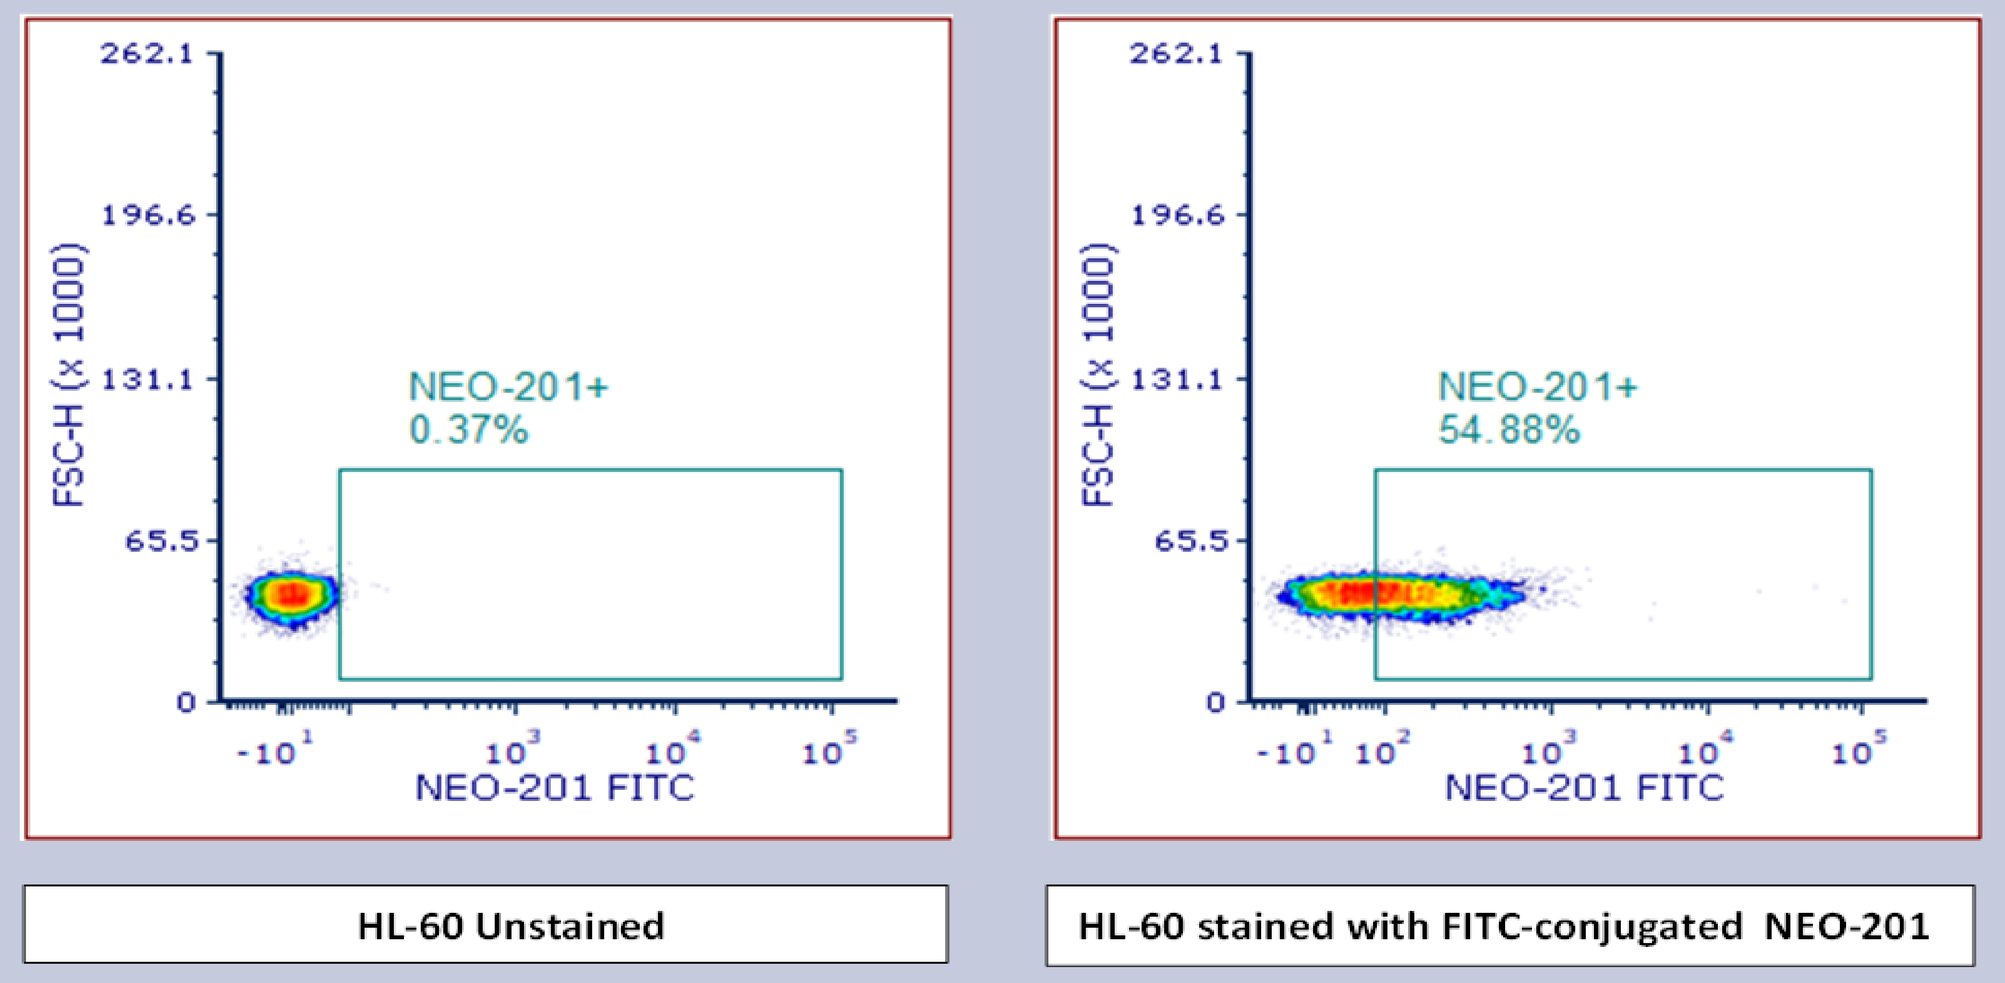
FSC-H (x 1000)
NEO-201+
0.37%
NEO-201 FITC
HL-60 Unstained
NEO-201+
54.88%
NEO-201 FITC
HL-60 stained with FITC-conjugated NEO-201

Flow cytometry analysis of the binding of NEO-201 to HL-60 (AML) cell line. Data are presented as percentage of cells expressing the antigen recognized by NEO-201. Reactivity with NEO-201 was determined by confronting unstained cells (left panel) with cell stained with 10 µg/mL FITC-conjugated NEO-201 (right panel). Positivity was defined as % of positive cells ≥ 5%.

ADCC assay

PBMCs

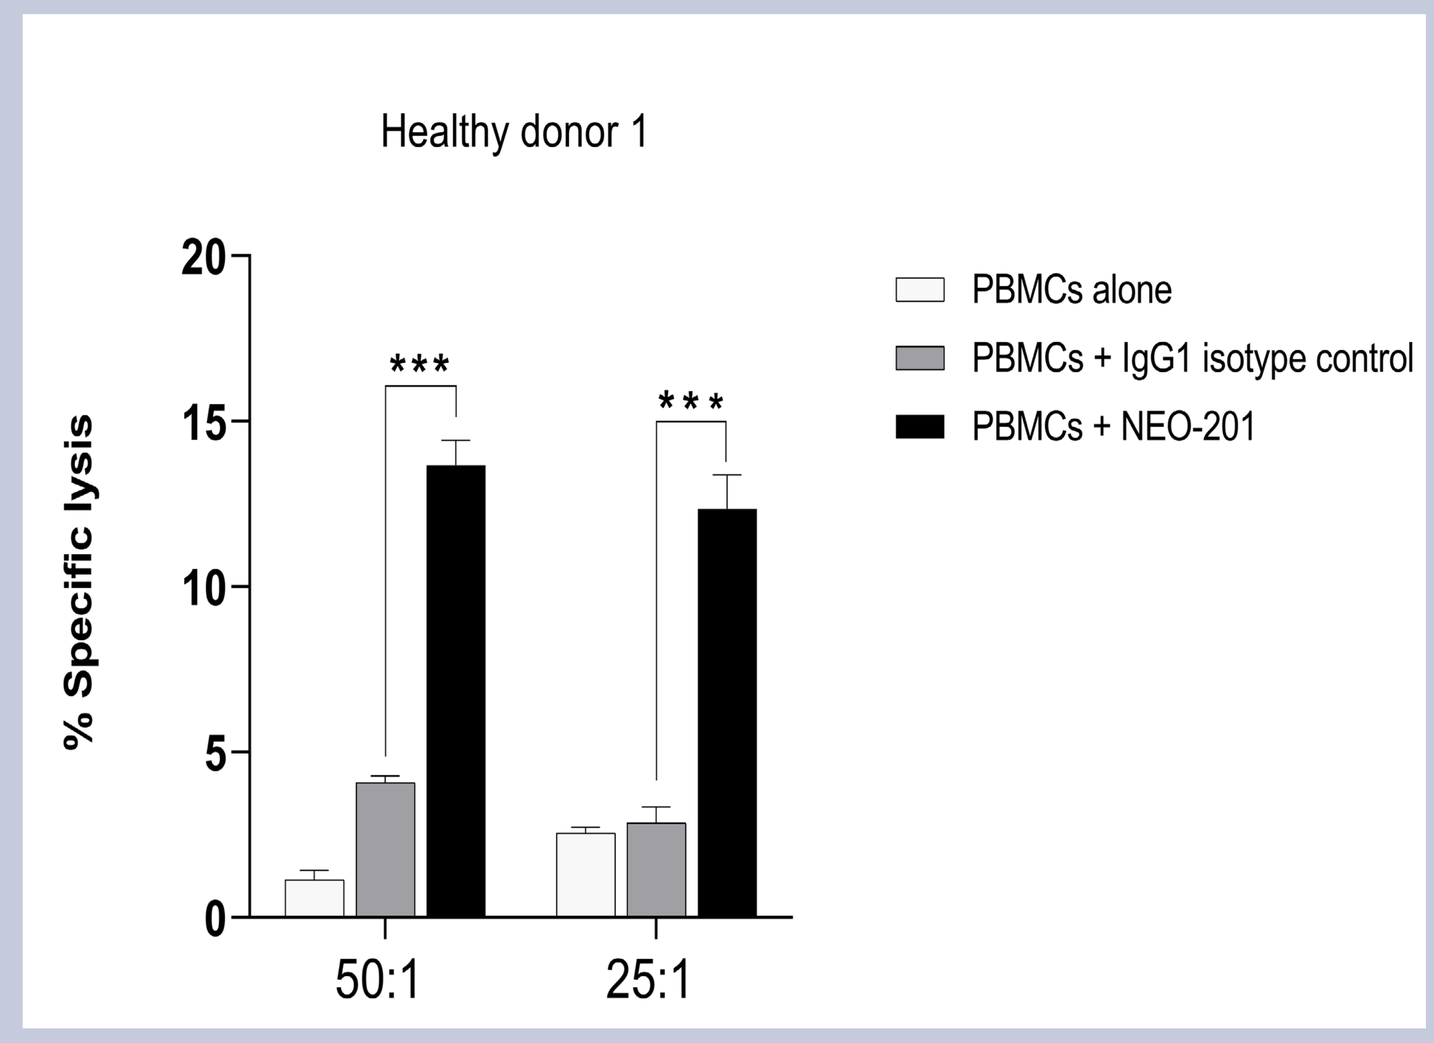
Healthy donor 1
% Specific lysis
PBMCs alone
PBMCs + IgG1 isotype control
PBMCs + NEO-201

50:1
25:1

*** statistically significant ($p < 0.001$) by 2way ANOVA (NEO-201 + PBMCs vs IgG1+ PBMCs).

Isolated NK cells

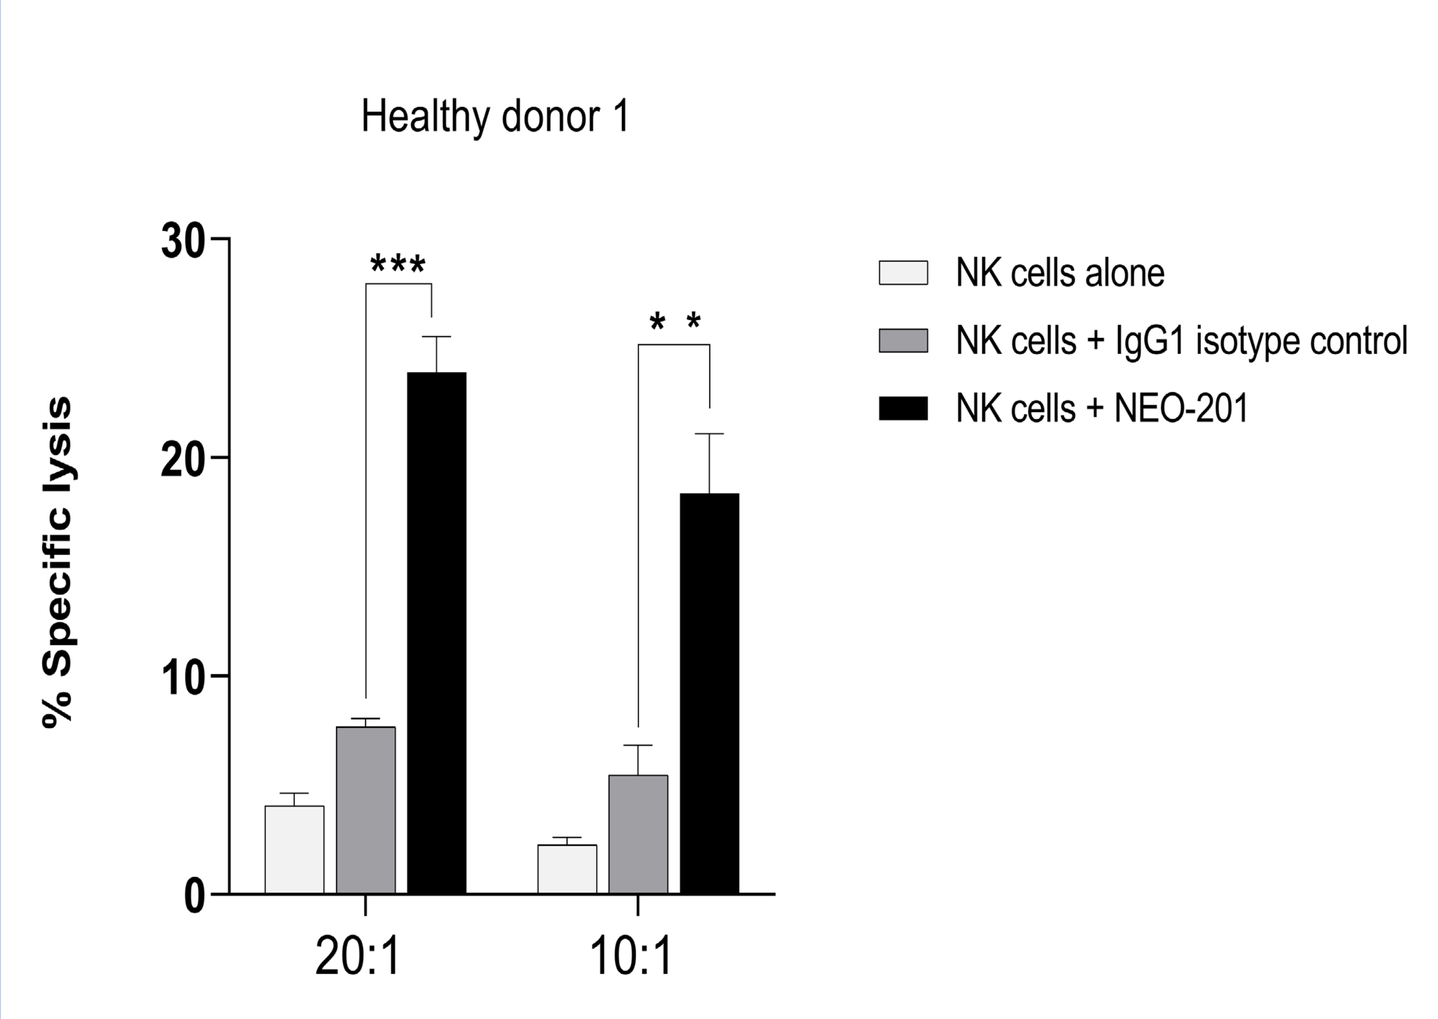
Healthy donor 1
% Specific lysis
NK cells alone
NK cells + IgG1 isotype control
NK cells + NEO-201

* *
20:1
10:1

*** statistically significant ($p < 0.001$) by 2way ANOVA (NEO-201 + NK cells vs IgG1+ NK cells).

** statistically significant ($p < 0.01$) by 2way ANOVA (NEO-201 + NK cells vs IgG1+ NK cells).

HL-60 cells were incubated with 10µg/mL of NEO-201 or human IgG1 (negative control). PBMCs and isolated NK cells from one healthy donor were used as effector cells at the indicated E:T ratios

Results are presented as mean of % specific lyisis ± SD (Standard deviation) from 3 replicate wells in each experiment.

4. NEO-201 mediates ADCC against neutrophils

Flow Cytometry

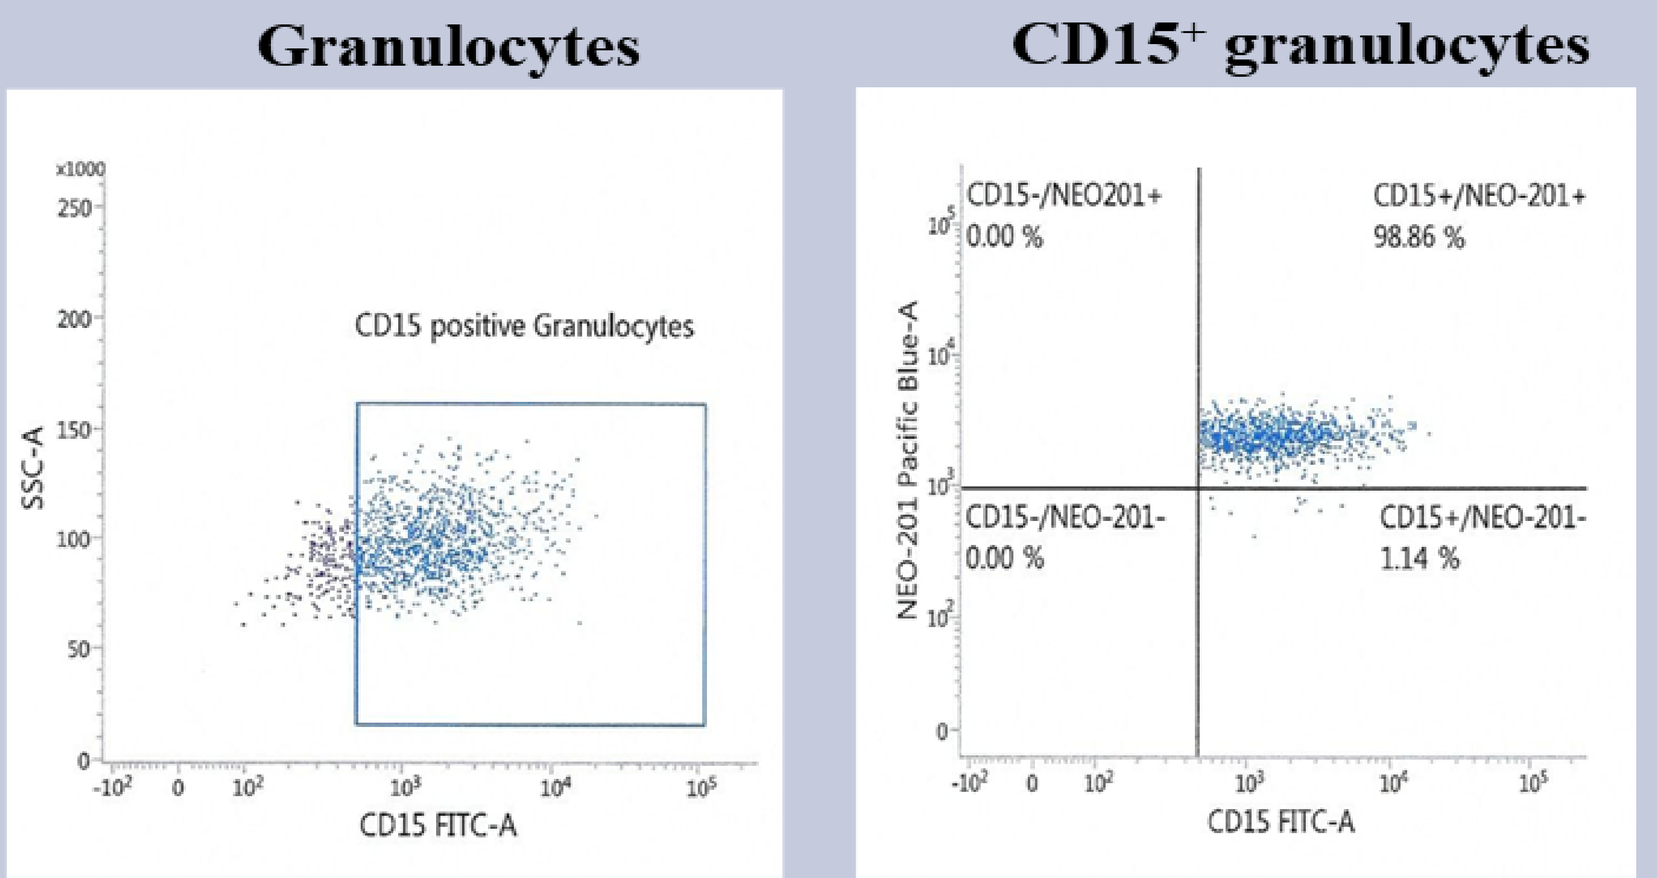
Granulocytes
CD15 positive Granulocytes
SSC-A
CD15 FITC-A
CD15⁺ granulocytes
CD15-/NEO201+
0.00 %
CD15+/NEO-201+
98.86 %
CD15-/NEO-201-
0.00 %
CD15+/NEO-201-
1.14 %
NEO-201 Pacific Blue-A
CD15 FITC-A

Flow cytometry analysis of the binding of NEO-201 to human neutrophils. Data are presented as percentage of CD15+ granulocytes expressing the antigen recognized by NEO-201. Reactivity with NEO-201 was determined using 10 µg/mL Pacific Blue-conjugated NEO-201 (right panel). Positivity was defined as % of positive cells ≥ 10%.

ADCC assay

PBMCs

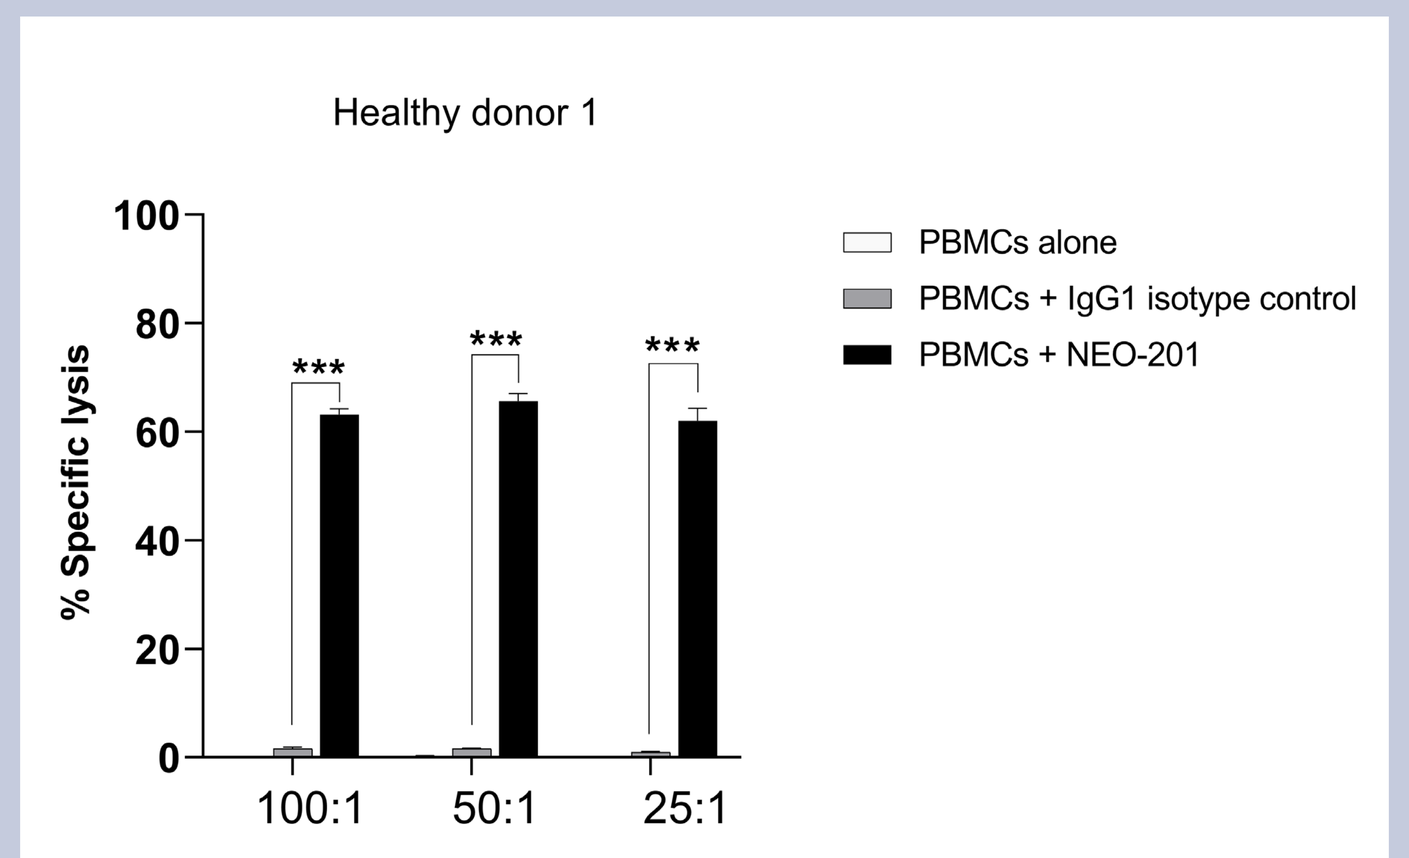
Healthy donor 1
% Specific lysis
PBMCs alone
PBMCs + IgG1 isotype control
PBMCs + NEO-201

100:1
50:1
25:1

Isolated human neutrophils from 1 healthy donor was incubated with 10µg/mL of NEO-201 or human IgG1 (negative control). PBMCs from the same healthy donor were used as effector cells at the indicated E:T ratios

Results are presented as mean of % specific lyisis ± SD (Standard deviation) from 3 replicate wells in each experiment.*** statistically significant ($p < 0.001$) by 2way ANOVA (NEO-201 + PBMCs vs IgG1+ PBMCs).